Clinical validation of a novel multi-omic algorithm for stratifying outcomes in a real-world cohort of advanced solid cancer patients treated with immune checkpoint inhibitors

Alia Zander[1], Rossin Erbe[1], Yan Liu[1], Ailin Jin[1] Seung Won Hyun[1], Sayantoni Mukhopadhyay[1] , Ben Terdich[1], Mario Rosasco[1] , Nirali Patel[1], Brett Mahon[1], Kate Sasser[1], Michelle Ting-Lin[1], Halla Nimeiri[1], Justin Guinney[1], Kyle Beauchamp[1], Chithra Sangli[1], Michelle Stein[1], Timothy Taxter[1], Timothy Chan[3], Sandip Pravin Patel[2], Ezra Cohen[1, 2]

[1]Tempus AI, Inc., Chicago, IL; [2]Division of Hematology/Oncology, Department of Medicine, University of California San Diego, Moores Cancer Center, La Jolla, CA; [3]Center for Immunotherapy and Precision Immuno-Oncology, Lerner Research Institute, Cleveland Clinic Foundation, Cleveland, OH

Poster/Abstract #
188

INTRODUCTION

Despite advances in immune checkpoint inhibitor (ICI) biomarker molecular testing, there remains an unmet clinical need for more sensitive and generalizable biomarkers to better predict patient outcomes on ICI. This has been challenging due to the limited availability of multi-omic testing and validation cohorts. An integrated DNA/RNA ICI biomarker can address this critical unmet need.

METHODS

A de-identified pan-cancer cohort from the Tempus multimodal real-world database was used for the development and validation of the Immune Profile Score (IPS) algorithm leveraging Tempus xT (648 gene DNA panel) and xR (RNAseq). The cohort (n=1707 training [T]; n=1600 validation [V]) consisted of advanced stage cancer patients treated with any ICI containing regimen as the first (1L) or second (2L) line of therapy. The IPS model was developed utilizing a machine learning framework that includes tumor mutational burden (TMB) and 11 RNA-based biomarkers as features. Cox Proportional Hazards (CoxPH) models were fit to demonstrate prognostic utility. Predictive utility of IPS was evaluated in an exploratory analysis using a Cox model for recurrent events.

COHORT

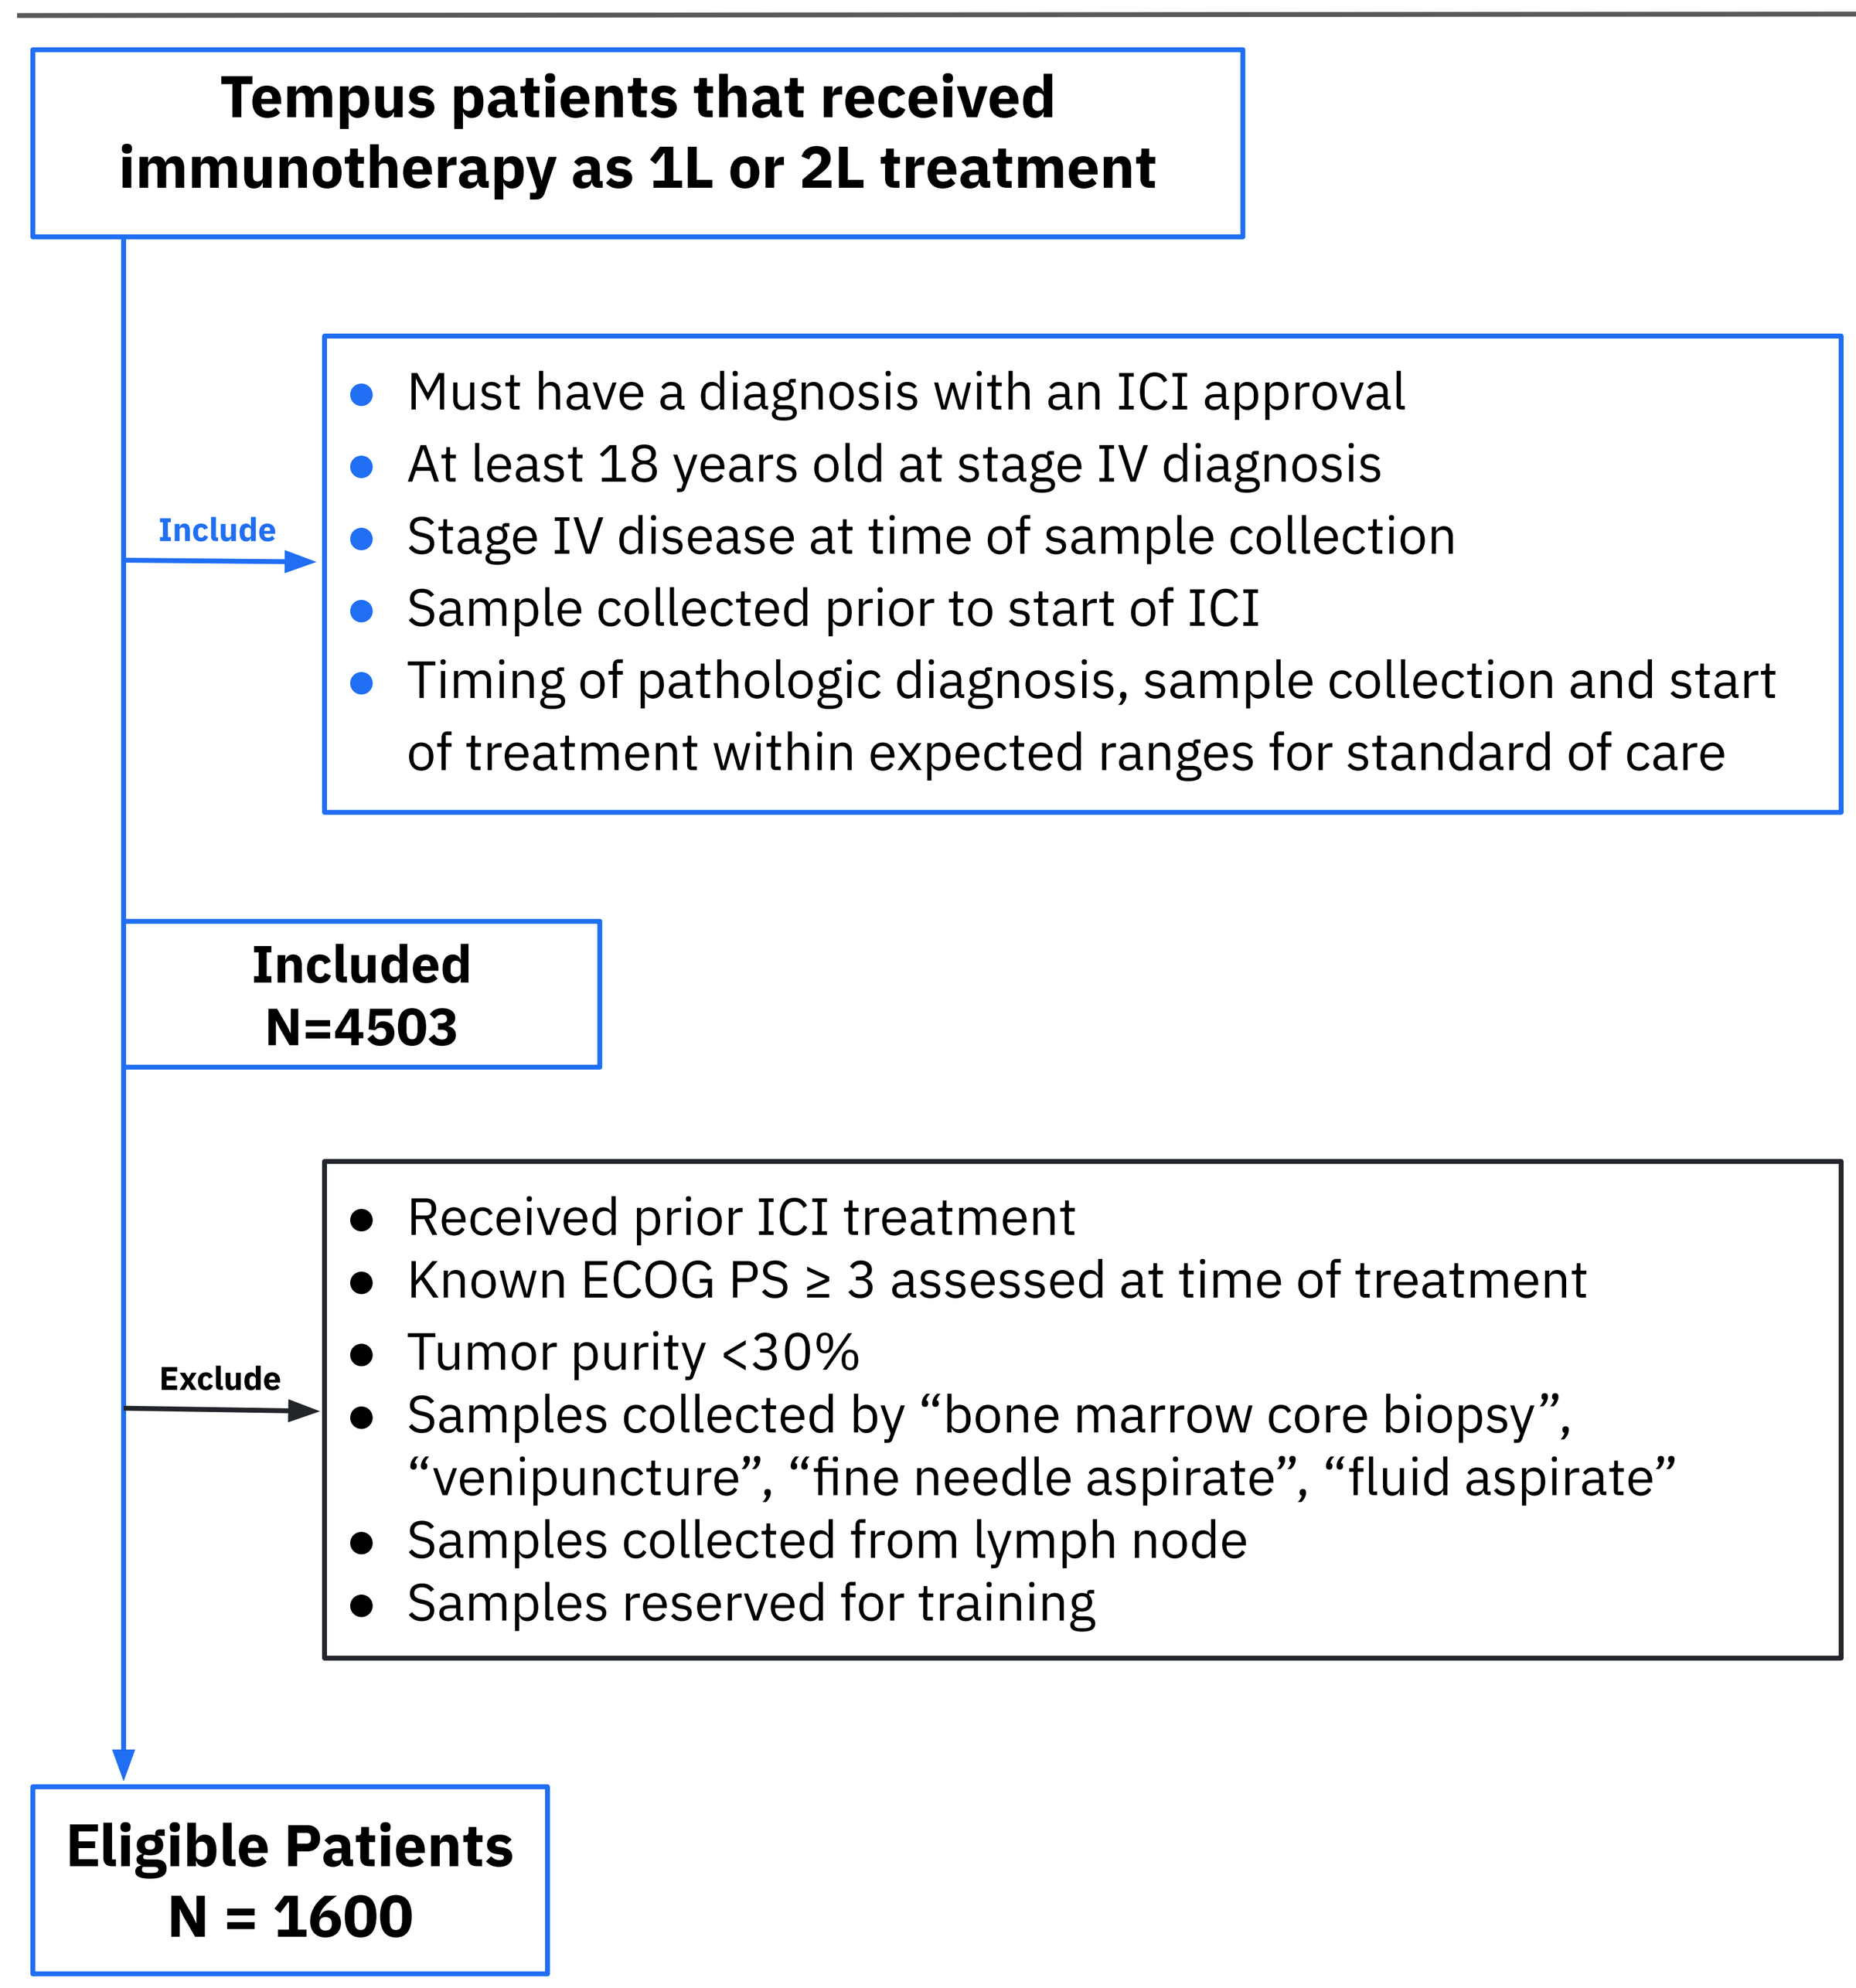
Tempus patients that received immunotherapy as 1L or 2L treatment
Include
Must have a diagnosis with an ICI approval
At least 18 years old at stage IV diagnosis
Stage IV disease at time of sample collection
Sample collected prior to start of ICI
Timing of pathologic diagnosis, sample collection and start of treatment within expected ranges for standard of care
Included N=4503
Exclude
Received prior ICI treatment
Known ECOG PS ≥ 3 assessed at time of treatment
Tumor purity <30%
Samples collected by "bone marrow core biopsy", "venipuncture", "fine needle aspirate", "fluid aspirate"
Samples collected from lymph node
Samples reserved for training
Eligible Patients N = 1600

Figure 1. Cohort funnel showing inclusion and exclusion criteria for entrance into the validation cohort. Maximum follow-up for this study was 24 months.

SUMMARY

- Our results demonstrate that IPS is a generalizable multi-omic biomarker that can be widely used clinically as a prognosticator of ICI-based regimens.
- IPS-high may identify patients (e.g. within TMB-L, MSS, PD-L1 low subgroups) who may benefit from ICI beyond what is predicted by standard biomarkers.
- An exploratory analysis is suggestive of predictive utility. Future prospective predictive utility studies are planned.

RESULTS

Figure 2. IPS model features

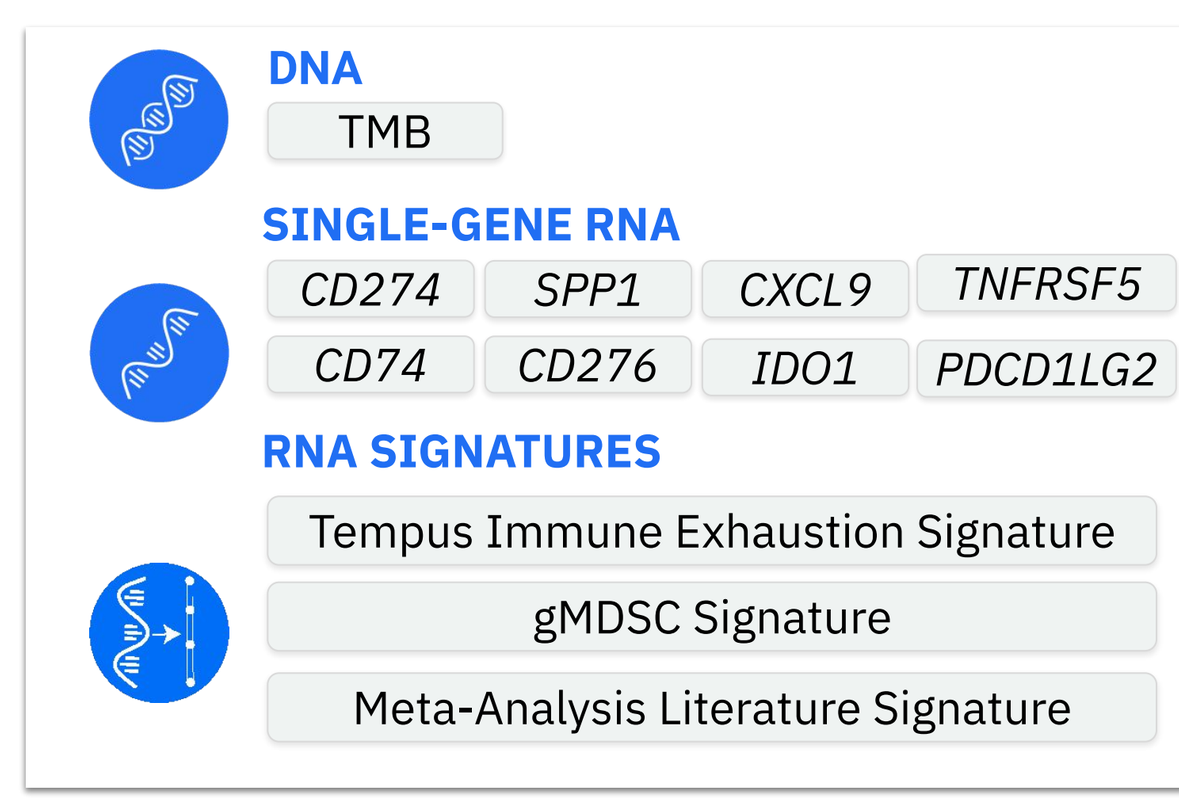
DNA
TMB
SINGLE-GENE RNA
CD274
SPP1
CXCL9
TNFRSF5
CD74
CD276
IDO1
PDCD1LG2
RNA SIGNATURES
Tempus Immune Exhaustion Signature
gMDSC Signature
Meta-Analysis Literature Signature

Figure 2: The Tempus IO Platform was leveraged for developing the IPS model. Various machine learning (ML) techniques were implemented to reduce the feature space. The final IPS model includes 11 RNA-based features and TMB.

Table 1. Patient characteristics

Characteristics	Overall N=1600	IPS-High N=576	IPS-Low N=943	Indeterminate N=81
Age				
Mean (SD)	64.6 (11.8)	64.9 (12.1)	64.5 (11.6)	64.2 (11.7)
Sex				
Female	645 (40%)	252 (44%)	360 (38%)	33 (41%)
Male	955 (60%)	324 (56%)	583 (62%)	48 (59%)
Brain metastases documented	265 (17%)	107 (19%)	143 (15%)	15 (19%)
Liver metastases documented	362 (23%)	94 (16%)	252 (27%)	16 (20%)
ECOG				
0	334 (21%)	137 (24%)	186 (20%)	11 (14%)
1	473 (30%)	168 (29%)	279 (30%)	26 (32%)
2	140 (9%)	45 (8%)	93 (10%)	2 (2%)
Unknown/Missing	653 (40%)	226 (39%)	385 (40%)	42 (52%)
Stage at primary Dx				
Stage I	47 (3%)	22 (4%)	23 (2%)	2 (2%)
Stage II	68 (4%)	25 (4%)	41 (4%)	2 (2%)
Stage III	94 (6%)	33 (6%)	58 (6%)	3 (4%)
Stage IV	1,219 (76%)	430 (75%)	721 (76%)	68 (84%)
Unknown/Missing	172 (11%)	66 (11%)	100 (11%)	6 (7%)
Cancer type				
Breast	86 (5%)	40 (47%)	41 (48%)	5 (6%)
Colorectal	46 (3%)	27 (59%)	18 (39%)	1 (2%)
Gastroesophageal	171 (11%)	26 (15%)	134 (78%)	11(6%)
Hepatocellular	40 (2%)	16 (40%)	22 (55%)	2 (5%)
HNSCC	125 (8%)	35 (28%)	86 (69%)	4 (3%)
Melanoma	102 (6%)	56 (55%)	42 (41%)	4 (4%)
NSCLC	647 (40%)	248 (38%)	367 (57%)	32 (5%)
Renal cell carcinoma	131 (8%)	69 (53%)	49 (37%)	13 (10%)
Urothelial	137 (9%)	36 (26%)	95 (69%)	6 (4%)
Other	115 (7%)	23 (20%)	89 (77%)	3 (3%)
Line of therapy				
1L	1,326 (83%)	482 (84%)	774 (82%)	70 (86%)
2L	274 (17%)	94 (16%)	169 (18%)	11 (14%)
Treatment regimen				
ICI Only	**534 (33%)**	**215 (37%)**	**292 (31%)**	**27 (33%)**
ICI mono	381 (24%)	146 (25%)	219 (23%)	16 (20%)
ICI doublet	153 (9.6%)	69 (12%)	73 (7.7%)	11 (14%)
ICI + Other	**1,066 (67%)**	**361 (63%)**	**651 (69%)**	**54 (67%)**
ICI+Chemo	869 (54%)	283 (49%)	542 (57%)	44 (54%)
ICI+Chemo+Other	72 (4.5%)	15 (2.6%)	55 (5.8%)	2 (2.5%)
ICI+Other	125 (7.8%)	63 (11%)	54 (5.7%)	8 (9.9%)
PD-L1 by IHC				
Negative	495 (31%)	149 (26%)	321 (34%)	25 (31%)
Positive	637 (40%)	250 (43%)	353 (37%)	34 (42%)
Unknown/Missing	468 (29%)	177 (31%)	269 (29%)	22 (27%)
TMB				
High	430 (27%)	250 (43%)	160 (17%)	20 (25%)
Low	1,170 (73%)	326 (57%)	783 (83%)	61 (75%)
MSI				
High	80 (5.0%)	45 (7.8%)	31 (3.3%)	4 (4.9%)
Stable	1517 (95%)	531 (92%)	909 (96%)	77 (95%)
Undetermined	3 (0.2%)	0 (0%)	3 (0.3%)	0 (0%)

Figure 3. OS significantly higher in IPS-H vs. IPS-L

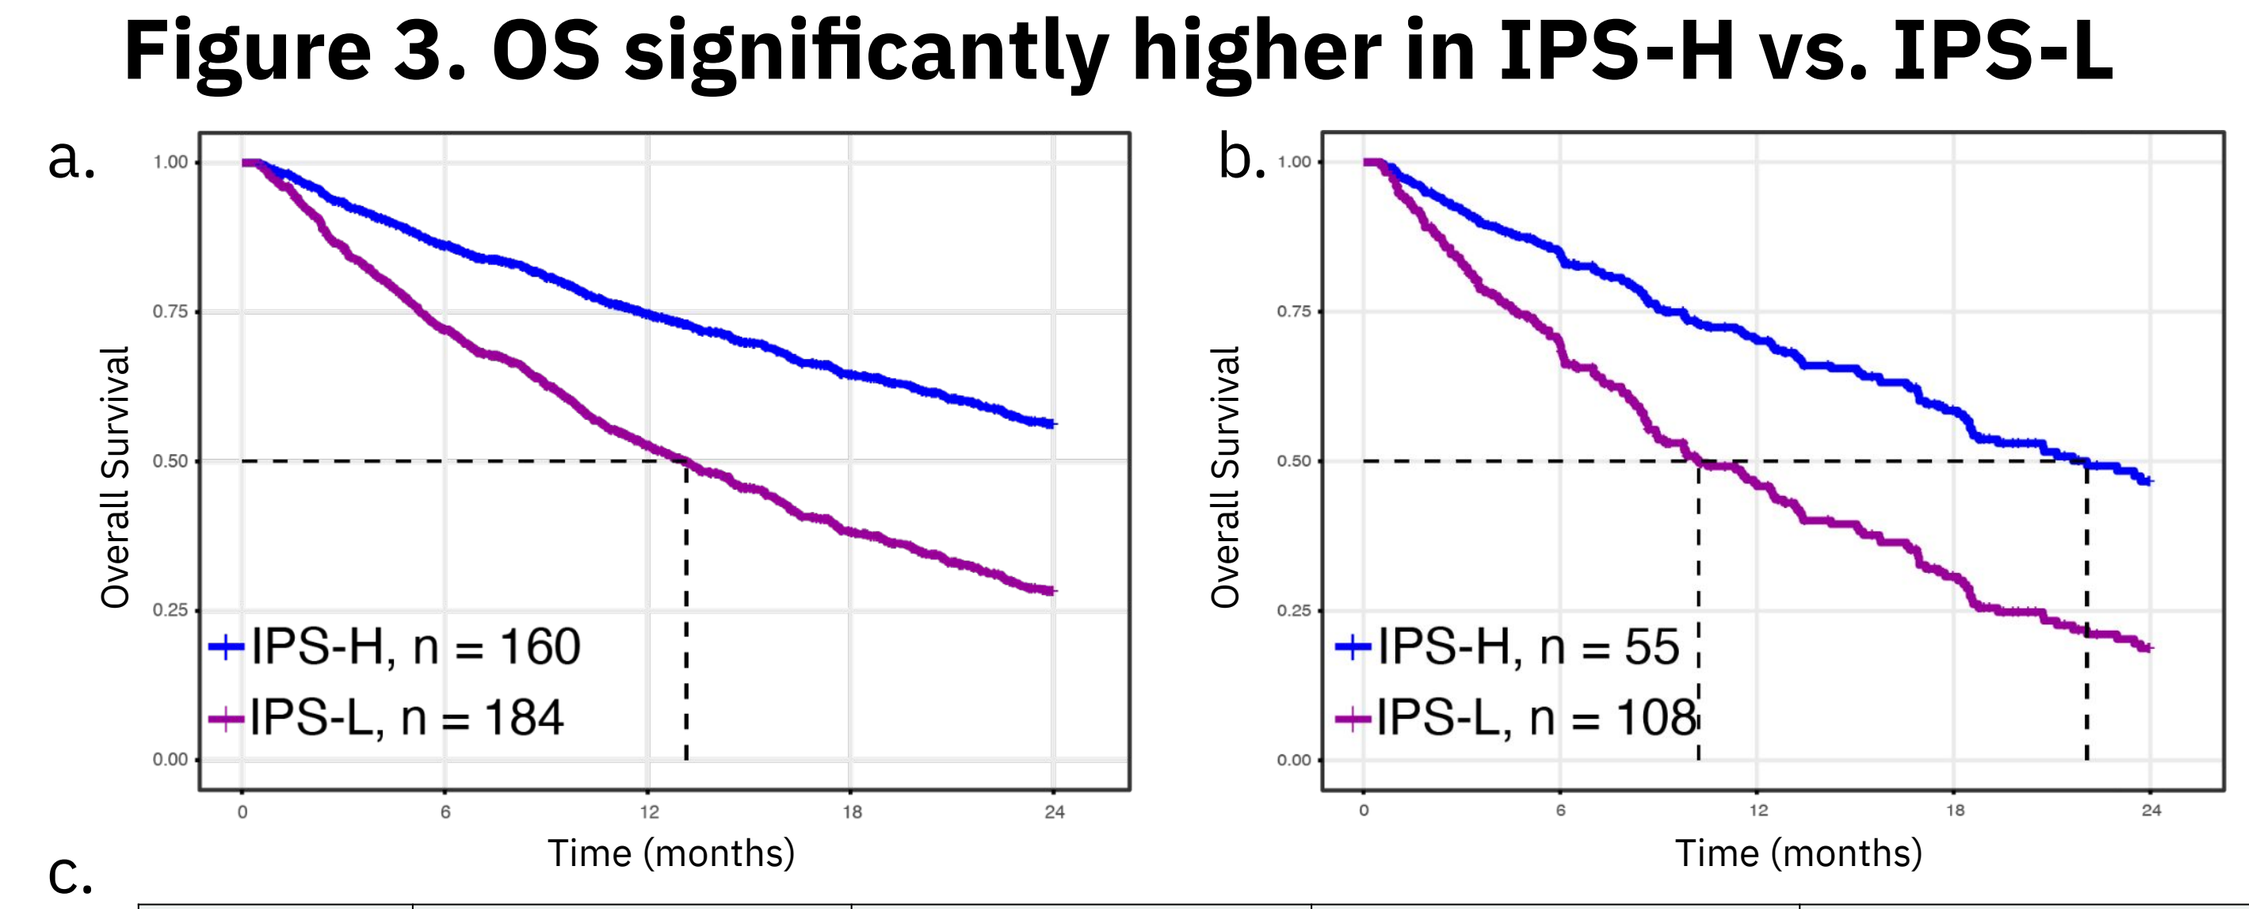
a.
Overall Survival
IPS-H, n = 160
IPS-L, n = 184
Time (months)
b.
Overall Survival
IPS-H, n = 55
IPS-L, n = 108
Time (months)

c.

	1L mono	2L mono	1L combo	2L combo
IPS-H	>24 (>24, >24)	22.1 (18.3, >24)	>24 (23.0, >24)	20.75 (16.95, >24)
IPS-L	13.1 (10.8, 15.6)	10.2 (8.56, 13.1)	12.2 (10.8, 13.8)	9.77 (8.23, 12.6)

Figure 3: The hazard ratio for IPS-H vs. IPS-L was evaluated using a CoxPH model stratified by line of therapy and controlling for treatment group (monotherapy vs. combination therapy). The HR was 0.45 (0.40, 0.52), p < 0.01. Predicted OS from the CoxPH model for **a**) 1L monotherapy and **b**) 2L monotherapy patients. Predicted survival for 1L and 2L combination therapy patients are similar to above. **c**) The median OS and 95% confidence interval for IPS-H and IPS-L groups for each line of therapy/treatment group combination.

Figure 4. HR consistent across subgroups

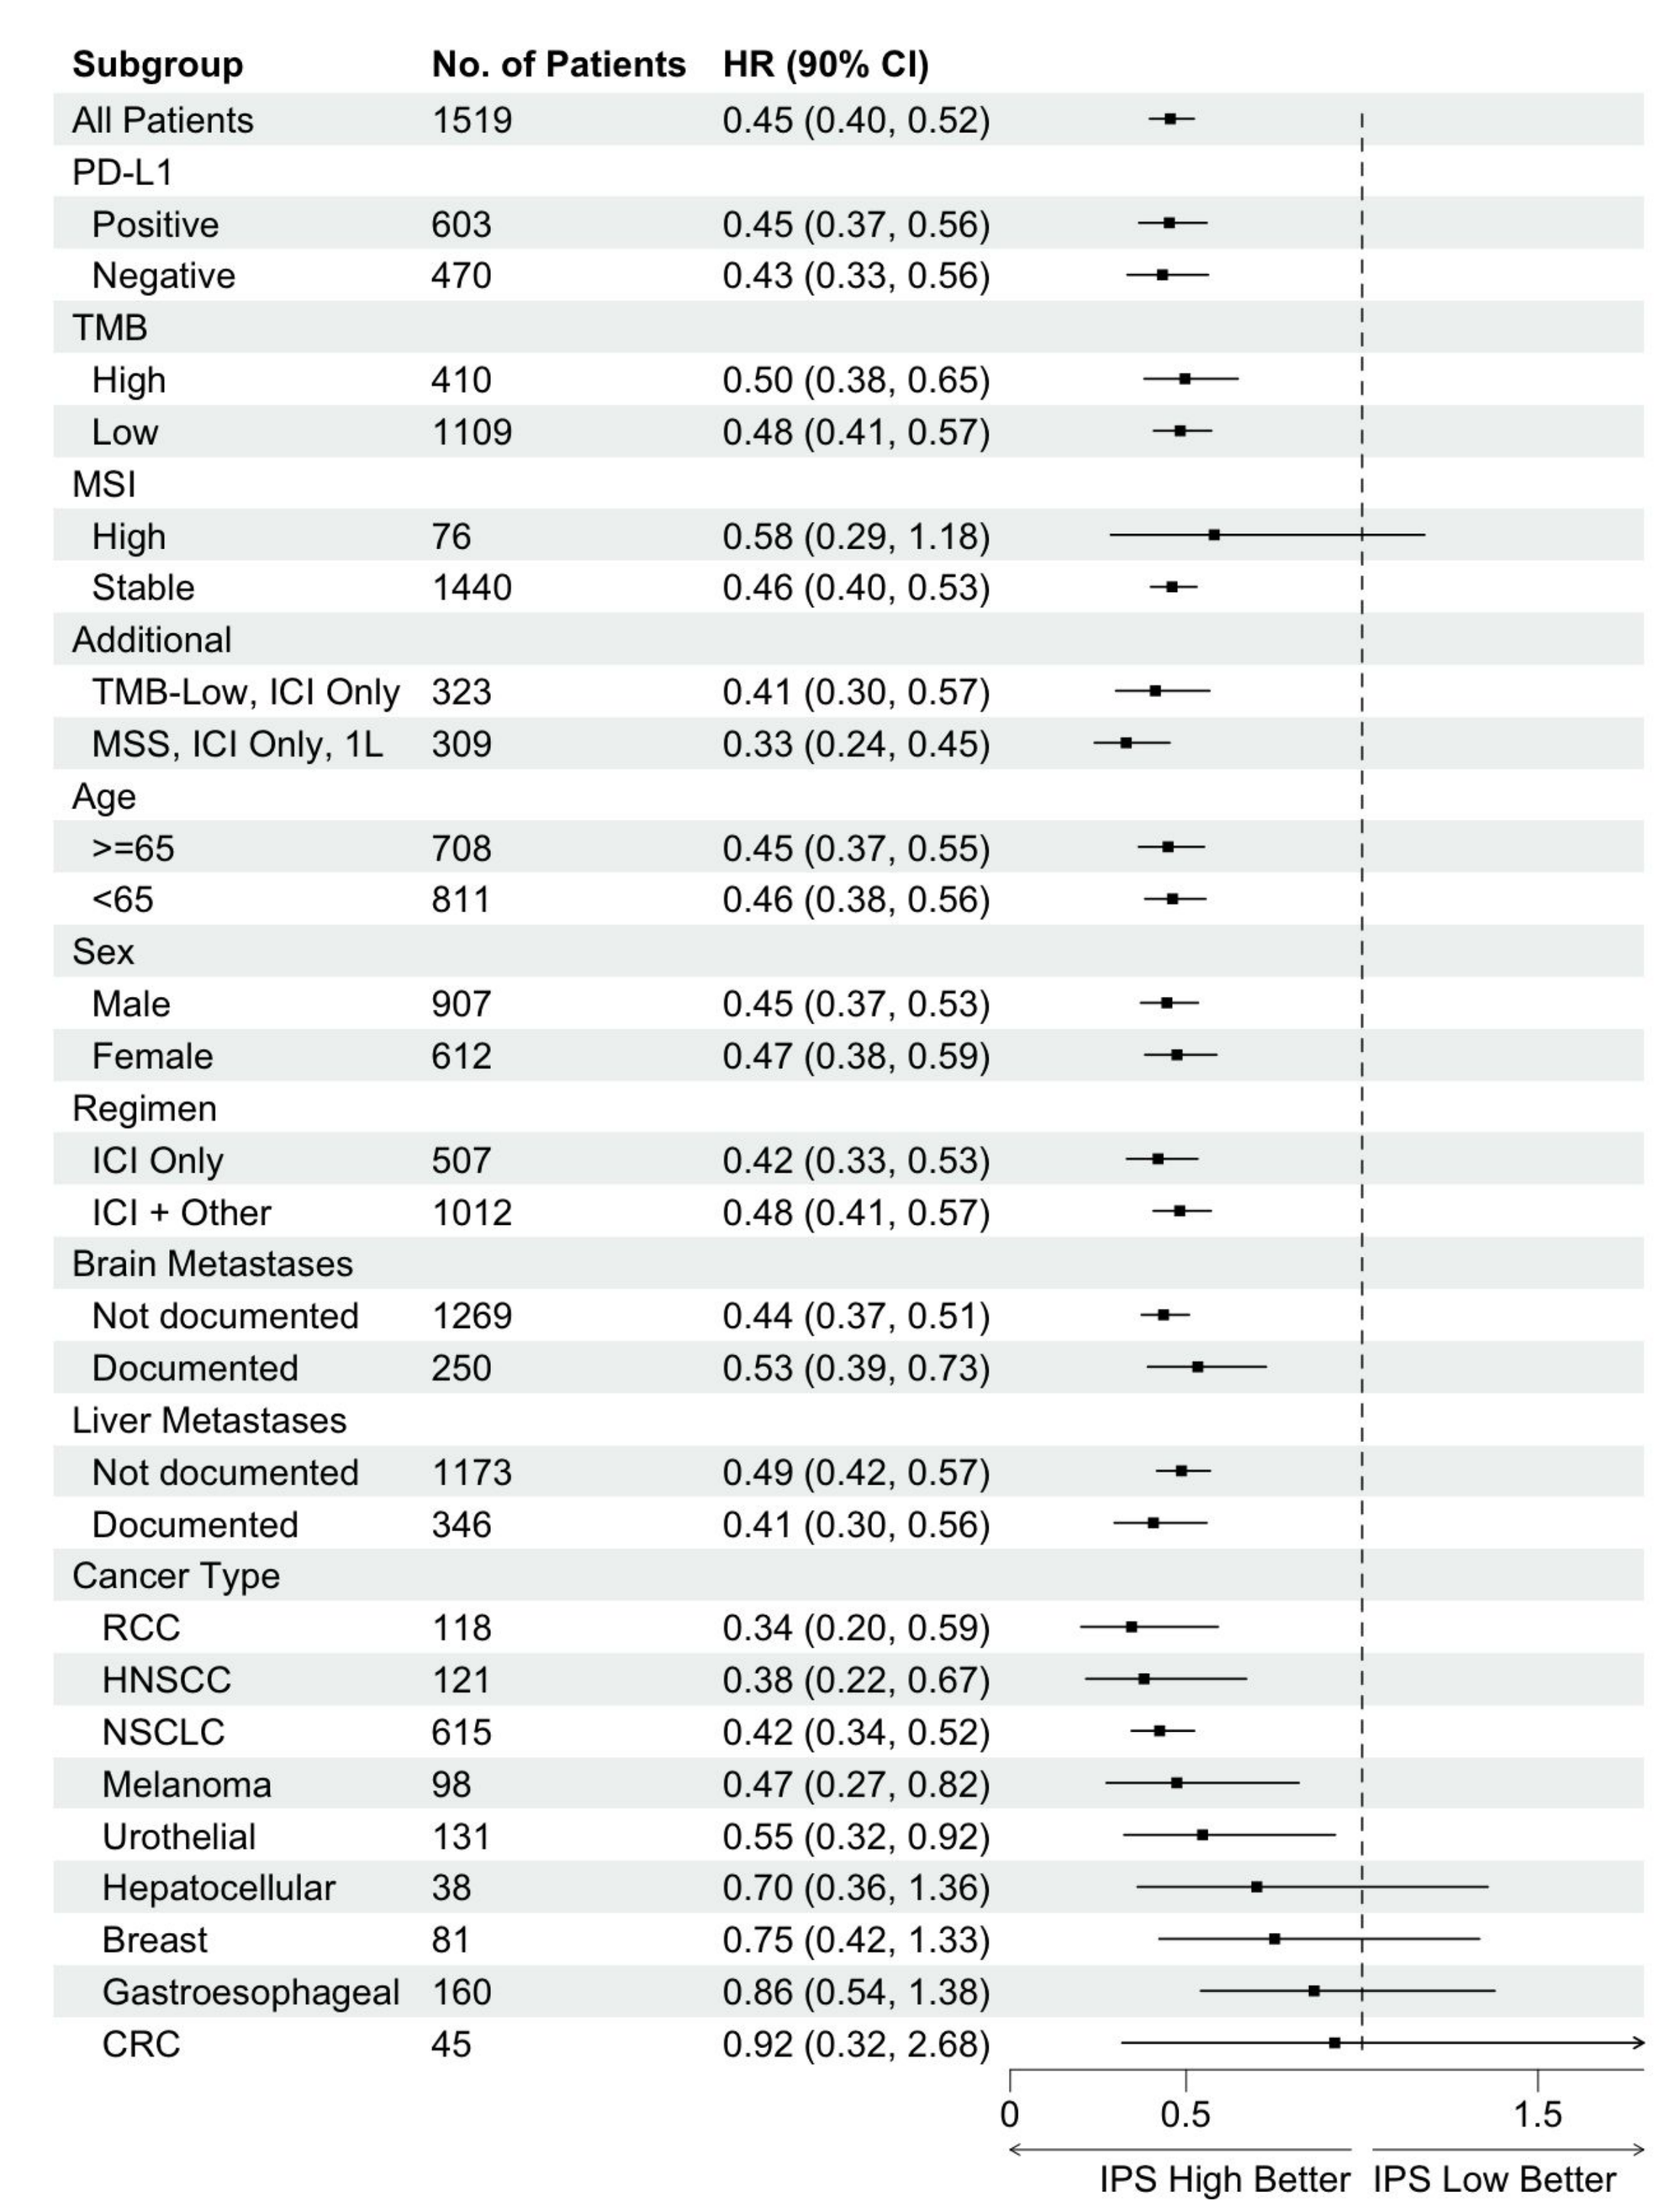
Subgroup | No. of Patients | HR (90% CI)
All Patients 1519 0.45 (0.40, 0.52)
PD-L1
Positive 603 0.45 (0.37, 0.56)
Negative 470 0.43 (0.33, 0.56)
TMB
High 410 0.50 (0.38, 0.65)
Low 1109 0.48 (0.41, 0.57)
MSI
High 76 0.58 (0.29, 1.18)
Stable 1440 0.46 (0.40, 0.53)
Additional
TMB-Low, ICI Only 323 0.41 (0.30, 0.57)
MSS, ICI Only, 1L 309 0.33 (0.24, 0.45)
Age
>=65 708 0.45 (0.37, 0.55)
<65 811 0.46 (0.38, 0.56)
Sex
Male 907 0.45 (0.37, 0.53)
Female 612 0.47 (0.38, 0.59)
Regimen
ICI Only 507 0.42 (0.33, 0.53)
ICI + Other 1012 0.48 (0.41, 0.57)
Brain Metastases
Not documented 1269 0.44 (0.37, 0.51)
Documented 250 0.53 (0.39, 0.73)
Liver Metastases
Not documented 1173 0.49 (0.42, 0.57)
Documented 346 0.41 (0.30, 0.56)
Cancer Type
RCC 118 0.34 (0.20, 0.59)
HNSCC 121 0.38 (0.22, 0.67)
NSCLC 615 0.42 (0.34, 0.52)
Melanoma 98 0.47 (0.27, 0.82)
Urothelial 131 0.55 (0.32, 0.92)
Hepatocellular 38 0.70 (0.36, 1.36)
Breast 81 0.75 (0.42, 1.33)
Gastroesophageal 160 0.86 (0.54, 1.38)
CRC 45 0.92 (0.32, 2.68)
0 0.5 1.5
IPS High Better IPS Low Better

Figure 4: Forest plot showing IPS-H vs. IPS-L hazard ratios and confidence intervals across demographics and clinically relevant subgroups. Subgroups may have <1519 patients due to availability of data.

Figure 5. IPS has significant prognostic utility beyond TMB, PD-L1, and MSI

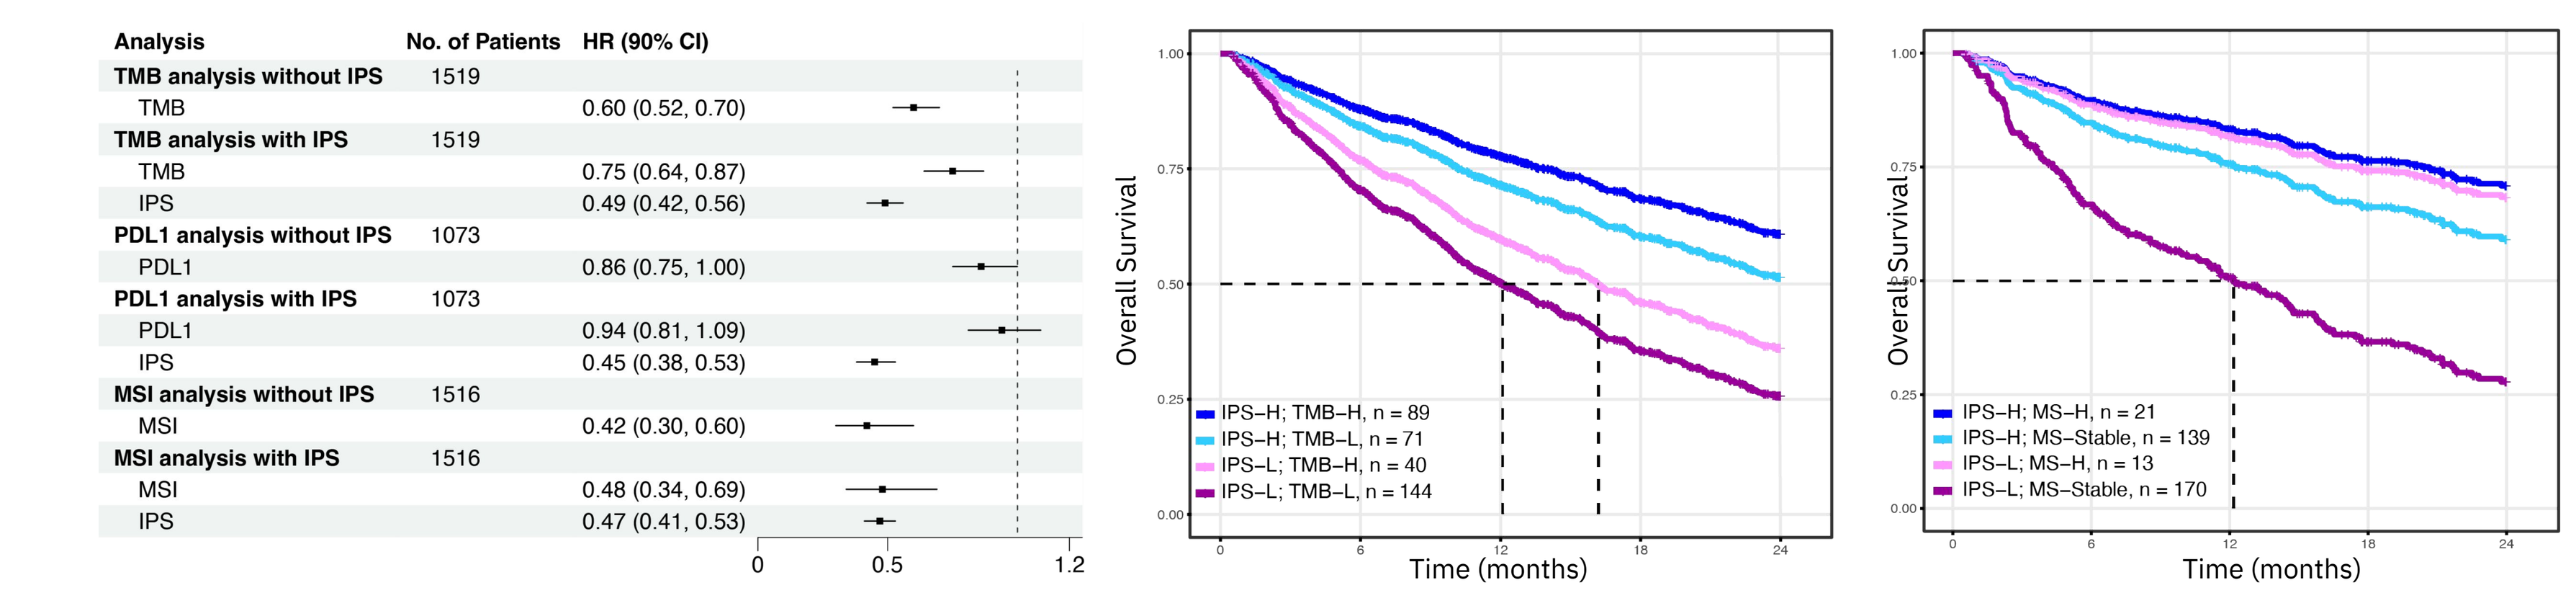
a. Hazard Ratios
Analysis | No. of Patients | HR (90% CI)
TMB analysis without IPS 1519
TMB 0.60 (0.52, 0.70)
TMB analysis with IPS 1519
TMB 0.75 (0.64, 0.87)
IPS 0.49 (0.42, 0.56)
PDL1 analysis without IPS 1073
PDL1 0.86 (0.75, 1.00)
PDL1 analysis with IPS 1073
PDL1 0.94 (0.81, 1.09)
IPS 0.45 (0.38, 0.53)
MSI analysis without IPS 1516
MSI 0.42 (0.30, 0.60)
MSI analysis with IPS 1516
MSI 0.48 (0.34, 0.69)
IPS 0.47 (0.41, 0.53)
0 0.5 1.2
b. OS: TMB x IPS
Overall Survival
IPS–H; TMB–H, n = 89
IPS–H; TMB–L, n = 71
IPS–L; TMB–H, n = 40
IPS–L; TMB–L, n = 144
Time (months)
c. OS: MSI x IPS
Overall Survival
IPS–H; MS–H, n = 21
IPS–H; MS–Stable, n = 139
IPS–L; MS–H, n = 13
IPS–L; MS–Stable, n = 170
Time (months)

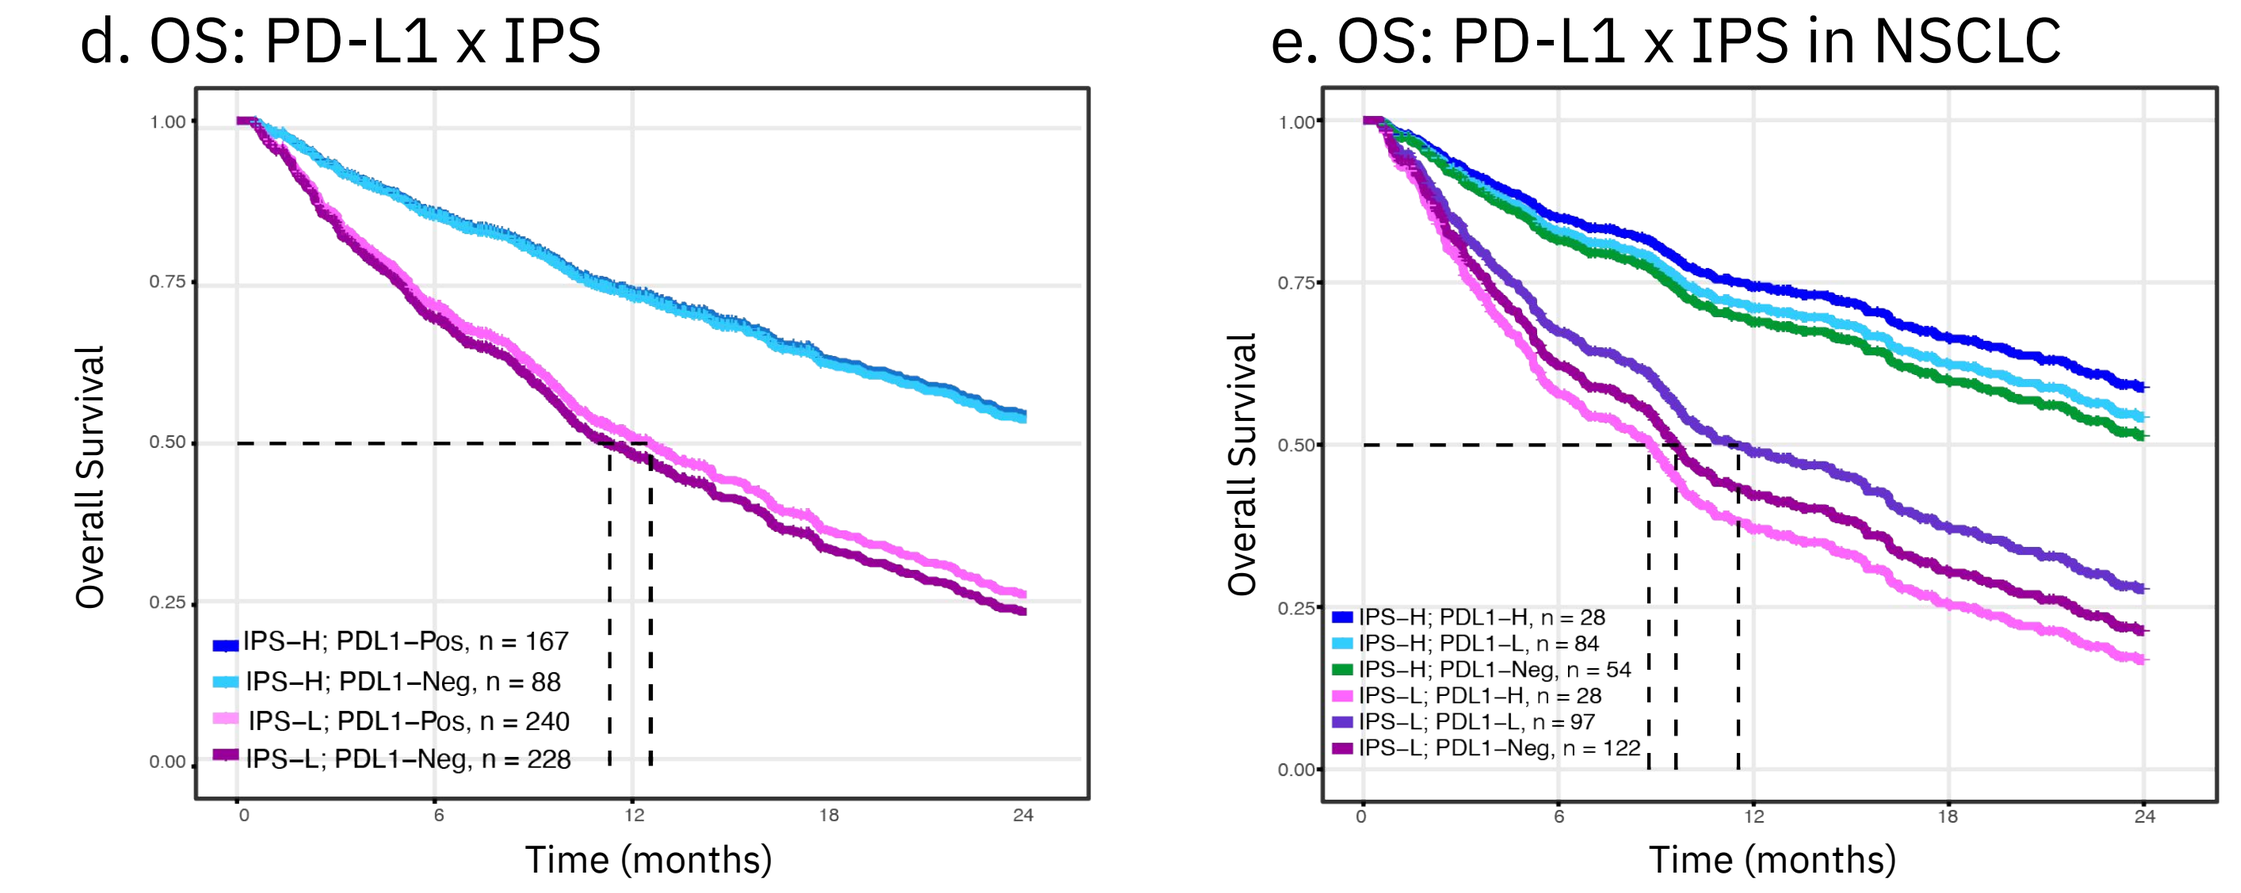
d. OS: PD-L1 x IPS
Overall Survival
IPS–H; PDL1–Pos, n = 167
IPS–H; PDL1–Neg, n = 88
IPS–L; PDL1–Pos, n = 240
IPS–L; PDL1–Neg, n = 228
Time (months)
e. OS: PD-L1 x IPS in NSCLC
Overall Survival
IPS–H; PDL1–H, n = 28
IPS–H; PDL1–L, n = 84
IPS–H; PDL1–Neg, n = 54
IPS–L; PDL1–H, n = 28
IPS–L; PDL1–L, n = 97
IPS–L; PDL1–Neg, n = 122
Time (months)

F. Hazard Ratios

Comparisons	HR
B. IPS-H/TMB-L to IPS-L/TMB-L	0.49 (0.41, 0.58)
C. IPS-H/MSS to IPS-L/MSS	0.41 (0.32, 0.53)
D. IPS-H/PDL1- to IPS-L/PDL1-	0.43 (0.33, 0.56)
E. IPS-H/PDL1- to IPS-L/PDL1- (within NSCLC)	0.43 (0.30, 0.63)

Figure 5: a) Forest plot showing univariate (UV) HRs for TMB, PD-L1, MSI and multivariate (MV) HRs that include IPS. A likelihood ratio test between the UV and MV models was significant ($p<0.01$) for all three biomarkers, indicating that IPS has significant prognostic utility beyond TMB, MSI, and PD-L1. Plots **b-e** show predicted OS from a model stratified by line of therapy and fit on IPS, treatment group, and the MV model with the listed biomarker: **b**) TMB pan-cancer, **c)** MSI pan-cancer, **d**) PD-L1 pan-cancer and **e)** PD-L1 in NSCLC patients. The predicted OS curves represent patients treated with monotherapy in 1L for TMB and MSI **(b-c)**, and combination therapy in 1L for PD-L1 and NSCLC **(d-e). f**) HR and 90% CI for the most relevant curves shown in the predicted OS plots in **(b-e).**

Figure 6. Predictive utility for IPS

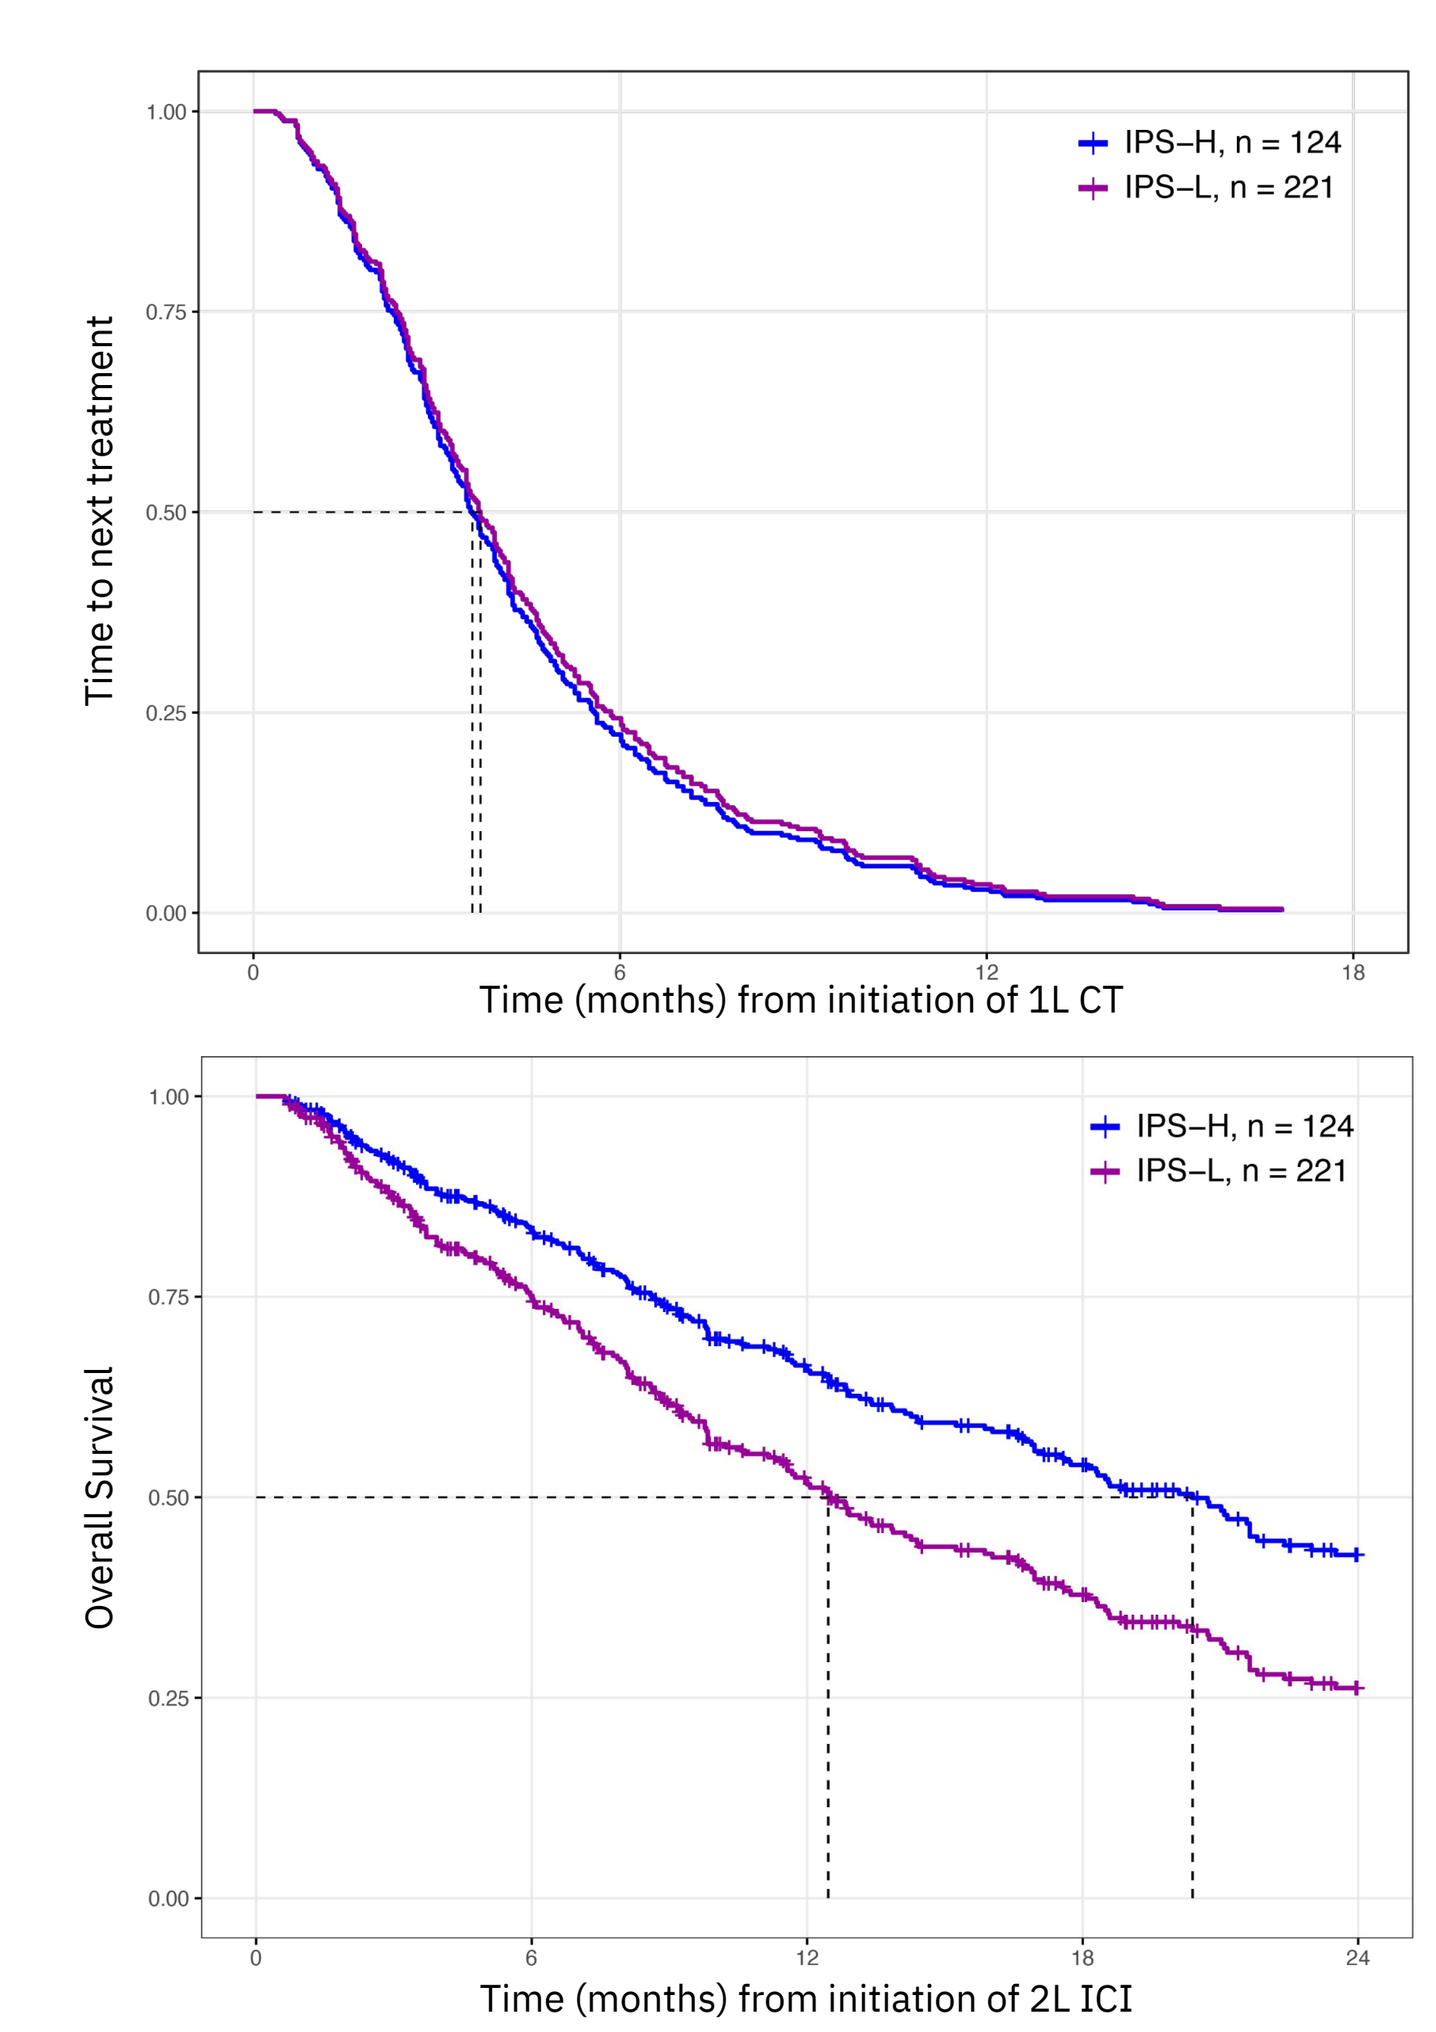
IPS–H, n = 124
IPS–L, n = 221
Time to next treatment
Time (months) from initiation of 1L CT
IPS–H, n = 124
IPS–L, n = 221
Overall Survival
Time (months) from initiation of 2L ICI

Figure 6: An exploratory analysis of the predictive utility of the IPS was performed by combining the training and validation cohorts of patients who received chemotherapy (CT) as first line treatment and ICI as second line treatment. Patients served as their own control in this analysis, and outcomes were evaluated for two lines of therapy: time to next treatment (TTNT) on CT and OS on ICI. A conditional model for recurrent events was fit. **Top**: Predicted TTNT for 1L CT with no significant effect for IPS (HR = 1.06 (0.85, 1.33)). **Bottom**: Predicted OS for 2L ICI shows that IPS does have a significant effect (HR = 0.63 (0.46, 0.86)). Interaction test $p < 0.01$, indicating that the HR in 2L ICI is significantly different from HR in 1L CT.

Figure 7. IPS prevalence in database

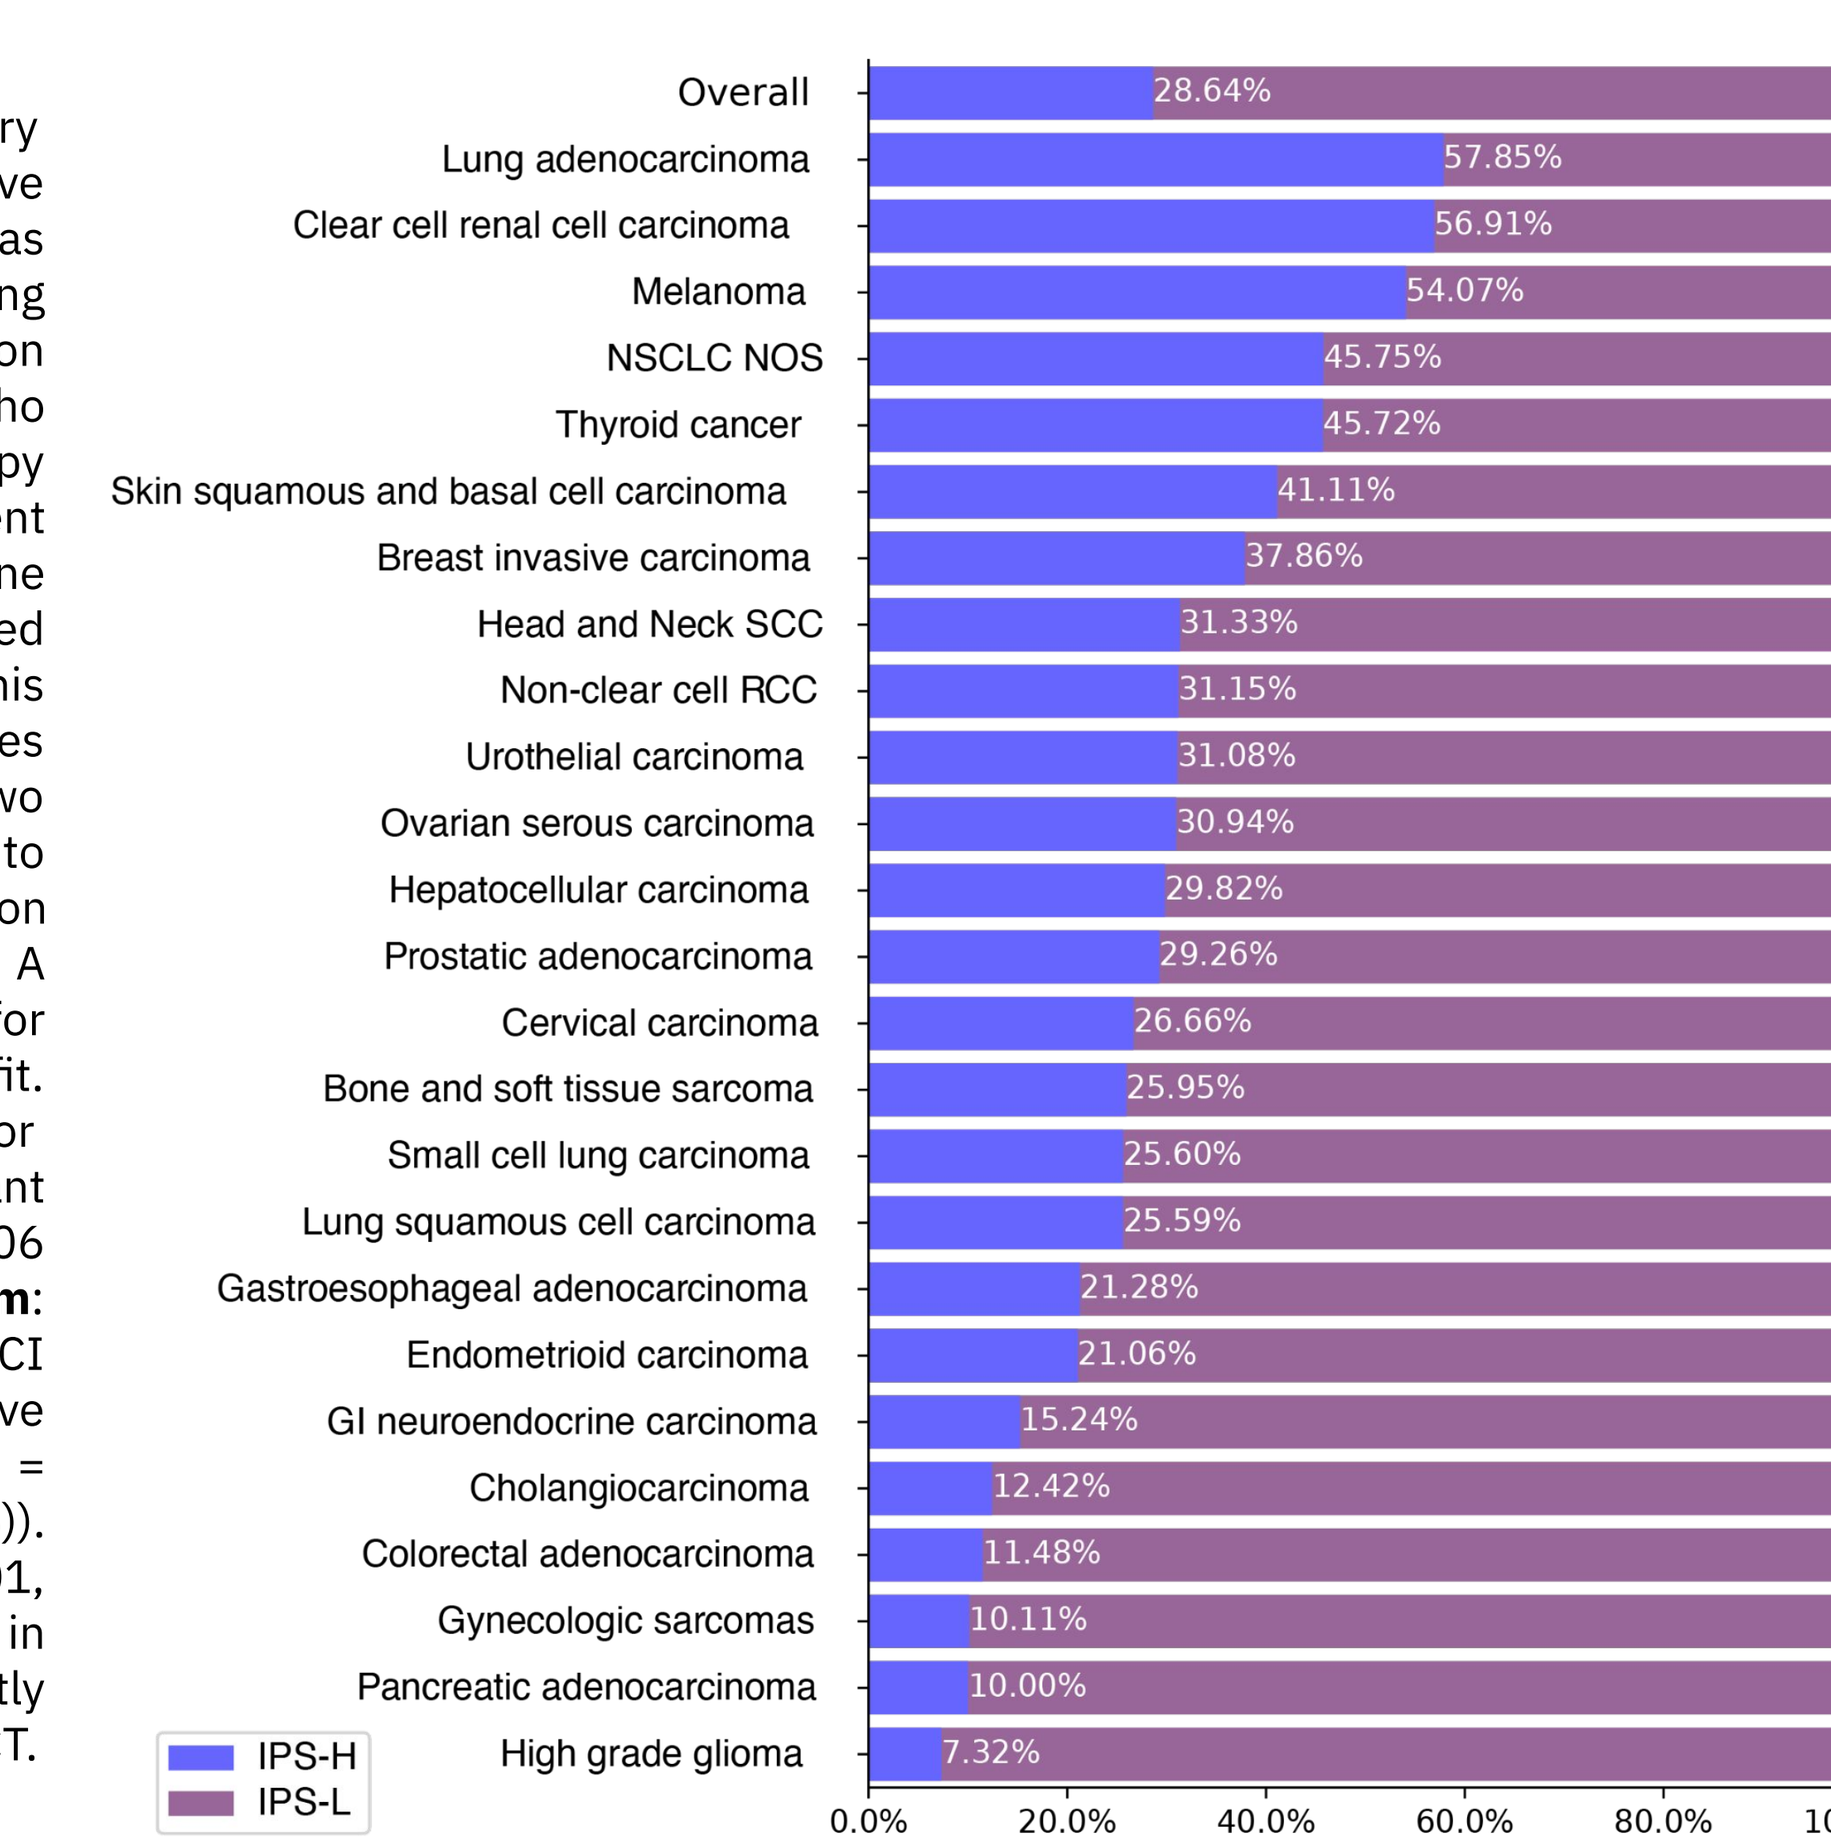
Overall 28.64%
Lung adenocarcinoma 57.85%
Clear cell renal cell carcinoma 56.91%
Melanoma 54.07%
NSCLC NOS 45.75%
Thyroid cancer 45.72%
Skin squamous and basal cell carcinoma 41.11%
Breast invasive carcinoma 37.86%
Head and Neck SCC 31.33%
Non-clear cell RCC 31.15%
Urothelial carcinoma 31.08%
Ovarian serous carcinoma 30.94%
Hepatocellular carcinoma 29.82%
Prostatic adenocarcinoma 29.26%
Cervical carcinoma 26.66%
Bone and soft tissue sarcoma 25.95%
Small cell lung carcinoma 25.60%
Lung squamous cell carcinoma 25.59%
Gastroesophageal adenocarcinoma 21.28%
Endometrioid carcinoma 21.06%
GI neuroendocrine carcinoma 15.24%
Cholangiocarcinoma 12.42%
Colorectal adenocarcinoma 11.48%
Gynecologic sarcomas 10.11%
Pancreatic adenocarcinoma 10.00%
High grade glioma 7.32%
IPS-H
IPS-L
0.0% 20.0% 40.0% 60.0% 80.0% 100.0%

Figure 7: Prevalence plot showing the percentage of IPS high patients in a large, representative cohort of patients from the Tempus multimodal database.

ACKNOWLEDGMENTS

We thank Dana DeSantis from the Tempus Science Communications team for poster development.

Correspondence: alia.zander@tempus.com