Variable associations between humoral immune features and immune checkpoint blockade-related outcomes across tissues in metastatic lung adenocarcinoma

Mario G. Rosasco[1], Rotem Ben-Shachar[1], Daniel Morgensztern[2], Michelle M. Stein[1], and Justin Guinney[1]
[1] Tempus AI, Inc., Chicago, IL // [2]Washington University, Division of Oncology, St Louis, MO, USA

Abstract #884

INTRODUCTION

Immune checkpoint blockade (ICB) has improved cancer patient outcomes, yet variable efficacy across metastatic sites motivates further study. Recently, humoral immune features have emerged as ICB biomarkers. Given strong correlations between many prognostic immune features, we evaluated heterogeneity in correlations between humoral and other features across metastatic sites, which may contribute to variable ICB efficacy.

METHODS

De-identified records of metastatic lung adenocarcinoma (mLUAD) patients receiving any ICB-containing regimen in the first line of therapy were selected from the Tempus Database.

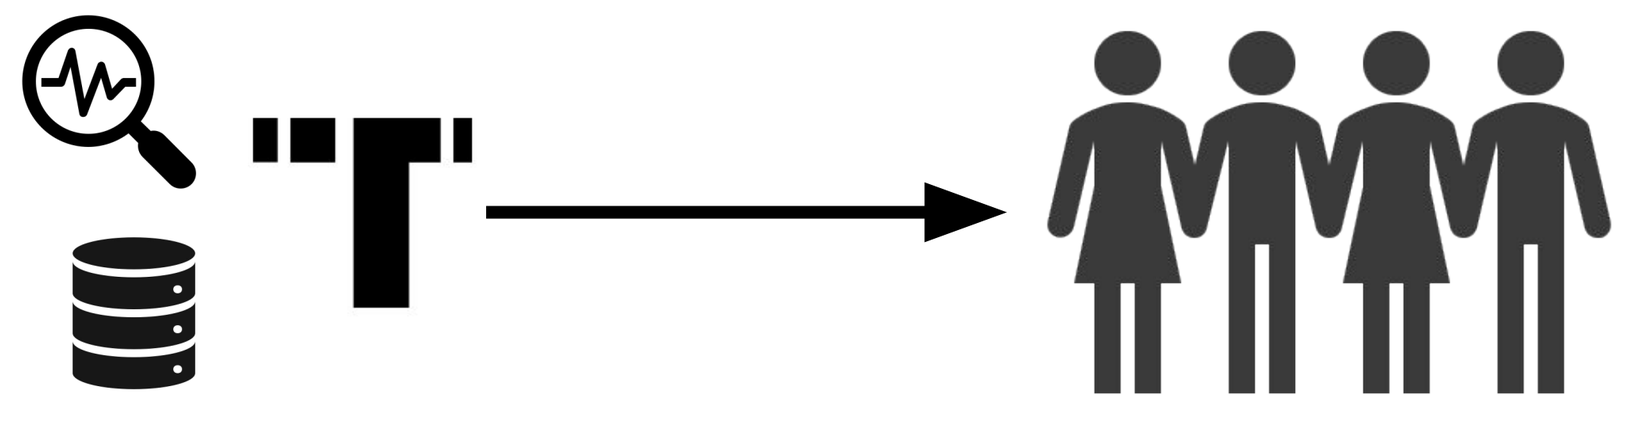

Retrospective analysis *N=1895 mLUAD cases*

Samples were labeled by tissue origin: primary (n=695), liver (n=124), lymphatic (n=306), neural (n=190), or other (n=580).

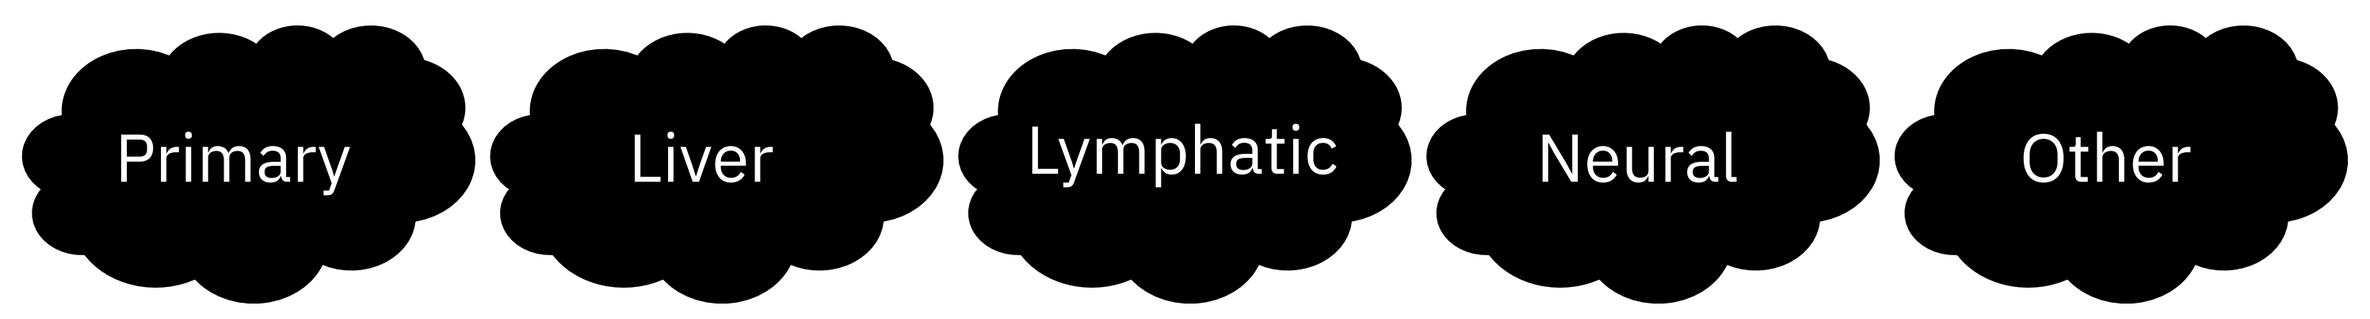
Primary
Liver
Lymphatic
Neural
Other

n=695 *n=306* *n=124* *n=190* *n=580*

From Tempus xR whole-exome capture RNA seq data, immunological trait scores were computed using the Tempus IO platform. Pearson correlations between traits were measured within each tissue, and differences were assessed using a z-test.

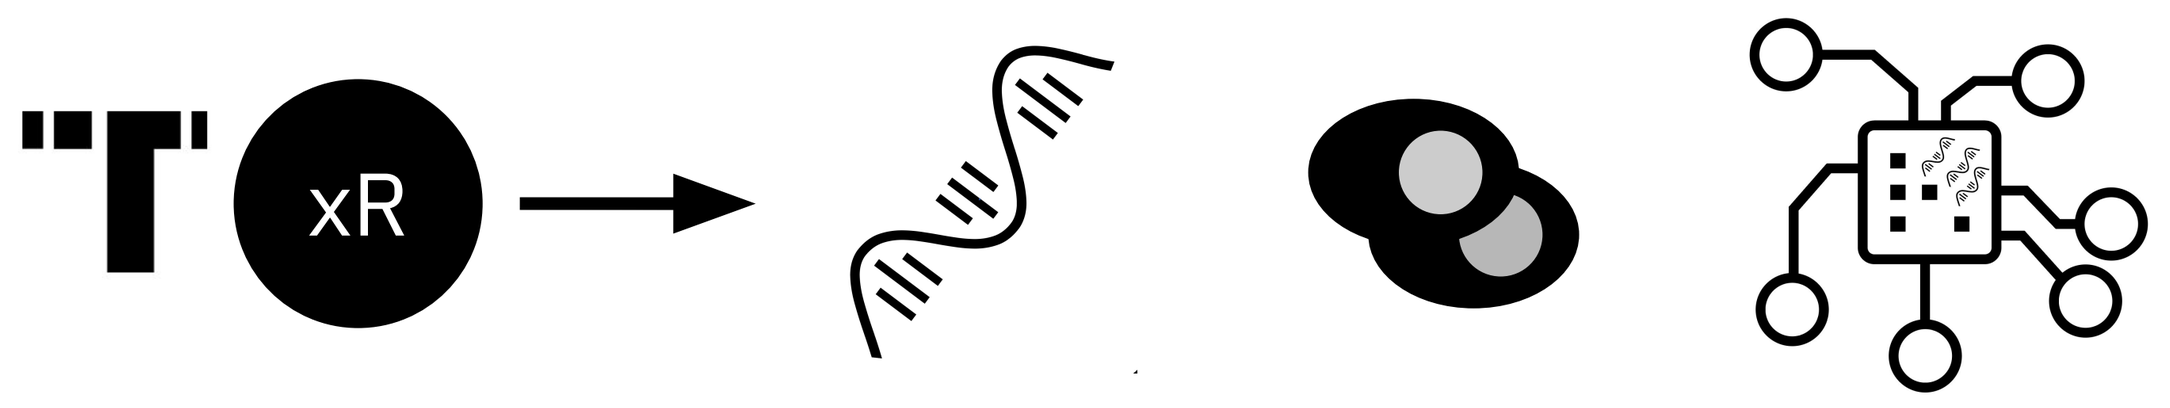
xR

Whole-exome capture RNA seq *Expression* *Infiltration* *Gene Expression Signature Scores*

In patients with a recorded death date or at least 2 years of follow-up (n=1125), features were tested for association with real-world overall survival (rwOS) at 2 years following metastatic diagnosis via a Wilcoxon rank-sum test with multiple testing correction using the Holm method.

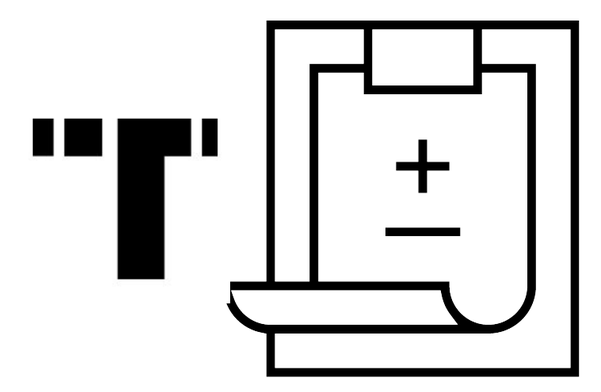

rwOS extracted from clinical records in the Tempus Database

SUMMARY

- In a large real-world cohort of metastatic lung adenocarcinoma patients, we observed significant immunological differences between tissues.
- Plasma cell score associations with survival varied between tissues, suggesting humoral immunity may have tissue-dependent effects on ICB-related clinical outcomes.

RESULTS

Figure 1. Immune trait correlations

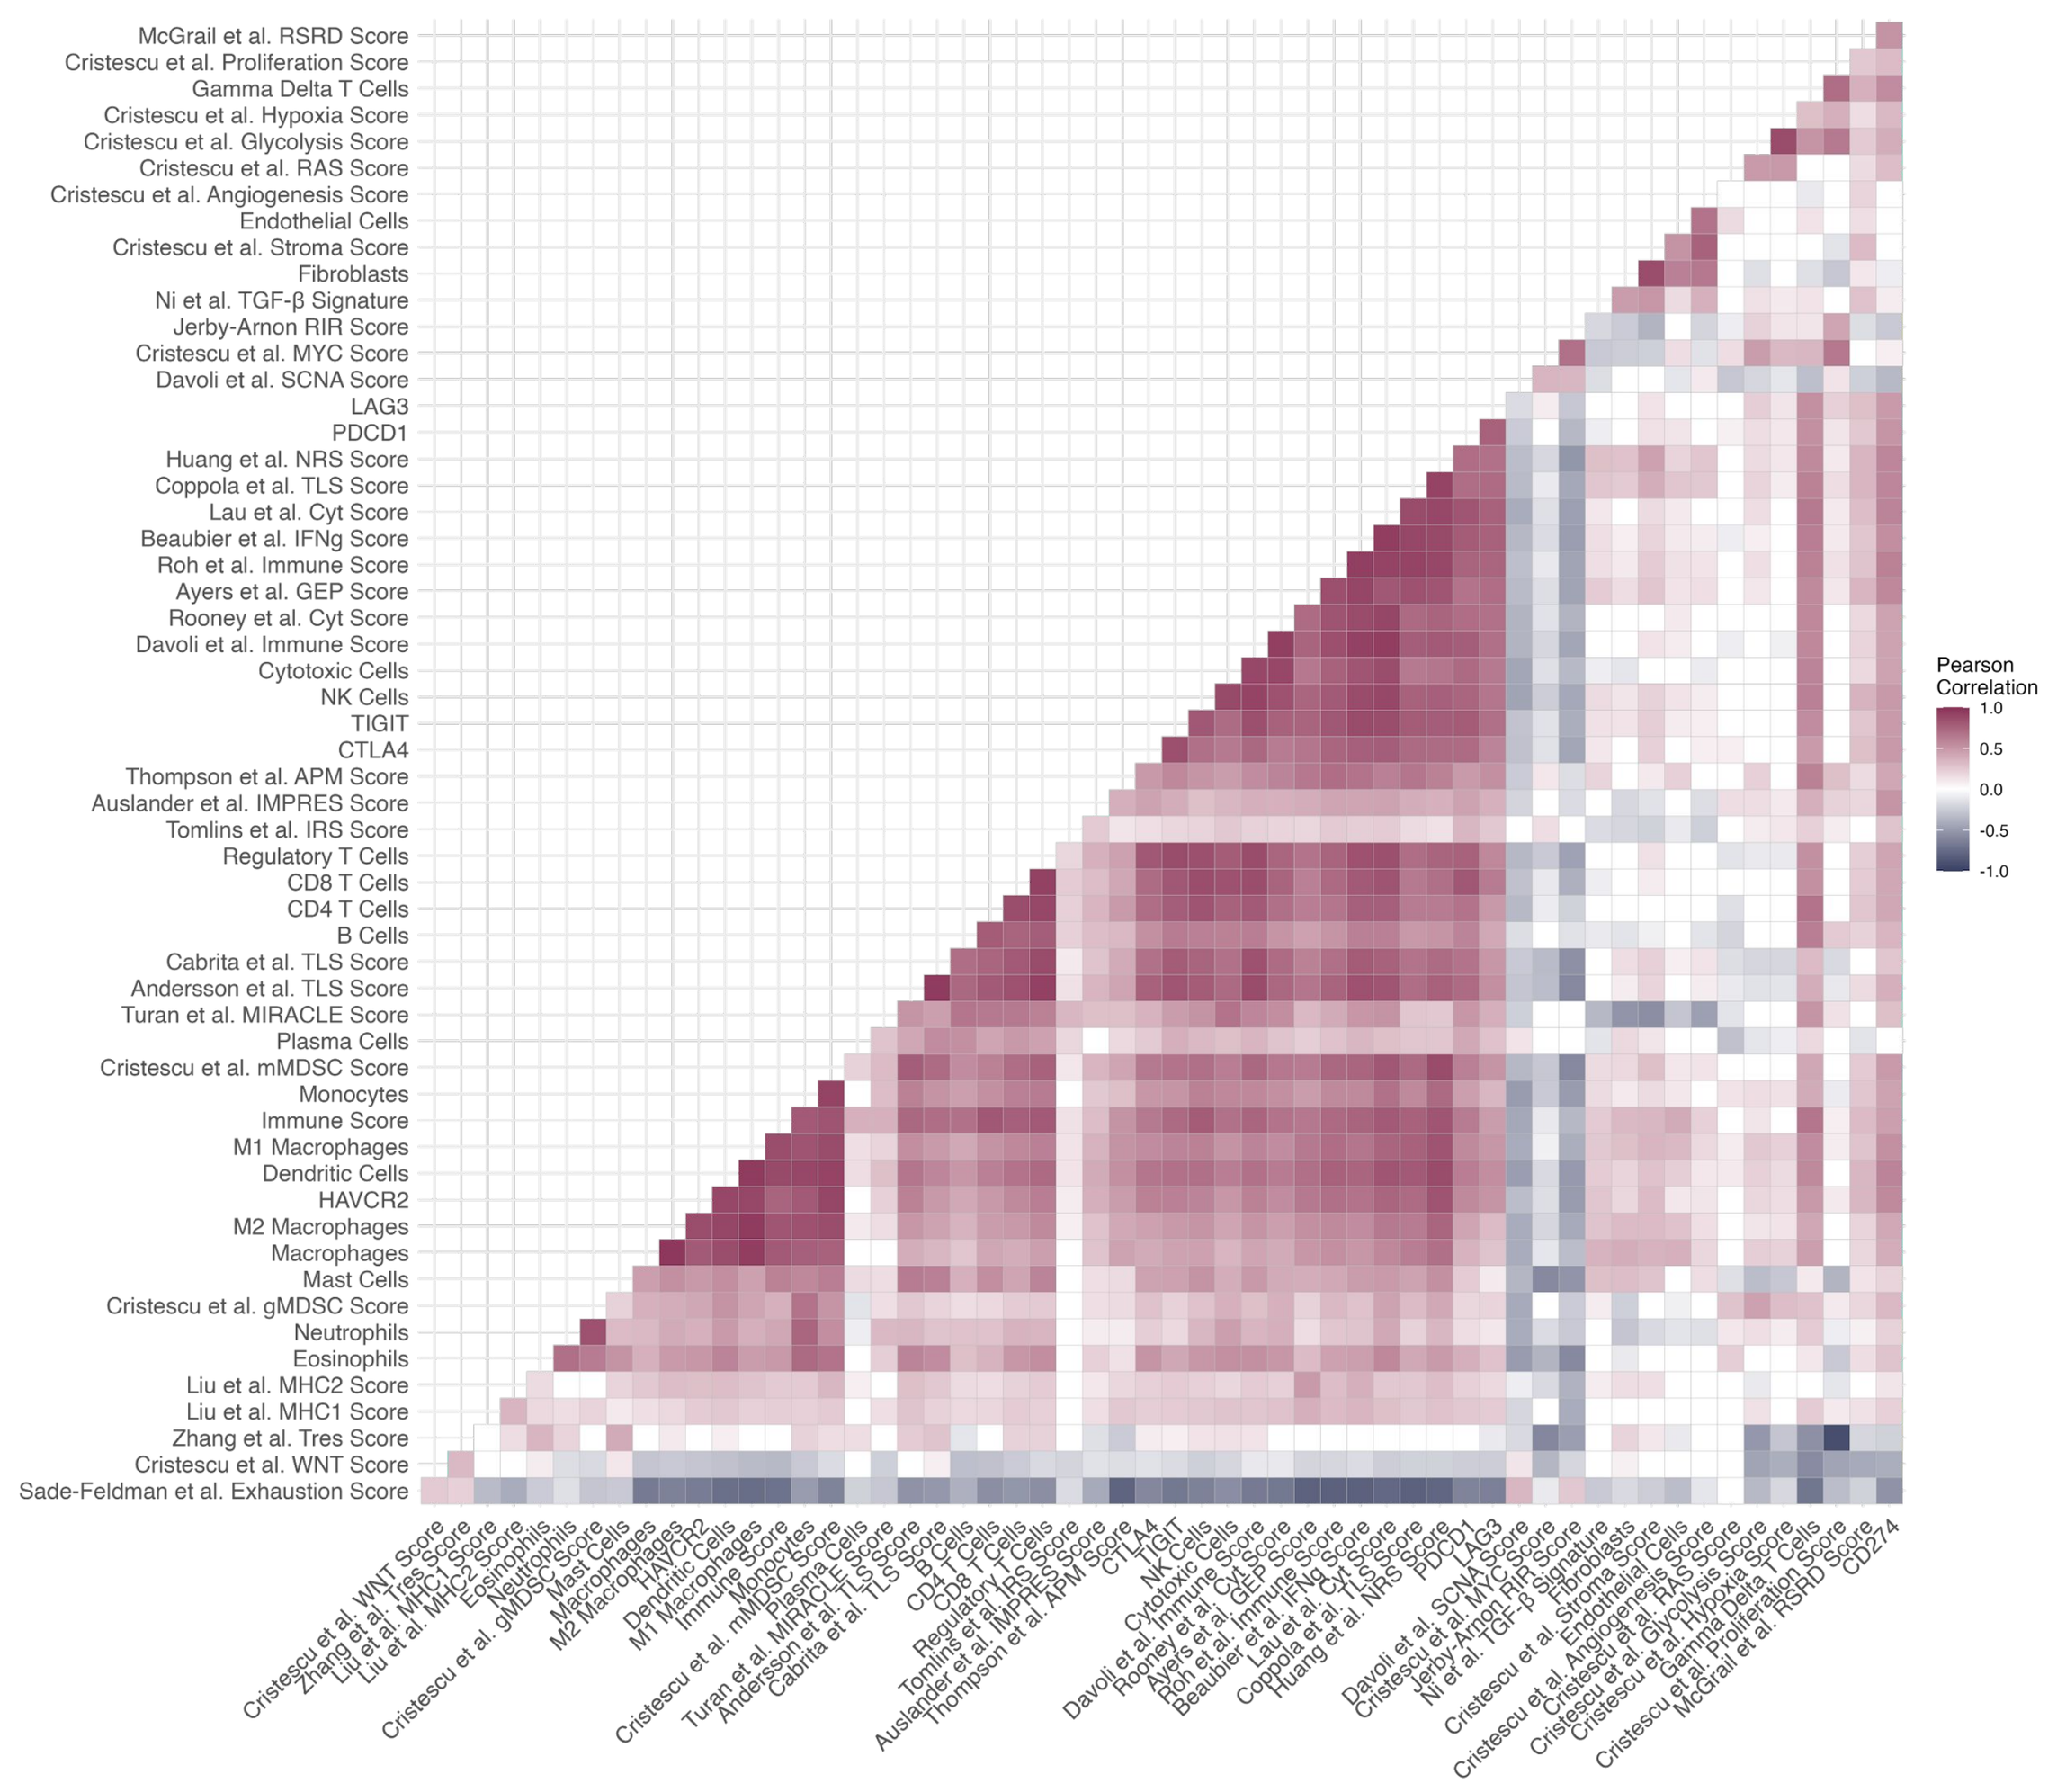
Pearson Correlation
1.0
0.5
0.0
-0.5
-1.0

Figure 1. Many immunological features are highly correlated.

Figure 2. Immune trait correlation differences

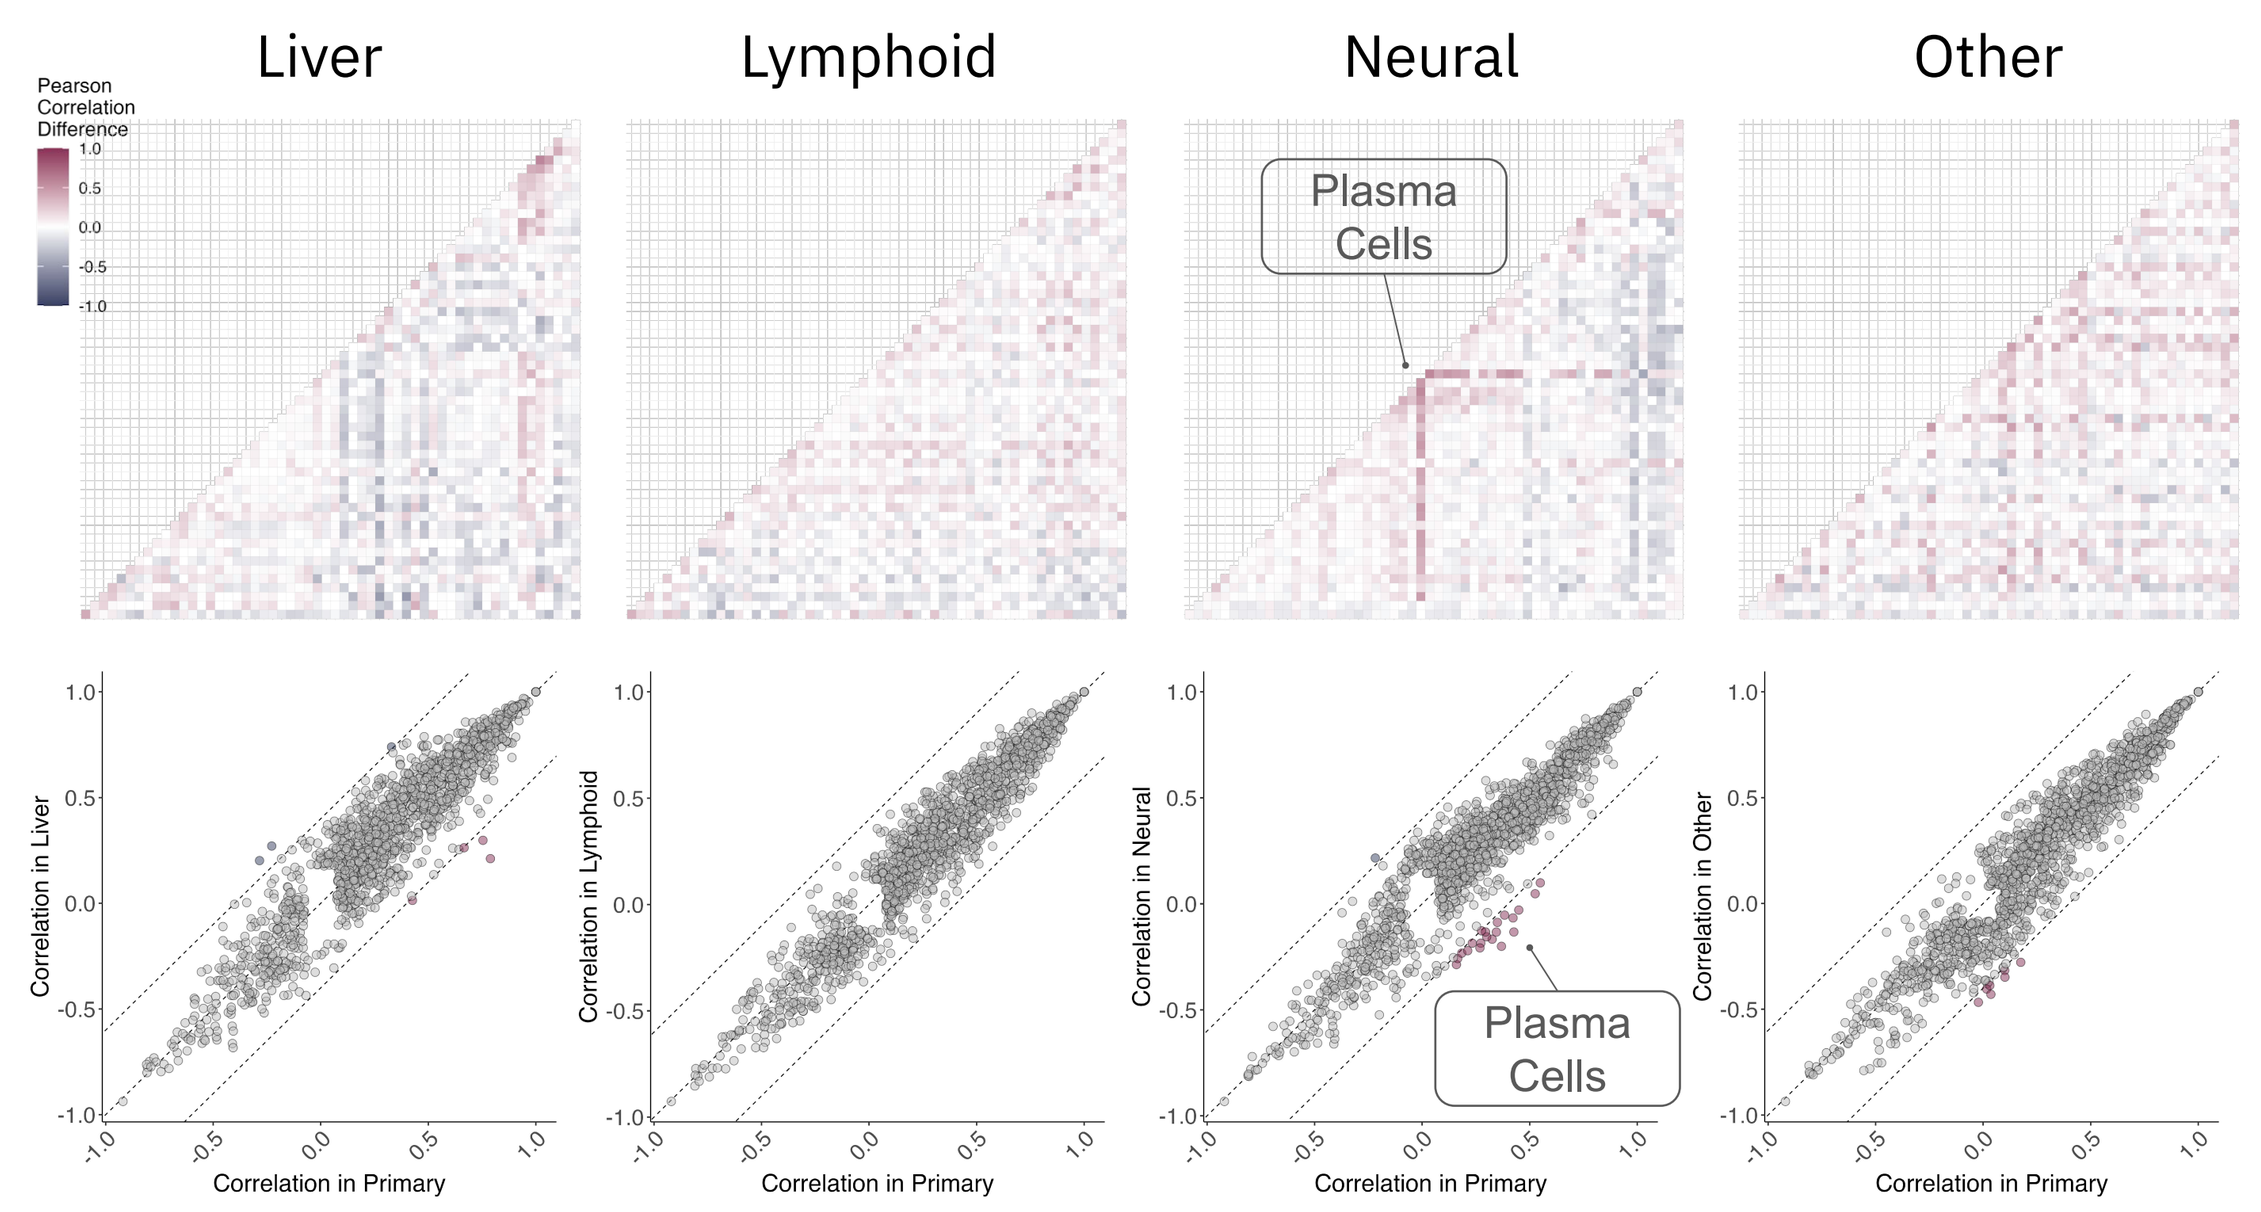
Liver
Lymphoid
Neural
Other
Pearson Correlation Difference
Plasma Cells
Plasma Cells
Correlation in Liver
Correlation in Lymphoid
Correlation in Neural
Correlation in Other
Correlation in Primary

Figure 2. Neural metastases showed pronounced differences in correlations between immunological features and plasma cell scores when compared to correlations in primary tissue. Dotted lines indicate correlation differences of -0.4, 0, and 0.4.

Figure 3. Immune trait correlations across tissues

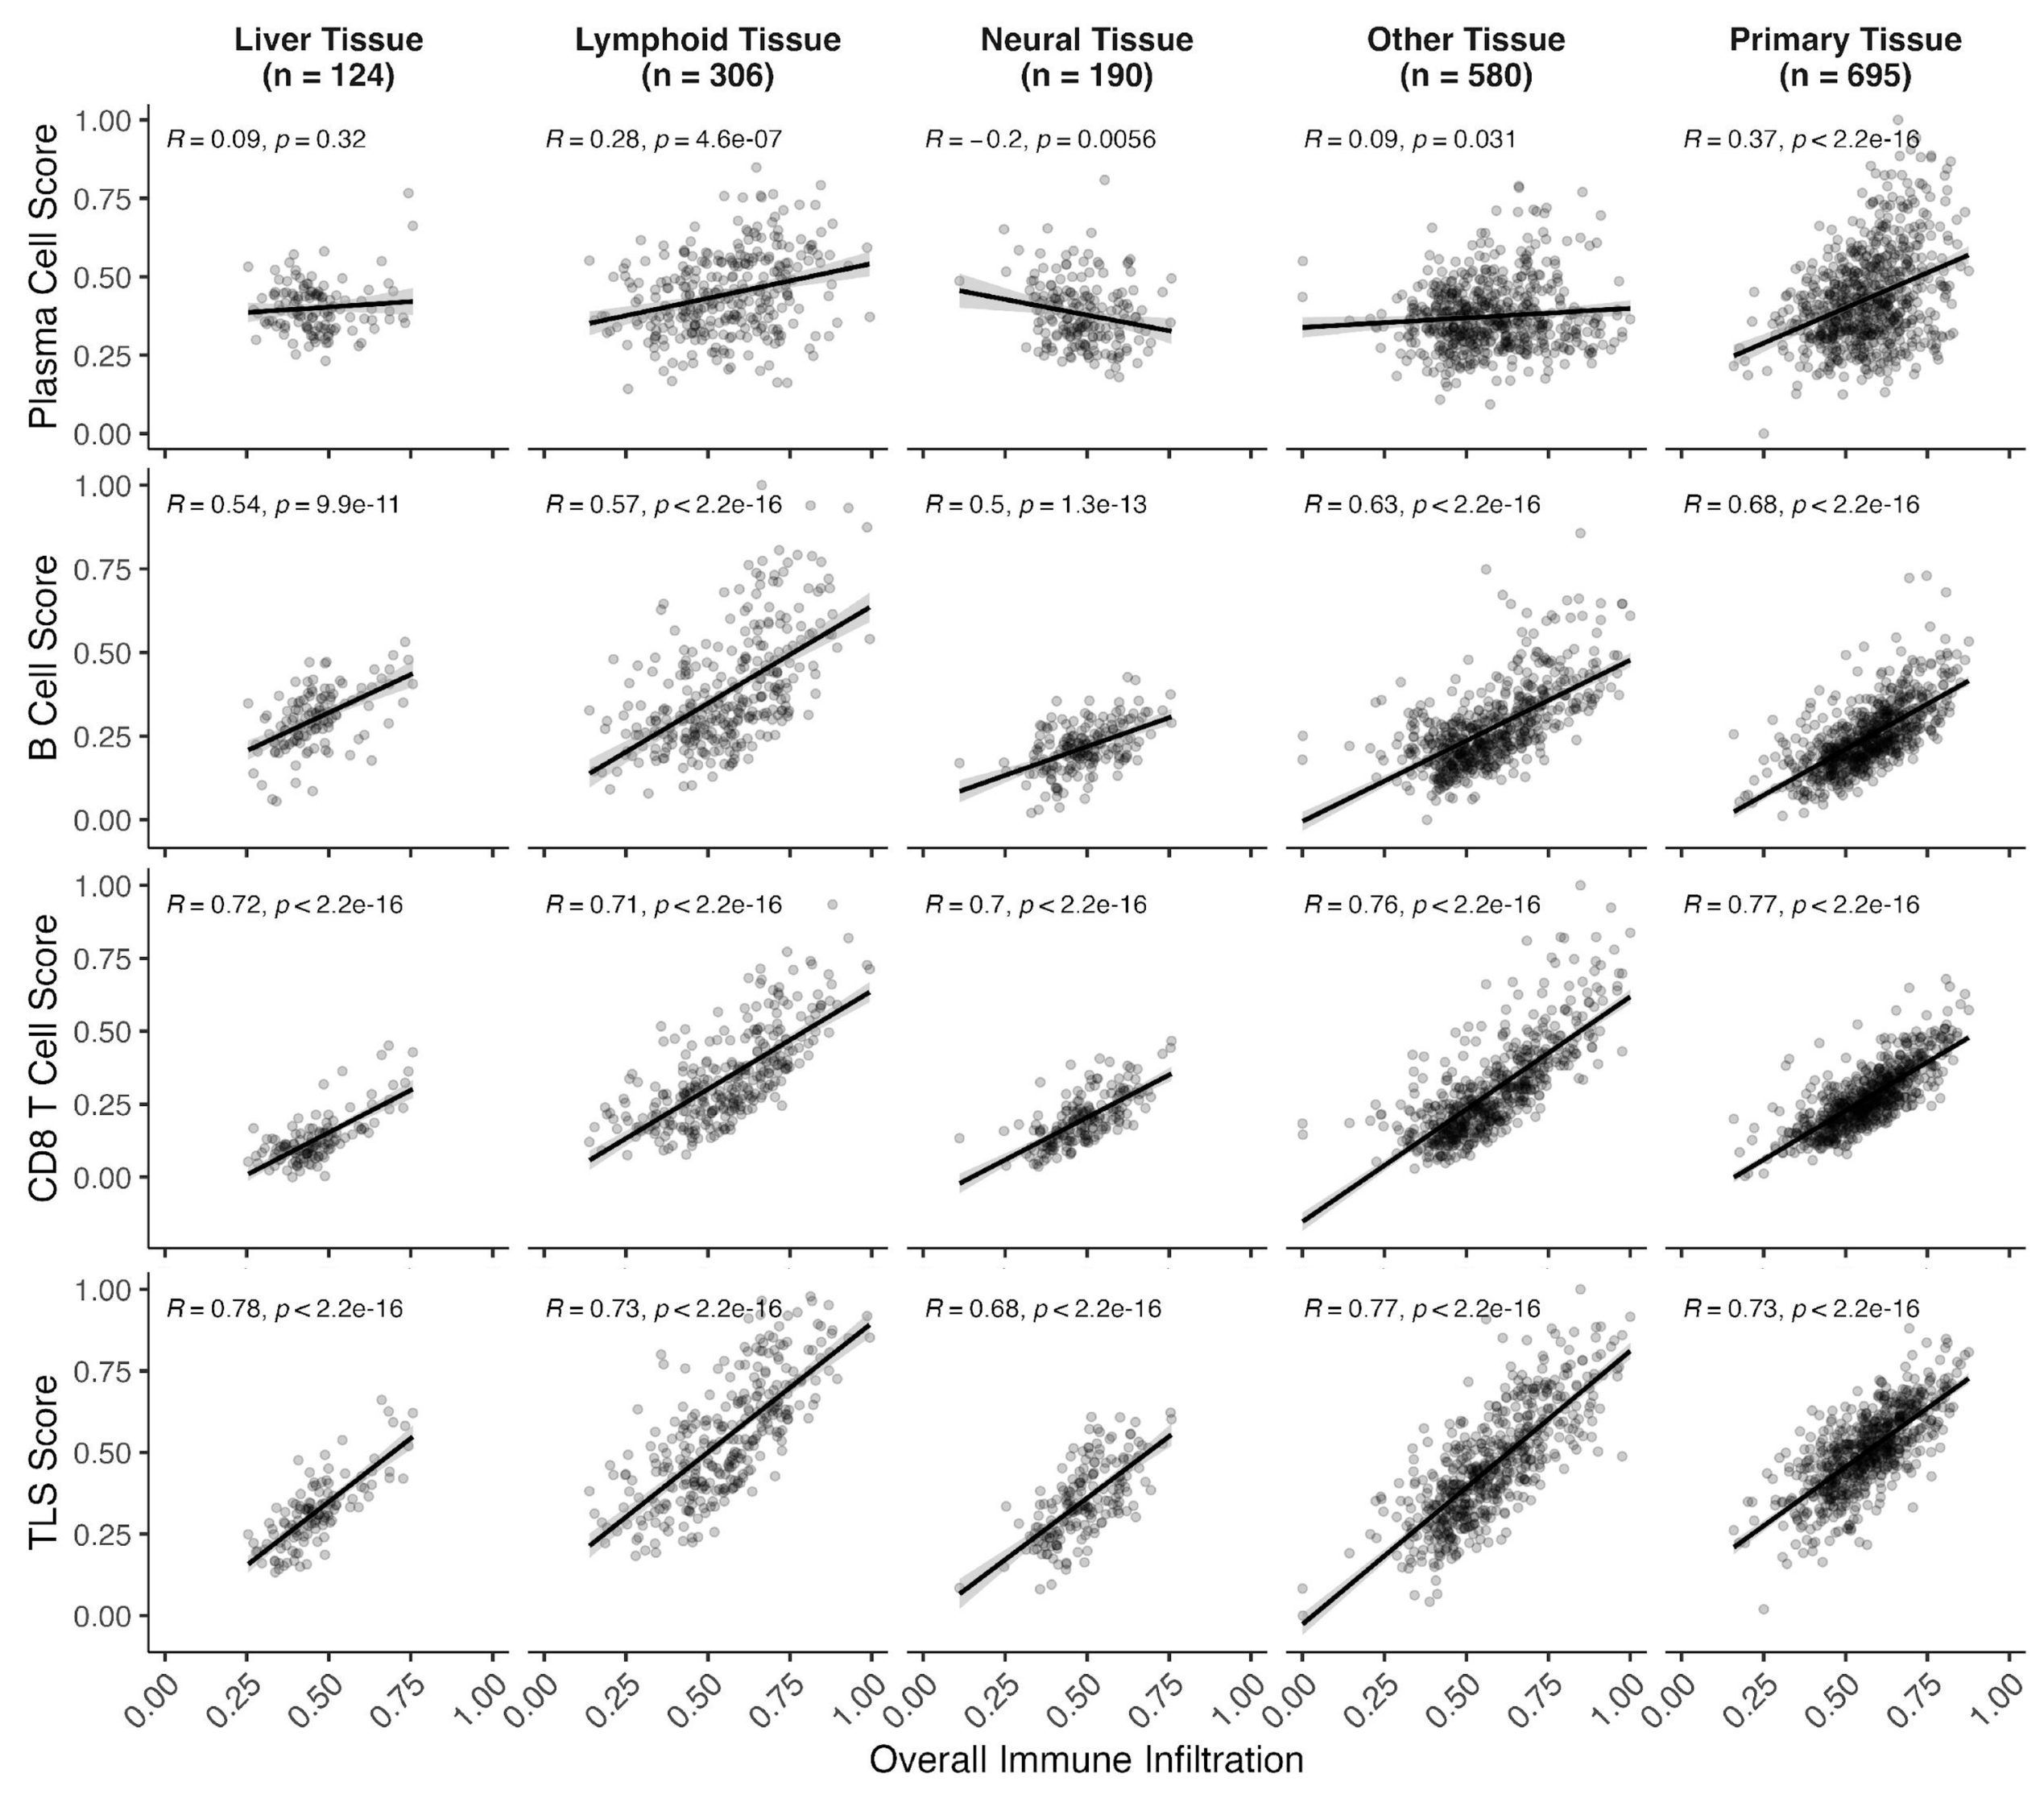
Liver Tissue (n = 124)
Lymphoid Tissue (n = 306)
Neural Tissue (n = 190)
Other Tissue (n = 580)
Primary Tissue (n = 695)
Plasma Cell Score
R = 0.09, p = 0.32
R = 0.28, p = 4.6e-07
R = −0.2, p = 0.0056
R = 0.09, p = 0.031
R = 0.37, p < 2.2e-16
B Cell Score
R = 0.54, p = 9.9e-11
R = 0.57, p < 2.2e-16
R = 0.5, p = 1.3e-13
R = 0.63, p < 2.2e-16
R = 0.68, p < 2.2e-16
CD8 T Cell Score
R = 0.72, p < 2.2e-16
R = 0.71, p < 2.2e-16
R = 0.7, p < 2.2e-16
R = 0.76, p < 2.2e-16
R = 0.77, p < 2.2e-16
TLS Score
R = 0.78, p < 2.2e-16
R = 0.73, p < 2.2e-16
R = 0.68, p < 2.2e-16
R = 0.77, p < 2.2e-16
R = 0.73, p < 2.2e-16
Overall Immune Infiltration

Figure 3. Most immune features remain correlated with overall infiltration across metastatic and primary tissues.

Figure 4. Plasma cell and overall immune score correlations by PD-L1 (*CD274*) expression

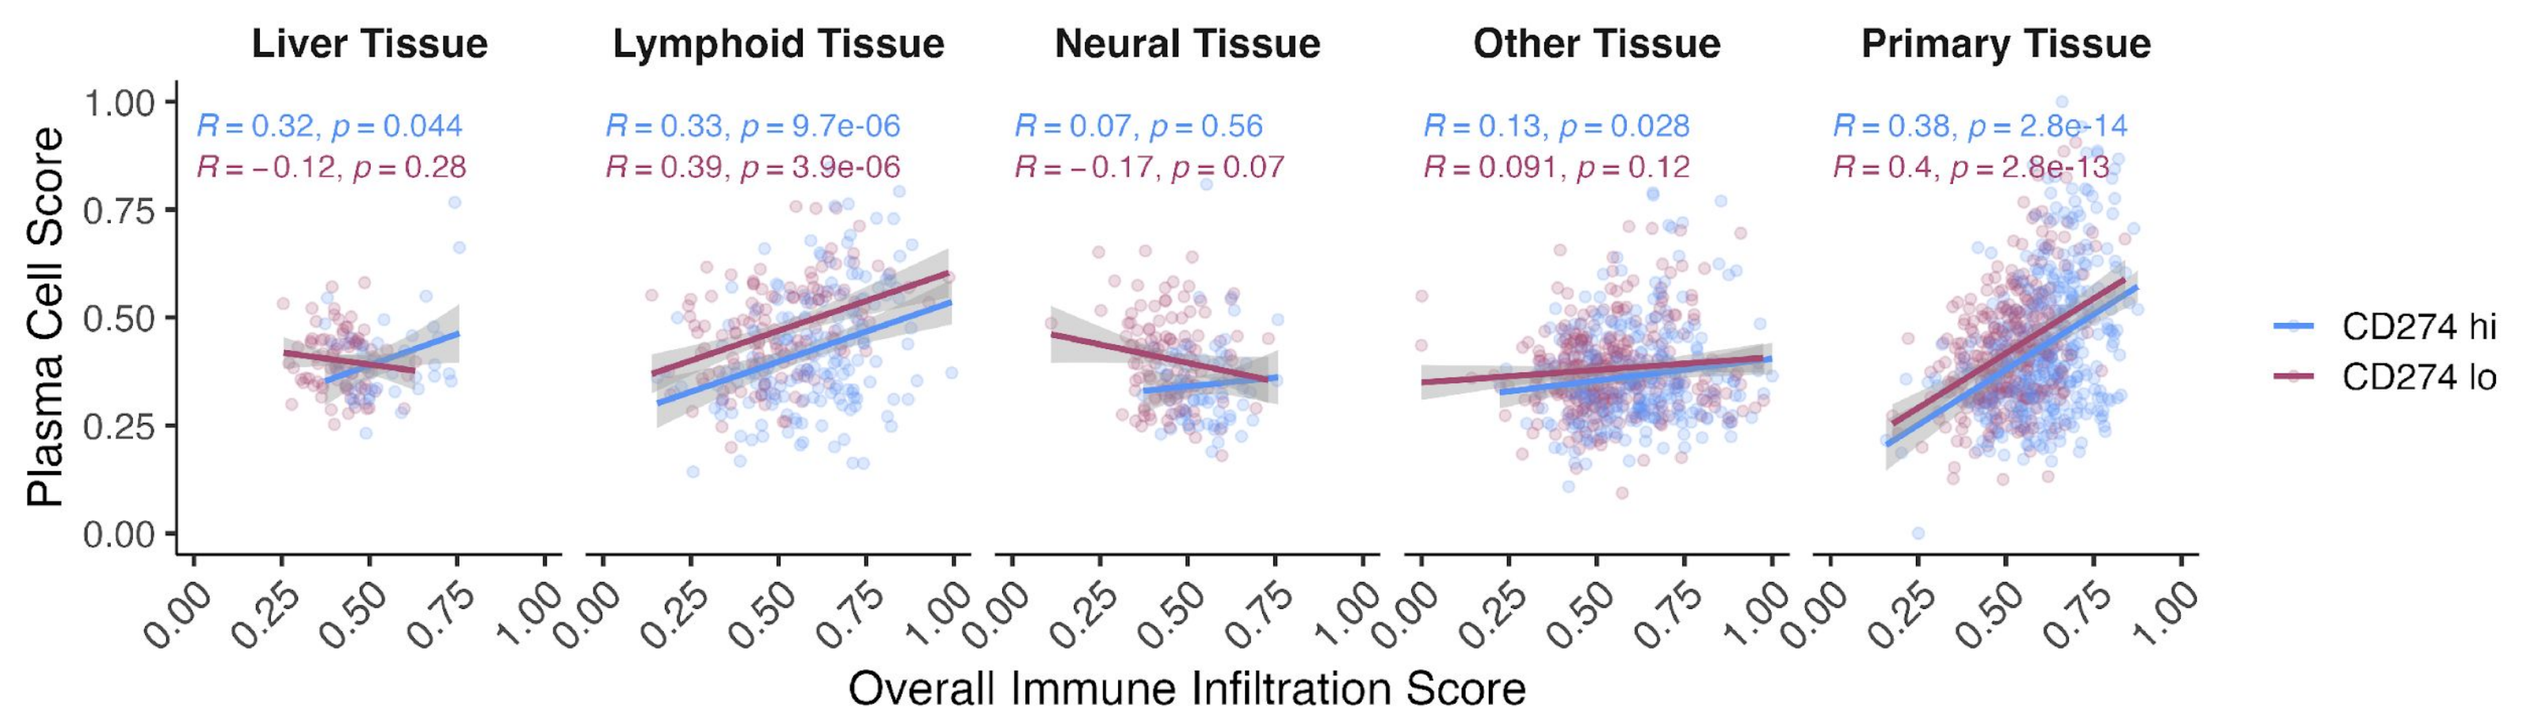
Liver Tissue
Lymphoid Tissue
Neural Tissue
Other Tissue
Primary Tissue
R = 0.32, p = 0.044
R = −0.12, p = 0.28
R = 0.33, p = 9.7e-06
R = 0.39, p = 3.9e-06
R = 0.07, p = 0.56
R = −0.17, p = 0.07
R = 0.13, p = 0.028
R = 0.091, p = 0.12
R = 0.38, p = 2.8e-14
R = 0.4, p = 2.8e-13
Plasma Cell Score
Overall Immune Infiltration Score
CD274 hi
CD274 lo

Figure 4. Differences in plasma cell score correlations with overall immune infiltration can be seen within *CD274* expression subgroups. *CD274* hi/lo groups were defined based on the median expression level in this cohort.

Figure 5. Immune traits and survival at 2 years

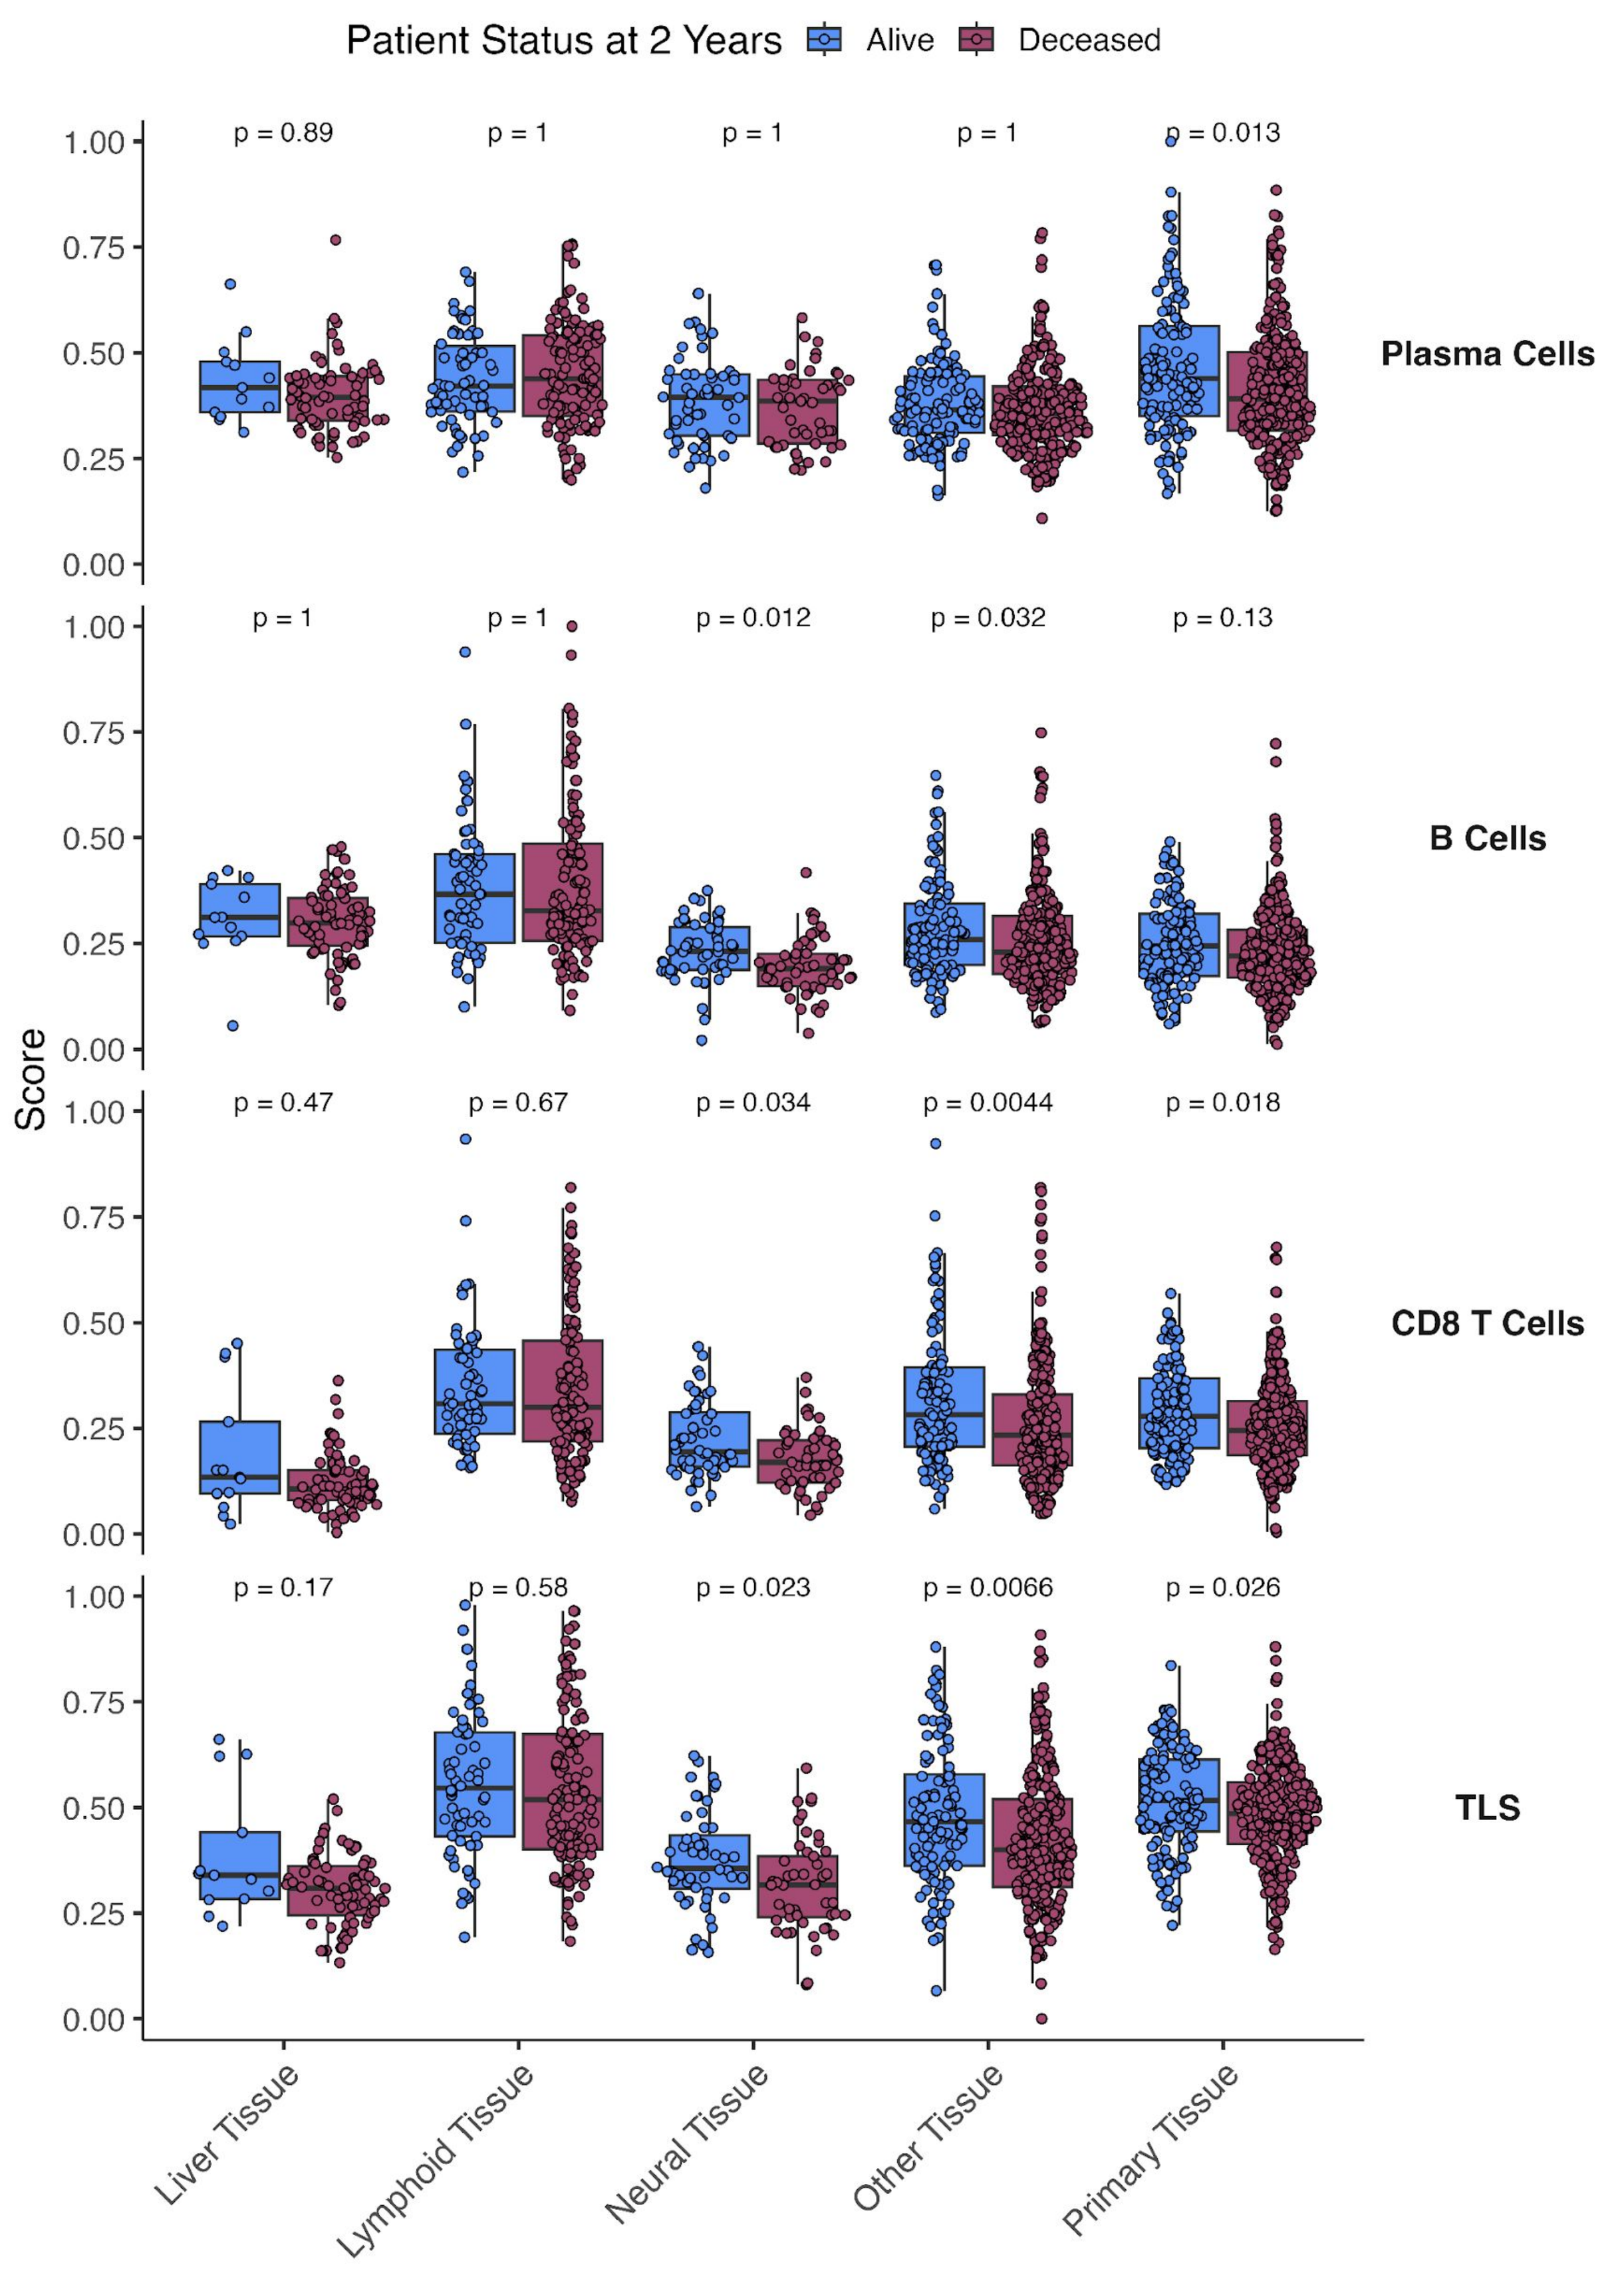
Patient Status at 2 Years
Alive
Deceased
p = 0.89
p = 1
p = 1
p = 1
p = 0.013
Plasma Cells
p = 1
p = 1
p = 0.012
p = 0.032
p = 0.13
B Cells
p = 0.47
p = 0.67
p = 0.034
p = 0.0044
p = 0.018
CD8 T Cells
p = 0.17
p = 0.58
p = 0.023
p = 0.0066
p = 0.026
TLS
Score
Liver Tissue
Lymphoid Tissue
Neural Tissue
Other Tissue
Primary Tissue

Figure 5. Immune features such as B cell, CD8+ T cell, and tertiary lymphoid structure scores associate with rwOS in neural metastases, whereas plasma cell scores only associate with rwOS when measured in primary tissue.

ACKNOWLEDGMENTS

We thank Matthew Kase for poster development.

Correspondence: Justin.Guinney@Tempus.com