Lipid Metabolic Gene Expression is Associated with Decreased Overall Survival and Immunogenicity in KRAS-STK11 NSCLC

Joshua Pothen[1], Jen Godden[2], Denise Shieh[2], Stamatina Fragkogianni[2], Jacob Mercer[2], Melissa Conrad Stoppler[2], Daniel Principe[3], Ryan Huu-Tuan Nguyen[1], Kamya Sankar[4], Koosha Paydary[5], Mary J. Fidler[5], Frank Weinberg[1]

[1]University of Illinois Chicago, Chicago, IL; [2]Tempus AI, Inc., Chicago, IL; [3]University of Wisconsin, Madison, WI; [4]Cedars-Sinai Medical Center, Los Angeles, CA; [5]Rush University, Chicago, IL

Abstract Presentation #8534

INTRODUCTION

- About 25% of patients with LAC and ~5% of patients with SCC will harbor *KRAS* mutations.
- It is appreciated that *KRAS* co-mutational status with *TP53, STK11, LRP1B* etc is prognostic and predictive for response to immunotherapy, but there is a lack of understanding between the discordant outcomes and responses to treatments.
- In a population of primarily underrepresented individuals with *KRAS* mutant NSCLC, those with *STK11* and *LRP1B* co-mutations had poor clinical outcomes.
- Additional RNA analysis demonstrated *KRAS-STK11* mutant tumors had decreased expression levels of genes important in lipid/cholesterol metabolism when compared to *KRAS-TP53* mutant tumors, which is also associated with worse clinical outcomes.
- This study aims to validate these findings in a larger cohort, to ultimately identify a novel pathway that may improve outcomes to standard of care therapy.

METHODS

Cohort Selection

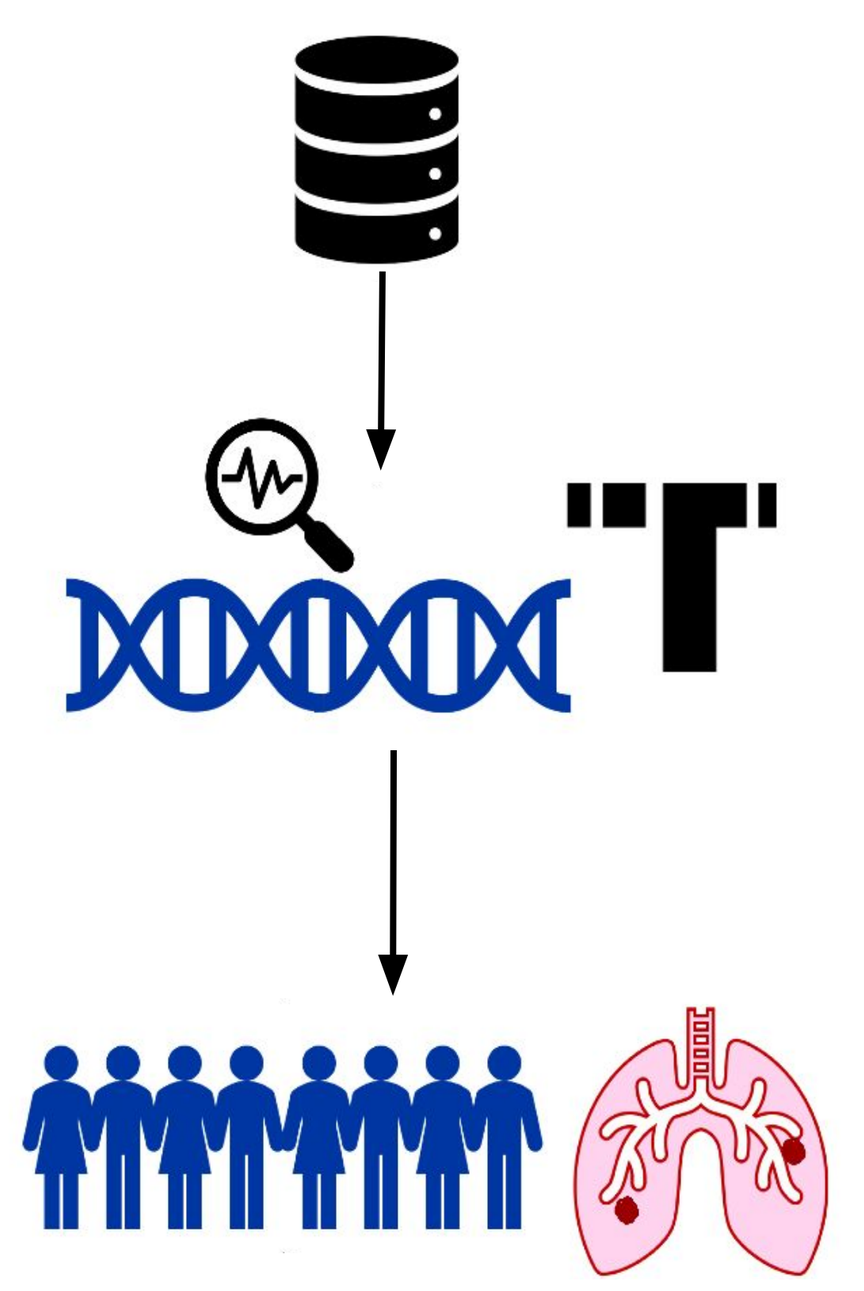

De-identified records of 2,187 patients diagnosed with *KRAS* G12C alt NSCLC

Sequencing via the Tempus xT/xR assay

Co-alterations in *TP53* (44%), *STK11* (17%), or *LRP1B* (4%) were selected. The groups are mutually exclusive.

- Single-sample GSEA (ssGSEA) based on 775 lipid metabolic genes (LMG) was used to calculate enrichment scores (LMG ES) for each pt.
- Pts were dichotomized into low vs. high groups based on their median LMG ES.
- Immune cell infiltration predicted from gene expression patterns, TMB, and PD-L1 from IHC was evaluated.
- Risk-set adjusted rwOS was calculated from sample collection date to death from any cause.
- Hazard ratios (HR) were calculated using Cox proportional hazards model, and p-values were calculated using the Wald test.

SUMMARY

- Percent immune infiltration was lower and median lipid metabolism gene enrichment score was higher in patients with *KRAS-STK11* tumors.
- Lipid gene expression and tumor immune cell infiltration were associated, suggesting that lipid metabolism may regulate tumor immunogenicity.
- Low expression of lipid metabolic genes in *KRAS-STK11* tumors is a prognostic marker associated with decreased OS.
- These data suggest that lipid metabolic genes should be further explored as potential therapeutic targets for patients with NSCLC and *KRAS-STK11* alt.

RESULTS

Table 1. Cohort Demographics

Characteristic	Overall, N = 2,187[1]	KRAS-G12C WT, N = 767[1]	KRAS-G12C TP53, N = 970[1]	KRAS-G12C STK11, N = 364[1]	KRAS-G12C LRP1B, N = 86[1]	p-value[2]
Age at Primary Diagnosis						<0.001
Median (IQR)	68 (63, 75)	70 (64, 77)	68 (62, 75)	67 (61, 73)	70 (65, 78)	
Range	27, 89	42, 88	37, 89	27, 87	45, 87	
Unknown	39	19	15	4	1	
Sex						0.2
Female	1,272 (58%)	461 (60%)	552 (57%)	203 (56%)	56 (65%)	
Male	915 (42%)	306 (40%)	418 (43%)	161 (44%)	30 (35%)	
Race						0.6
White	1,239 (84%)	428 (84%)	550 (83%)	216 (86%)	45 (82%)	
Black or African American	141 (9.6%)	45 (8.8%)	71 (11%)	19 (7.6%)	6 (11%)	
Other Race	71 (4.8%)	31 (6.1%)	27 (4.1%)	11 (4.4%)	2 (3.6%)	
Asian	23 (1.6%)	6 (1.2%)	11 (1.7%)	4 (1.6%)	2 (3.6%)	
Unknown	713	257	311	114	31	
Ethnicity						0.008
Not Hispanic or Latino	976 (96%)	339 (93%)	442 (98%)	163 (97%)	32 (97%)	
Hispanic or Latino	42 (4.1%)	26 (7.1%)	10 (2.2%)	5 (3.0%)	1 (3.0%)	
Unknown	1,169	402	518	196	53	
Smoking Status						0.4
Ex-smoker	857 (57%)	297 (59%)	374 (54%)	154 (59%)	32 (62%)	
Current-smoker	621 (41%)	195 (39%)	305 (44%)	101 (39%)	20 (38%)	
Never-smoker	33 (2.2%)	14 (2.8%)	13 (1.9%)	6 (2.3%)	0 (0%)	
Unknown	676	261	278	103	34	

[1] n (%)
[2] Kruskal-Wallis rank sum test; Pearson's Chi-squared test; Fisher's Exact Test for Count Data with simulated p-value (based on 2000 replicates)

Table 1: Among pts with *KRAS* G12C alt, the median age was 68, 58% were female, and 84% were White.

Table 2. Immune Biomarker Status Across Co-Alterations

Characteristic	Overall, N = 2,187[1]	*KRAS*-G12C WT, N = 767[1]	*KRAS*-G12C *TP53*, N = 970[1]	*KRAS*-G12C *STK11*, N = 364[1]	*KRAS*-G12C *LRP1B*, N = 86[1]	p-value[2]
TMB (Continuous)						<0.001
Median (IQR)	7.4 (4.7, 10.1)	6.3 (4.0, 9.5)	8.4 (5.3, 12.1)	5.8 (4.2, 7.9)	7.4 (4.8, 9.9)	
Range	0.0, 58.9	0.0, 34.7	0.0, 58.9	0.0, 17.7	0.0, 32.6	
TMB (Categorical)						<0.001
Low	1,557 (71%)	601 (78%)	572 (59%)	320 (88%)	64 (74%)	
High	630 (29%)	166 (22%)	398 (41%)	44 (12%)	22 (26%)	
Neoantigen Tumor Burden						<0.001
Median (IQR)	11 (7, 16)	9 (6, 14)	12 (8, 18)	9 (6, 12)	11 (7, 16)	
Range	1, 60	1, 60	1, 58	1, 31	1, 49	
Unknown	46	22	18	3	3	
PDL1 Result from Internal IHC						<0.001
Positive	1,059 (69%)	345 (65%)	596 (86%)	75 (30%)	43 (74%)	
Negative	473 (31%)	187 (35%)	100 (14%)	171 (70%)	15 (26%)	
Unknown	655	235	274	118	28	
RNA expression levels of PD-L1 (CD274)						<0.001
Median (IQR)	3.09 (2.36, 3.96)	2.92 (2.32, 3.72)	3.55 (2.88, 4.39)	2.19 (1.72, 2.69)	3.27 (2.55, 3.78)	
Range	-0.33, 7.31	-0.33, 7.31	-0.33, 6.97	0.34, 5.90	0.62, 7.19	
Unknown	3	2	1	0	0	

[1] n (%)
[2] Kruskal-Wallis rank sum test; Pearson's Chi-squared test

Table 2: *KRAS* G12C/*STK11* alt patients had the lowest TMB, neoantigen burden, and PD-L1 positivity compared to other cohorts (p < 0.001 for all).

Figure 1. Comparison of Immune Infiltration Levels Between Co-Alterations

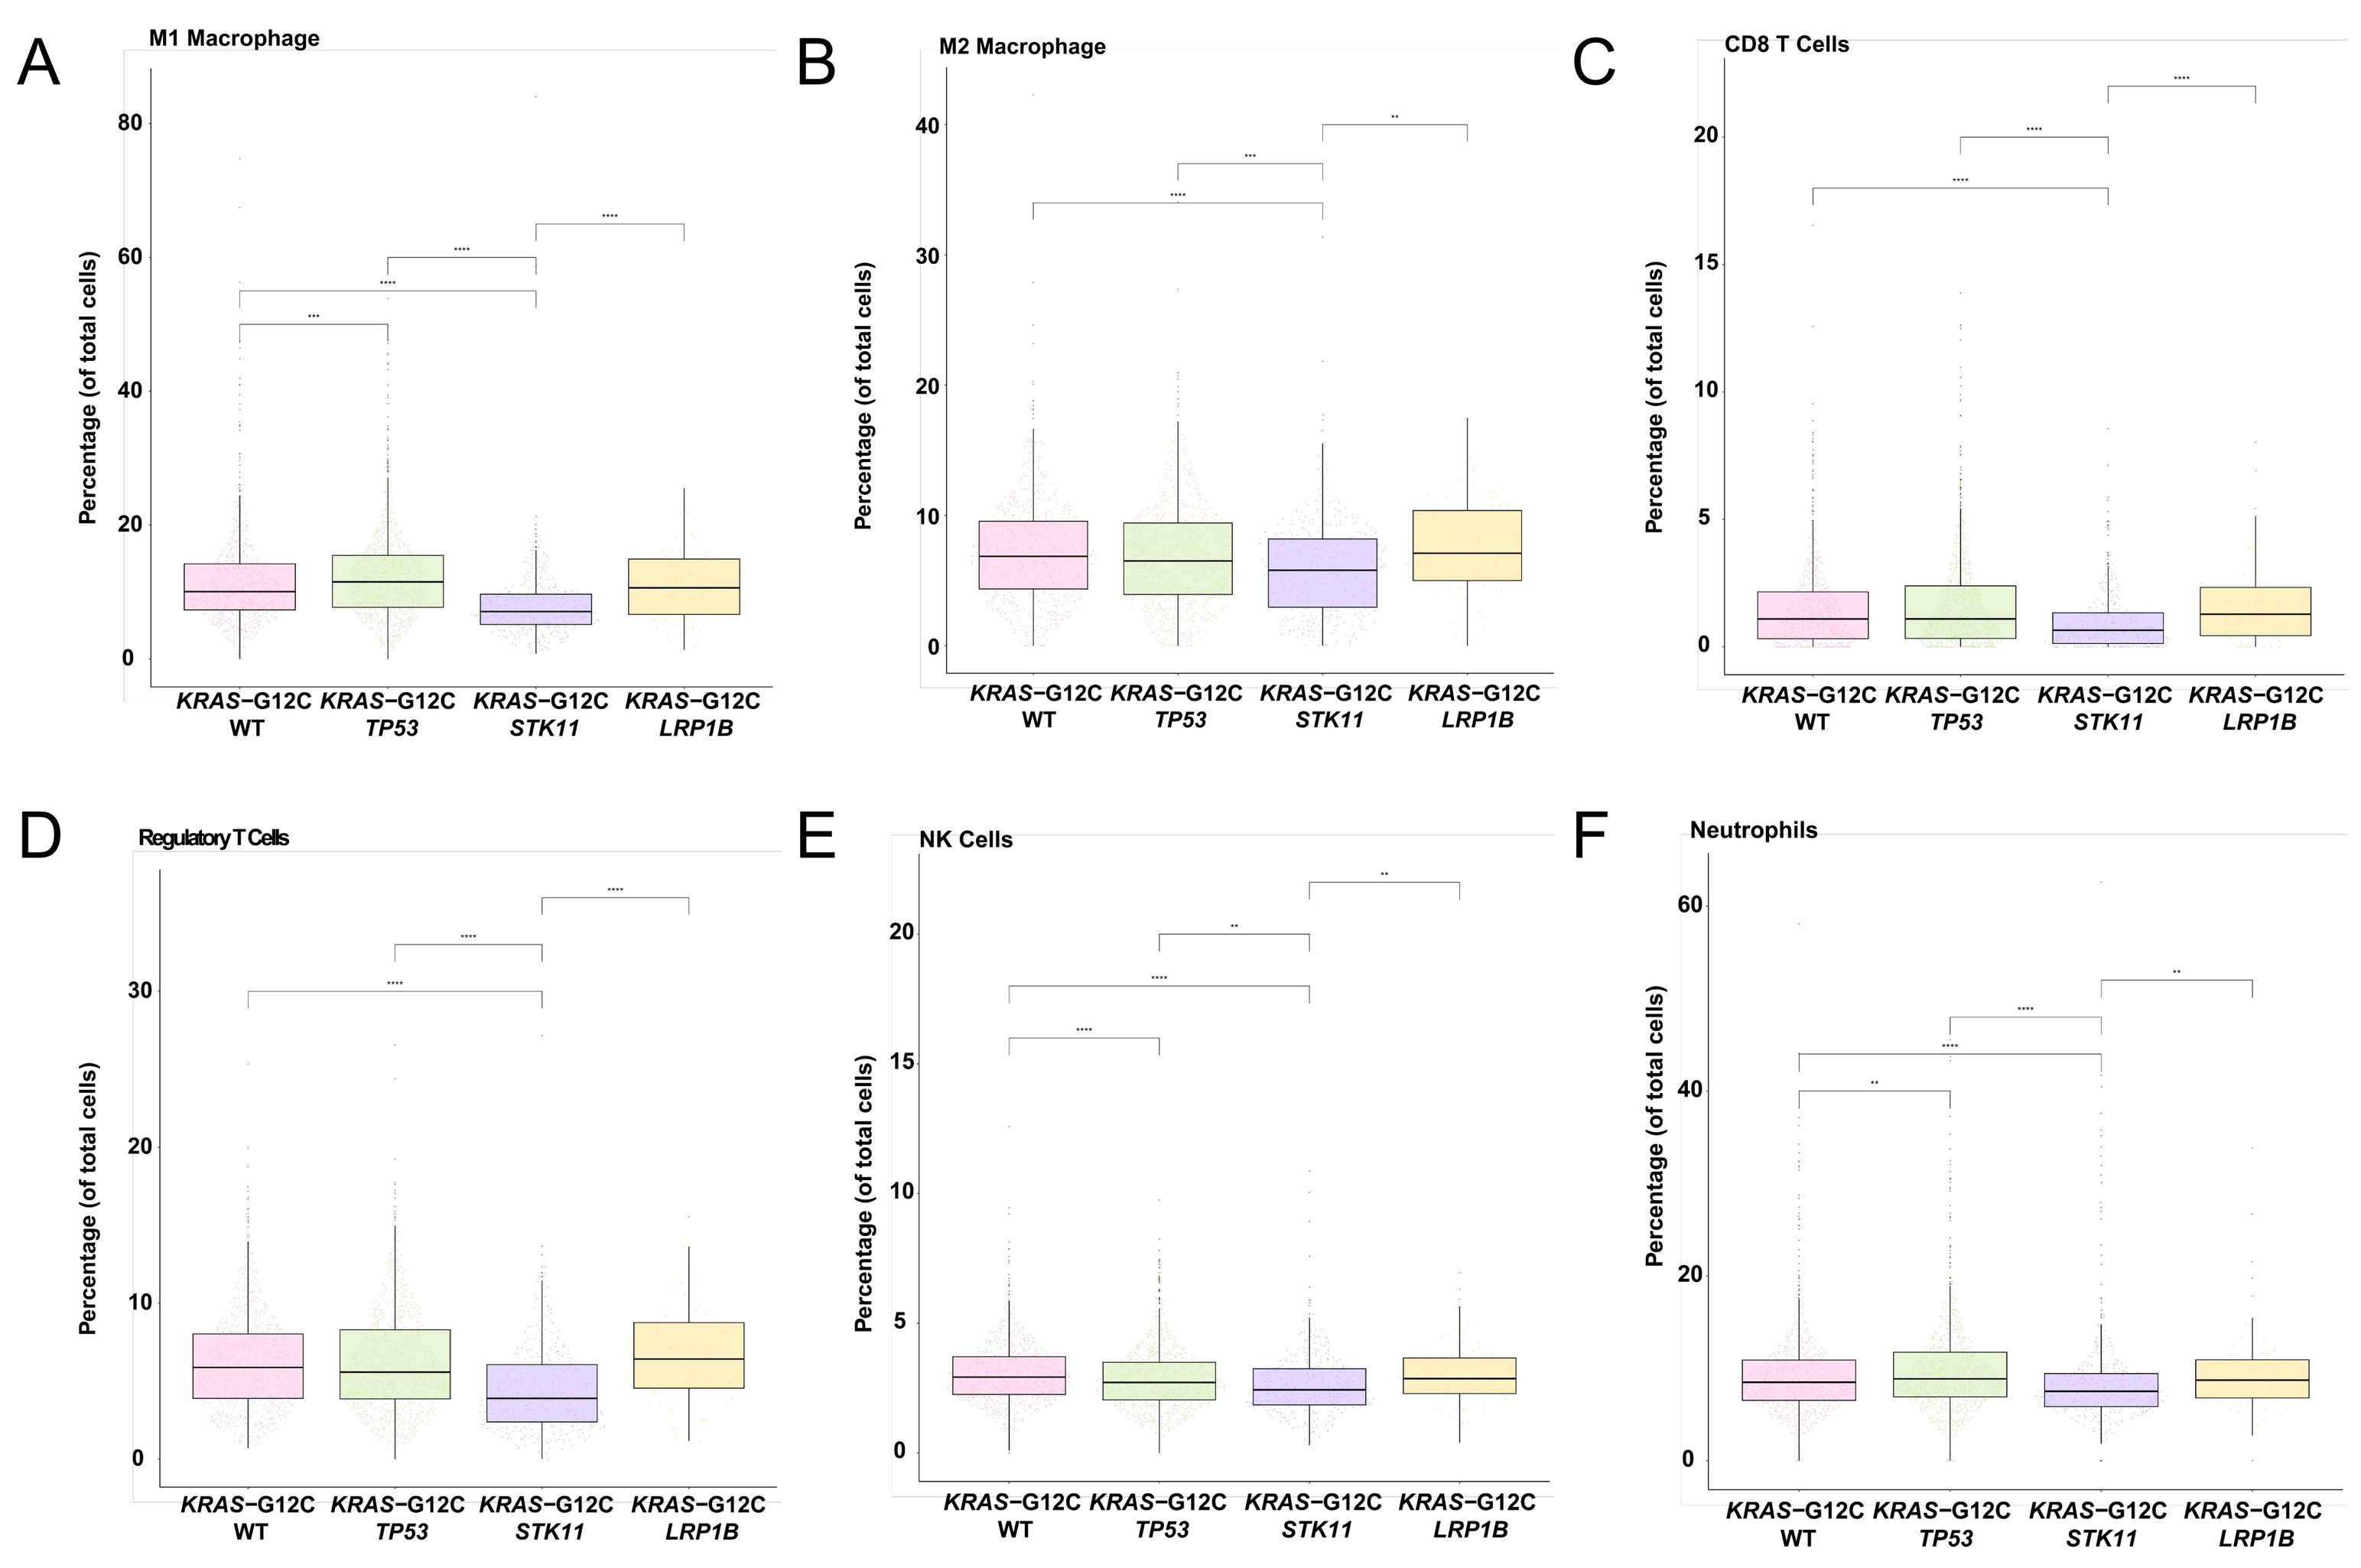
A
M1 Macrophage
B
M2 Macrophage
C
CD8 T Cells
D
Regulatory T Cells
E
NK Cells
F
Neutrophils
Percentage (of total cells)
KRAS−G12C WT
KRAS−G12C TP53
KRAS−G12C STK11
KRAS−G12C LRP1B

Characteristic	Overall N = 1,384	KRAS-G12C WT N = 533	KRAS-G12C TP53 N = 564	KRAS-G12C STK11 N = 231	KRAS-G12C LRP1B N = 56	p-value[1]
Percent of B cells						0.8
Median (IQR)	5.46 (3.88, 8.17)	5.36 (3.99, 8.12)	5.43 (3.77, 8.27)	5.54 (3.89, 8.24)	5.74 (4.29, 7.82)	
Percent of dendritic cells						0.5
Median (IQR)	0.00 (0.00, 0.00)	0.00 (0.00, 0.00)	0.00 (0.00, 0.00)	0.00 (0.00, 0.00)	0.00 (0.00, 0.00)	
Percent of M1 macrophages						<0.001
Median (IQR)	10.53 (7.36, 14.52)	10.31 (7.65, 14.27)	12.02 (8.50, 15.87)	7.52 (5.58, 9.91)	12.06 (8.63, 15.11)	
Percent of M2 macrophages						<0.001
Median (IQR)	7.68 (5.45, 10.03)	7.57 (5.44, 10.15)	7.99 (5.68, 10.27)	7.10 (4.89, 8.73)	8.46 (5.94, 11.35)	
Percent of monocytes						0.4
Median (IQR)	0.00 (0.00, 0.00)	0.00 (0.00, 0.00)	0.00 (0.00, 0.00)	0.00 (0.00, 0.00)	0.00 (0.00, 0.00)	
Percent of NK cells						<0.001
Median (IQR)	2.99 (2.37, 3.76)	3.14 (2.52, 3.84)	2.94 (2.34, 3.74)	2.74 (2.04, 3.58)	3.21 (2.62, 3.87)	
Percent of neutrophils						<0.001
Median (IQR)	8.30 (6.51, 10.46)	8.30 (6.56, 10.40)	8.76 (6.95, 11.27)	6.99 (5.68, 8.89)	8.99 (7.10, 10.62)	
Percent of other cells						<0.001
Median (IQR)	52.44 (43.69, 61.21)	52.14 (43.52, 60.53)	49.61 (41.01, 58.70)	60.25 (53.24, 66.47)	50.23 (41.06, 58.67)	
Percent of CD4 T cells						0.004
Median (IQR)	0.00 (0.00, 0.60)	0.00 (0.00, 1.06)	0.00 (0.00, 0.00)	0.00 (0.00, 1.15)	0.00 (0.00, 0.11)	
Percent of CD8 T cells						<0.001
Median (IQR)	1.19 (0.44, 2.30)	1.16 (0.42, 2.20)	1.41 (0.57, 2.59)	0.87 (0.35, 1.47)	1.52 (0.75, 2.39)	
Percent of Tregs						<0.001
Median (IQR)	6.49 (4.56, 8.61)	6.62 (4.78, 8.40)	6.77 (4.77, 9.27)	5.03 (3.62, 6.97)	7.15 (5.01, 9.29)	

[1] Kruskal-Wallis rank sum test

Figure 1: The proportion of total immune cells, M1, M2, NK cells, neutrophils, CD8 T cells and regulatory T cells was lowest in tumors with KRAS G12C/STK11 alt (global p < 0.001 for all).
: q ≤0.01; *: q ≤0.001; ****: q ≤0.0001.

Figure 2. Comparison of Lipid Metabolism Gene Enrichment Scores Between Co-Alterations

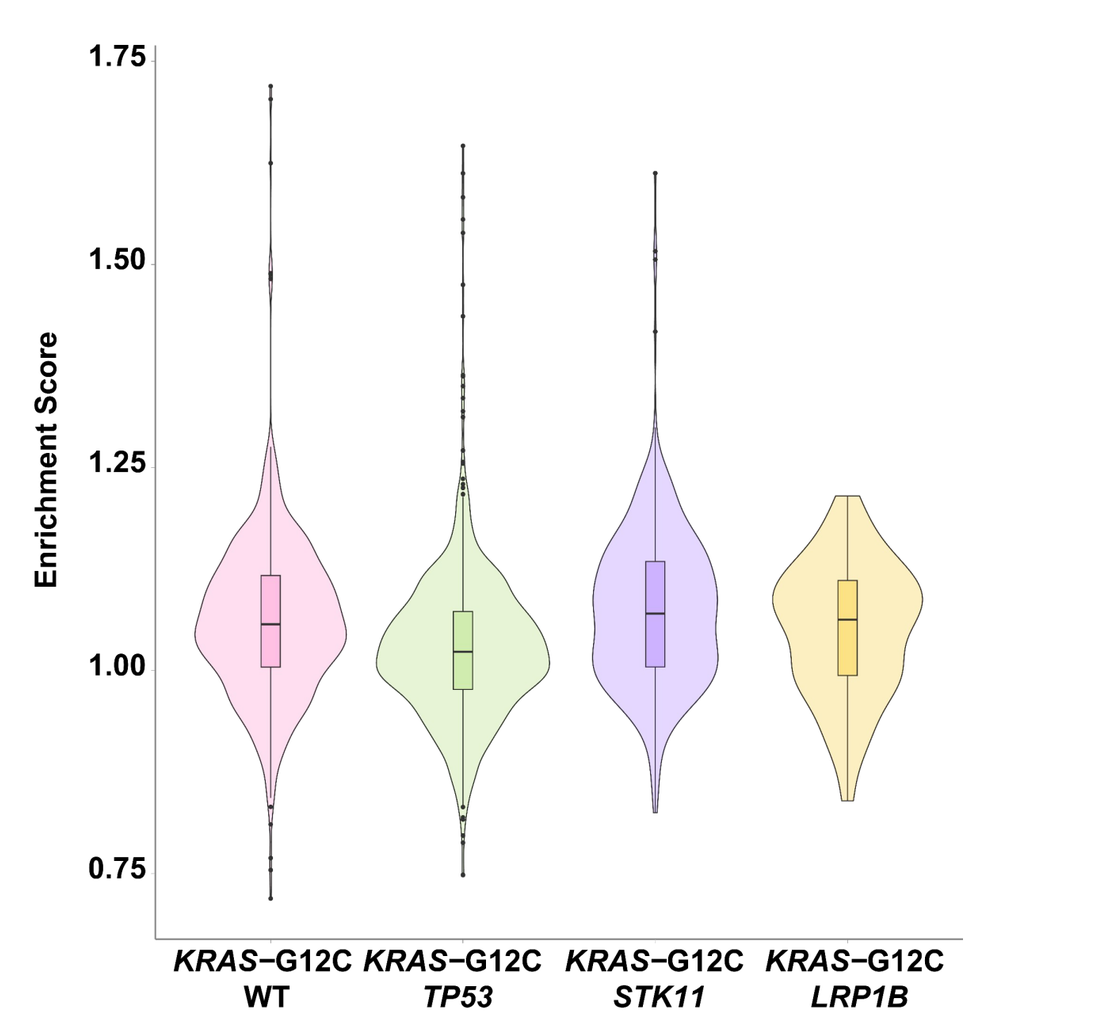
Enrichment Score
KRAS−G12C WT
KRAS−G12C TP53
KRAS−G12C STK11
KRAS−G12C LRP1B

Characteristic	Overall N = 2,187	KRAS-G12C WT N = 767	KRAS-G12C TP53 N = 970	KRAS-G12C STK11 N = 364	KRAS-G12C LRP1B N = 86	p-value[1]
Enrichment Score						<0.001
Median (IQR)	1.04 (0.99, 1.10)	1.06 (1.00, 1.12)	1.02 (0.98, 1.07)	1.07 (1.00, 1.13)	1.06 (0.99, 1.11)	
Range	0.72, 1.72	0.72, 1.72	0.75, 1.65	0.83, 1.61	0.84, 1.21	

Figure 2: Patients with KRAS G12C/STK11 alt had the highest median lipid metabolism gene enrichment score compared to other cohorts (p <0.001).

Figure 3. Multivariable Logistic Regression of High vs. Low Lipid Metabolic Gene Enrichment Score

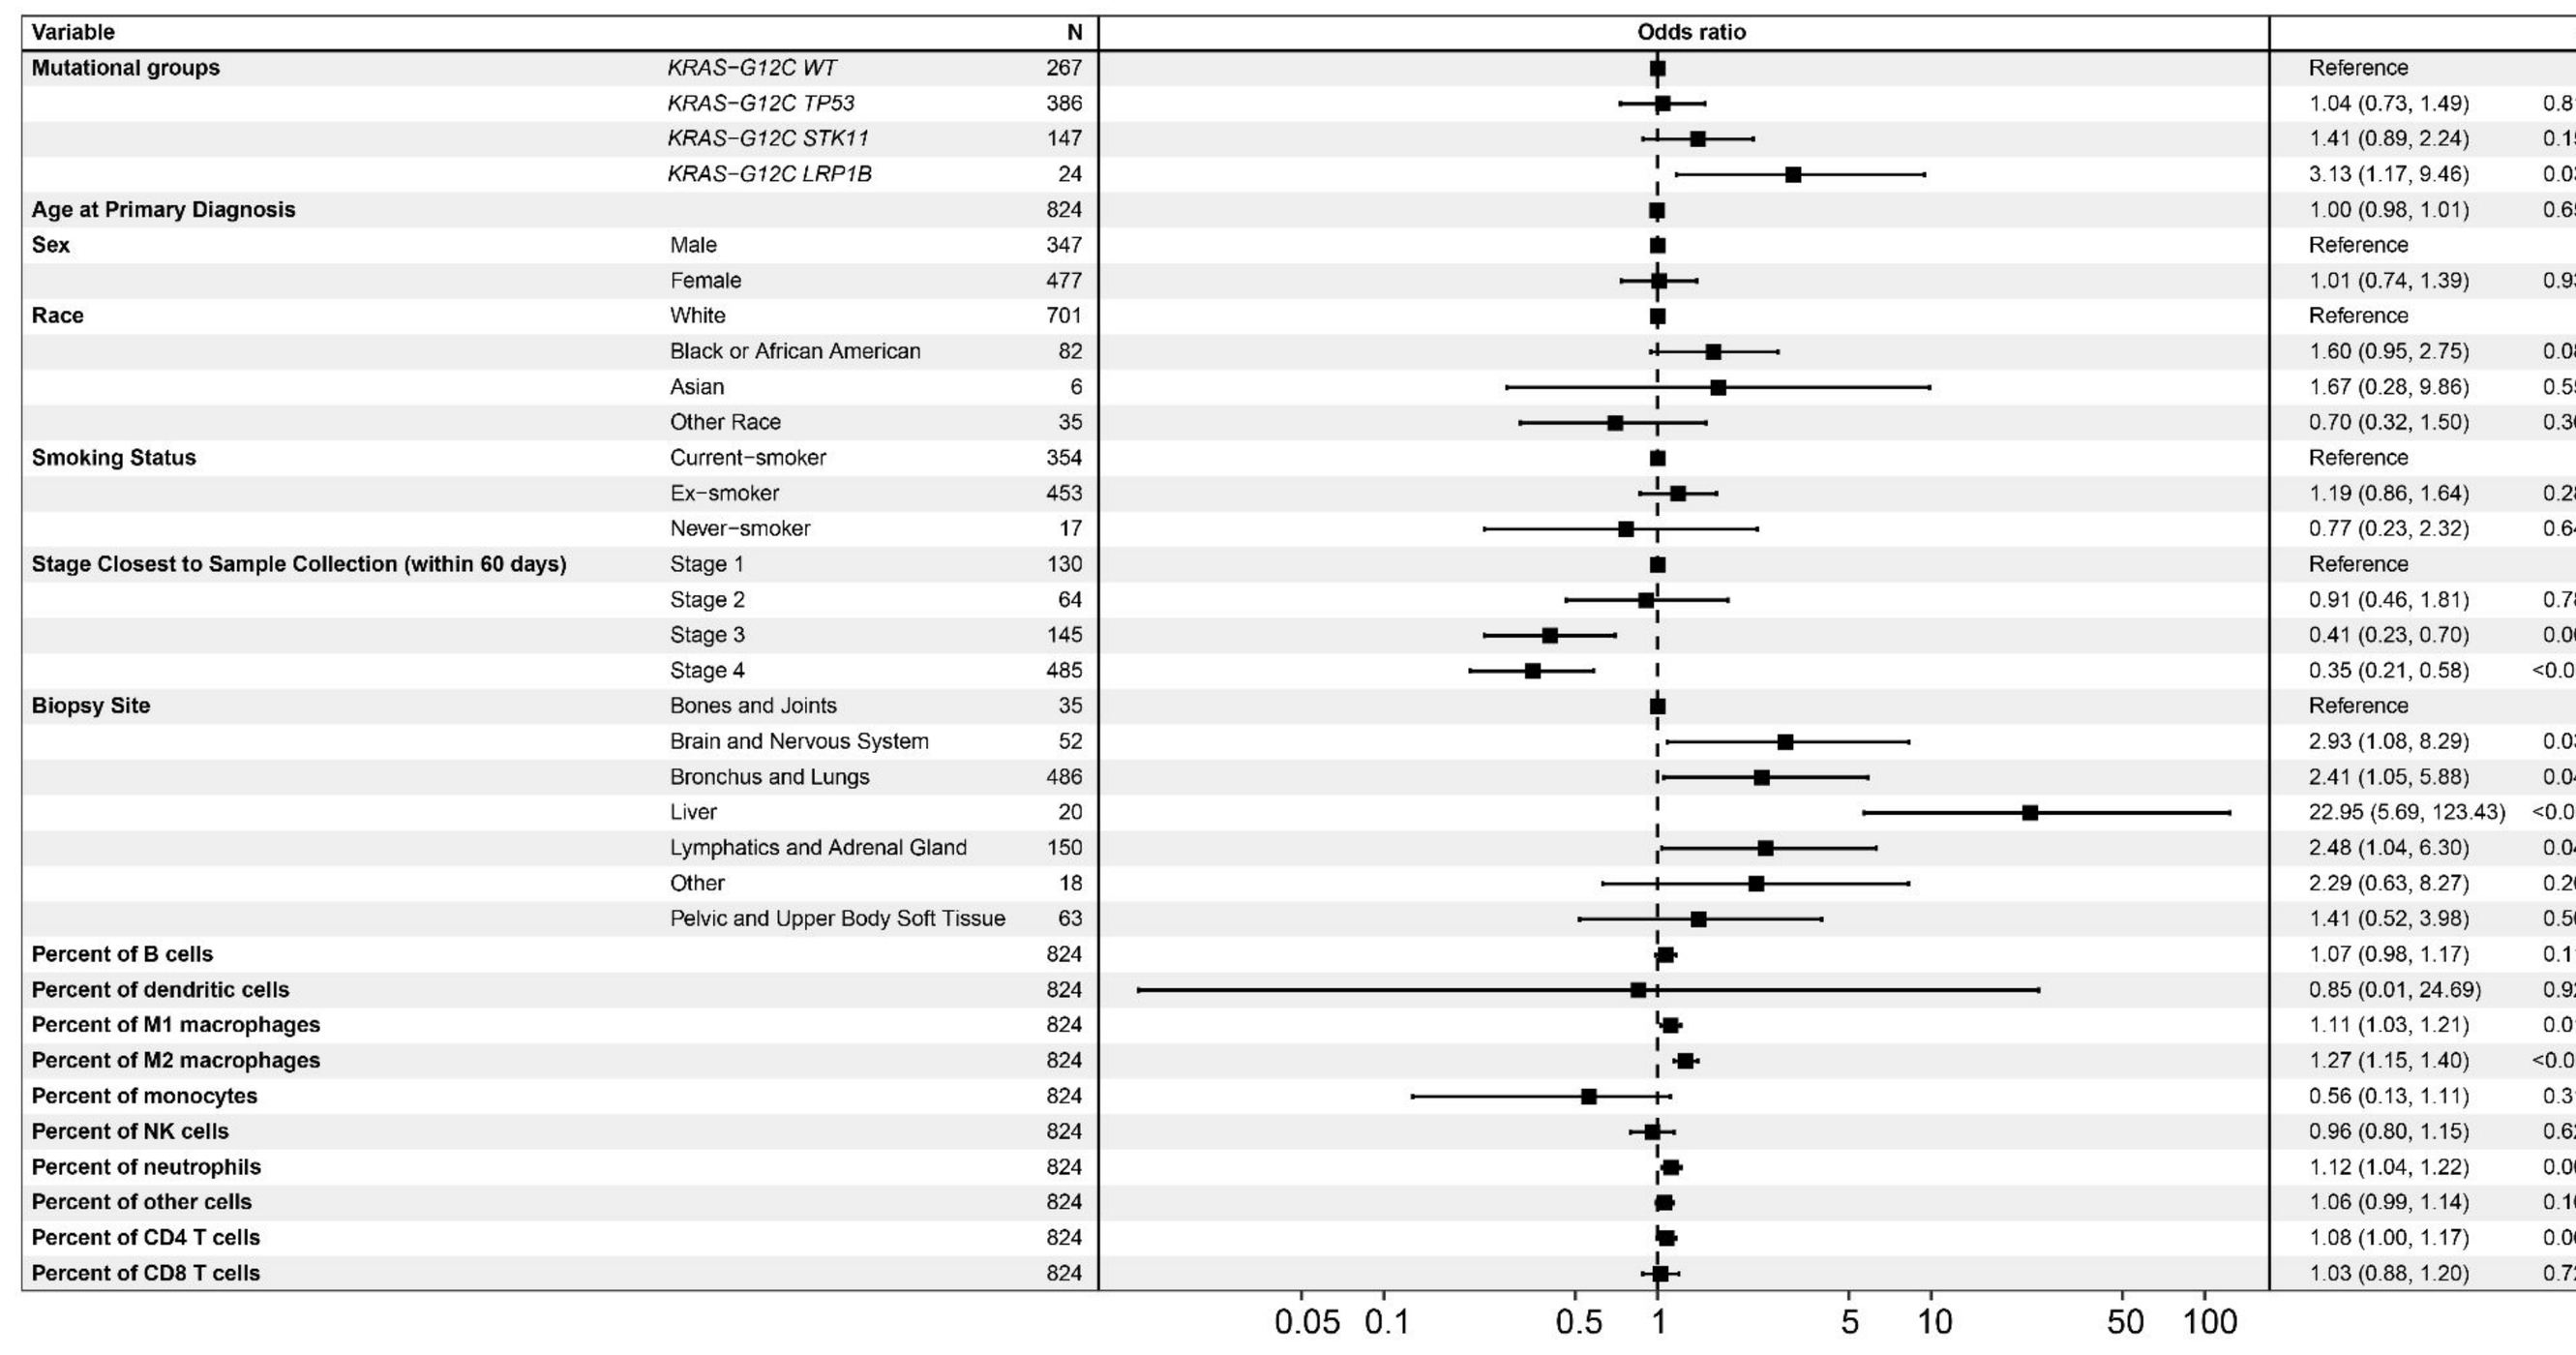

Variable		N	Odds ratio	p
Mutational groups	*KRAS−G12C WT*	267	Reference	
	KRAS−G12C TP53	386	1.04 (0.73, 1.49)	0.813
	KRAS−G12C STK11	147	1.41 (0.89, 2.24)	0.150
	KRAS−G12C LRP1B	24	3.13 (1.17, 9.46)	0.030
Age at Primary Diagnosis		824	1.00 (0.98, 1.01)	0.657
Sex	Male	347	Reference	
	Female	477	1.01 (0.74, 1.39)	0.933
Race	White	701	Reference	
	Black or African American	82	1.60 (0.95, 2.75)	0.082
	Asian	6	1.67 (0.28, 9.86)	0.554
	Other Race	35	0.70 (0.32, 1.50)	0.369
Smoking Status	Current−smoker	354	Reference	
	Ex−smoker	453	1.19 (0.86, 1.64)	0.289
	Never−smoker	17	0.77 (0.23, 2.32)	0.644
Stage Closest to Sample Collection (within 60 days)	Stage 1	130	Reference	
	Stage 2	64	0.91 (0.46, 1.81)	0.788
	Stage 3	145	0.41 (0.23, 0.70)	0.001
	Stage 4	485	0.35 (0.21, 0.58)	<0.001
Biopsy Site	Bones and Joints	35	Reference	
	Brain and Nervous System	52	2.93 (1.08, 8.29)	0.037
	Bronchus and Lungs	486	2.41 (1.05, 5.88)	0.043
	Liver	20	22.95 (5.69, 123.43)	<0.001
	Lymphatics and Adrenal Gland	150	2.48 (1.04, 6.30)	0.047
	Other	18	2.29 (0.63, 8.27)	0.202
	Pelvic and Upper Body Soft Tissue	63	1.41 (0.52, 3.98)	0.503
Percent of B cells		824	1.07 (0.98, 1.17)	0.114
Percent of dendritic cells		824	0.85 (0.01, 24.69)	0.927
Percent of M1 macrophages		824	1.11 (1.03, 1.21)	0.012
Percent of M2 macrophages		824	1.27 (1.15, 1.40)	<0.001
Percent of monocytes		824	0.56 (0.13, 1.11)	0.319
Percent of NK cells		824	0.96 (0.80, 1.15)	0.627
Percent of neutrophils		824	1.12 (1.04, 1.22)	0.005
Percent of other cells		824	1.06 (0.99, 1.14)	0.103
Percent of CD4 T cells		824	1.08 (1.00, 1.17)	0.062
Percent of CD8 T cells		824	1.03 (0.88, 1.20)	0.729

Figure 3: To determine if lipid genes were associated with immunogenic changes, a multivariable logistic regression model was fit with LMG ES (high vs. low) as the outcome variable. The ES was significantly associated with immune cell infiltration percentages for M1 macrophages, M2 macrophages, and neutrophils, with a trend towards significant association with CD4 T cells. Regulatory T cells were omitted from the model due to multicollinearity.

Figure 4. rwOS Association with Lipid Metabolic Gene Expression Levels in Patients with *KRAS* G12C/*STK11* alt

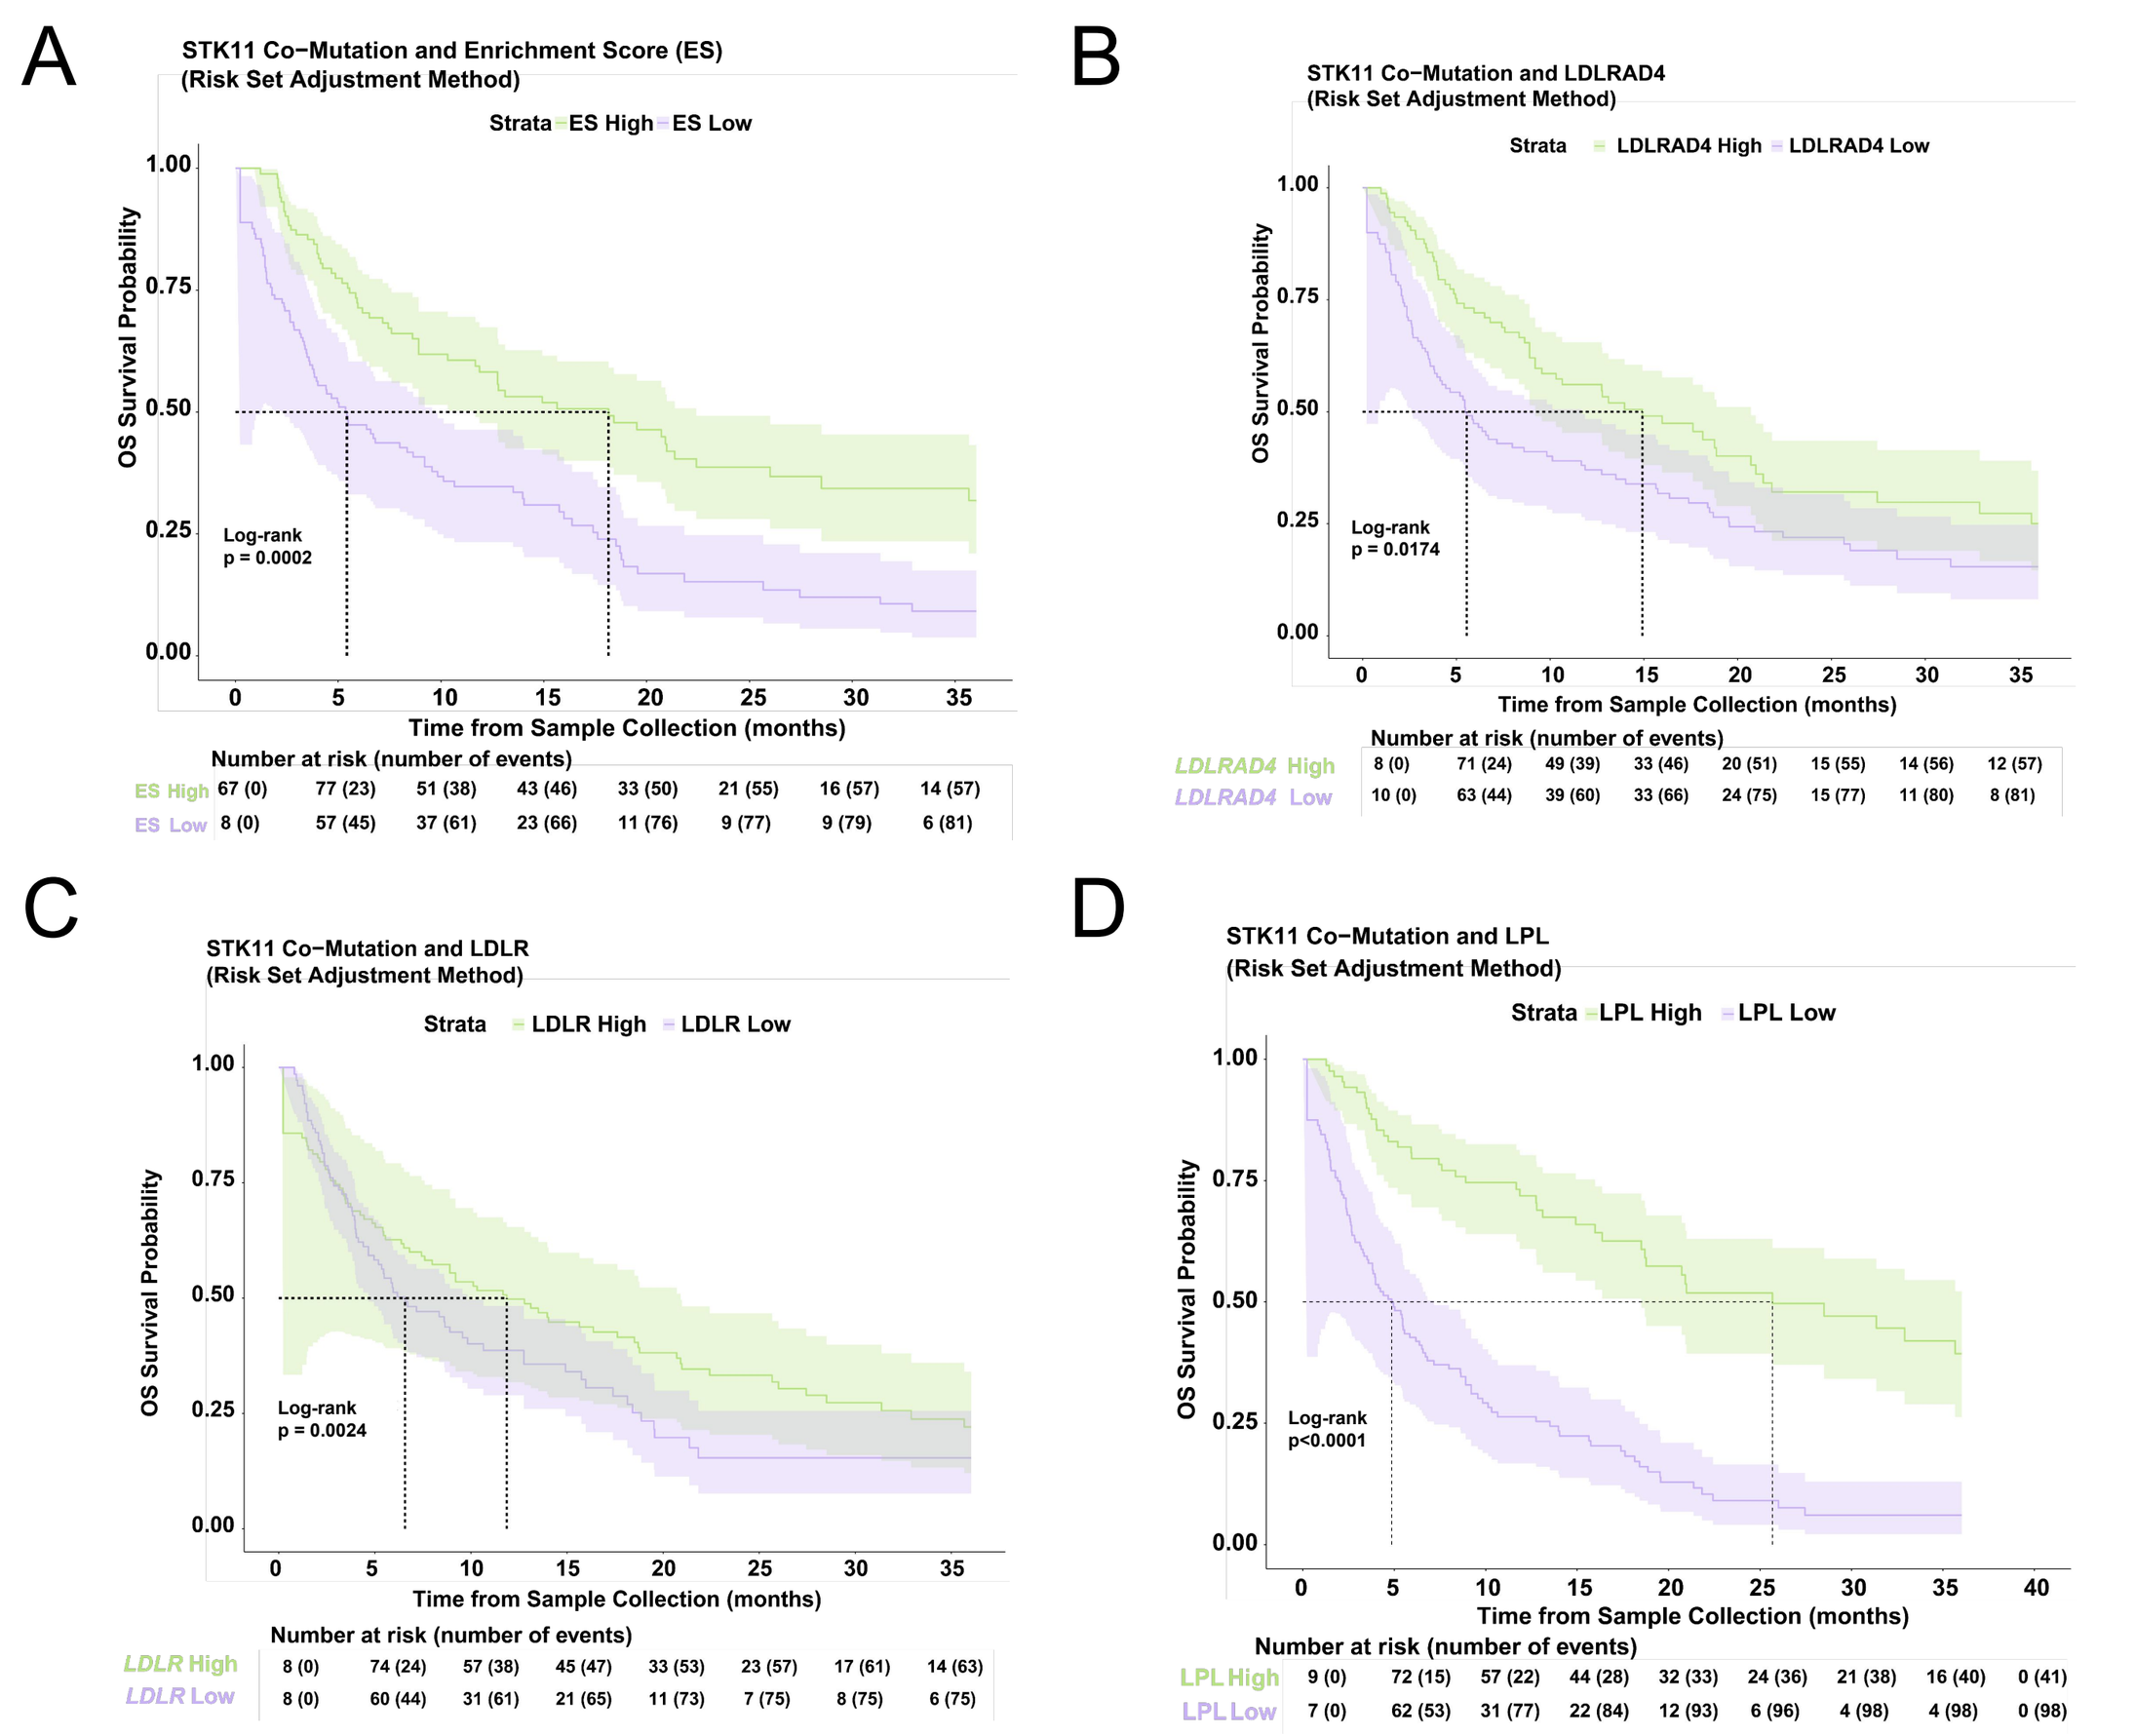
A
STK11 Co−Mutation and Enrichment Score (ES)
(Risk Set Adjustment Method)
Strata ES High ES Low
Log-rank
p = 0.0002
B
STK11 Co−Mutation and LDLRAD4
(Risk Set Adjustment Method)
Strata LDLRAD4 High LDLRAD4 Low
Log-rank
p = 0.0174
C
STK11 Co−Mutation and LDLR
(Risk Set Adjustment Method)
Strata LDLR High LDLR Low
Log-rank
p = 0.0024
D
STK11 Co−Mutation and LPL
(Risk Set Adjustment Method)
Strata LPL High LPL Low
Log-rank
p<0.0001
OS Survival Probability
Time from Sample Collection (months)
Number at risk (number of events)

Figure 4: Patients with KRAS G12C/STK11 alt and low LMG ES had decreased median rwOS (5.4 vs 18.2 months, p = 0.0002) compared to patients with a high ES. Additionally, low LDLRAD4 (5.6 vs 14.9 months, p = 0.0174), LDLR (6.6 vs 11.9 months, p = 0.0024), and LPL (4.9 vs 25.7 months, p <0.0001) expression was associated with poorer rwOS in patients with KRAS G12C/STK11 alt.

ACKNOWLEDGMENTS

We thank Vanessa Nepomuceno from the Tempus Science Communications team for poster development.

Correspondence: fweinb1@uic.edu