Integrative modeling of multimodal real-world data for improved risk stratification of first-line CDK4/6 inhibitor outcomes in patients with estrogen receptor (ER)-positive/human epidermal growth factor receptor 2 (HER2)-negative metastatic breast cancer

Francisco Sanchez-Vega[1]*, Subhiksha Nandakumar[1], Gráinne Long[2], Walid Chatila[1], Sameet Sreenivasan[3], Maureen Carey[4], Mohamed Reda Keddar[2], Jessica Davies[5], Matthew Zatzman[1], Martin Miller[2†], Elza de Bruin[6], Sajan Khosla[2], Nick Ceglia[1], Hannah Lees[1], Fatema Nagib[1], Yixiao Gong[1], Mark Donoghue[1], Nikolaus Schultz[1], Sohrab Shah[1].

1. Memorial Sloan Kettering Cancer Center, New York, NY, USA; 2. Clinical Data Sciences, AstraZeneca, Cambridge, UK; 3. Clinical Data Sciences, AstraZeneca, Gaithersburg, MD, USA; 4. Tempus AI Inc., Chicago, IL, USA;
5. Clinical Data Sciences, AstraZeneca, Barcelona, Spain; 6. Translational Medicine, Oncology R&D AstraZeneca, Cambridge, UK.

*Presenting author. †At time of study.

Corresponding author email address: sanchezf@mskcc.org

Memorial Sloan Kettering Cancer Center

Background

- CDK4/6 inhibition in combination with endocrine therapy is a cornerstone of treatment for hormone receptor-positive metastatic breast cancer.
- We developed a machine learning model that used clinical, genomic, and transcriptomic features to stratify patients based on response to first-line CDK4/6 inhibitor plus endocrine treatment and identify predictors of response.

Methods

Patient selection

- Patients with ER-positive/HER2-negative metastatic breast cancer biopsy samples from before or <30 days after initiation of first-line treatment.

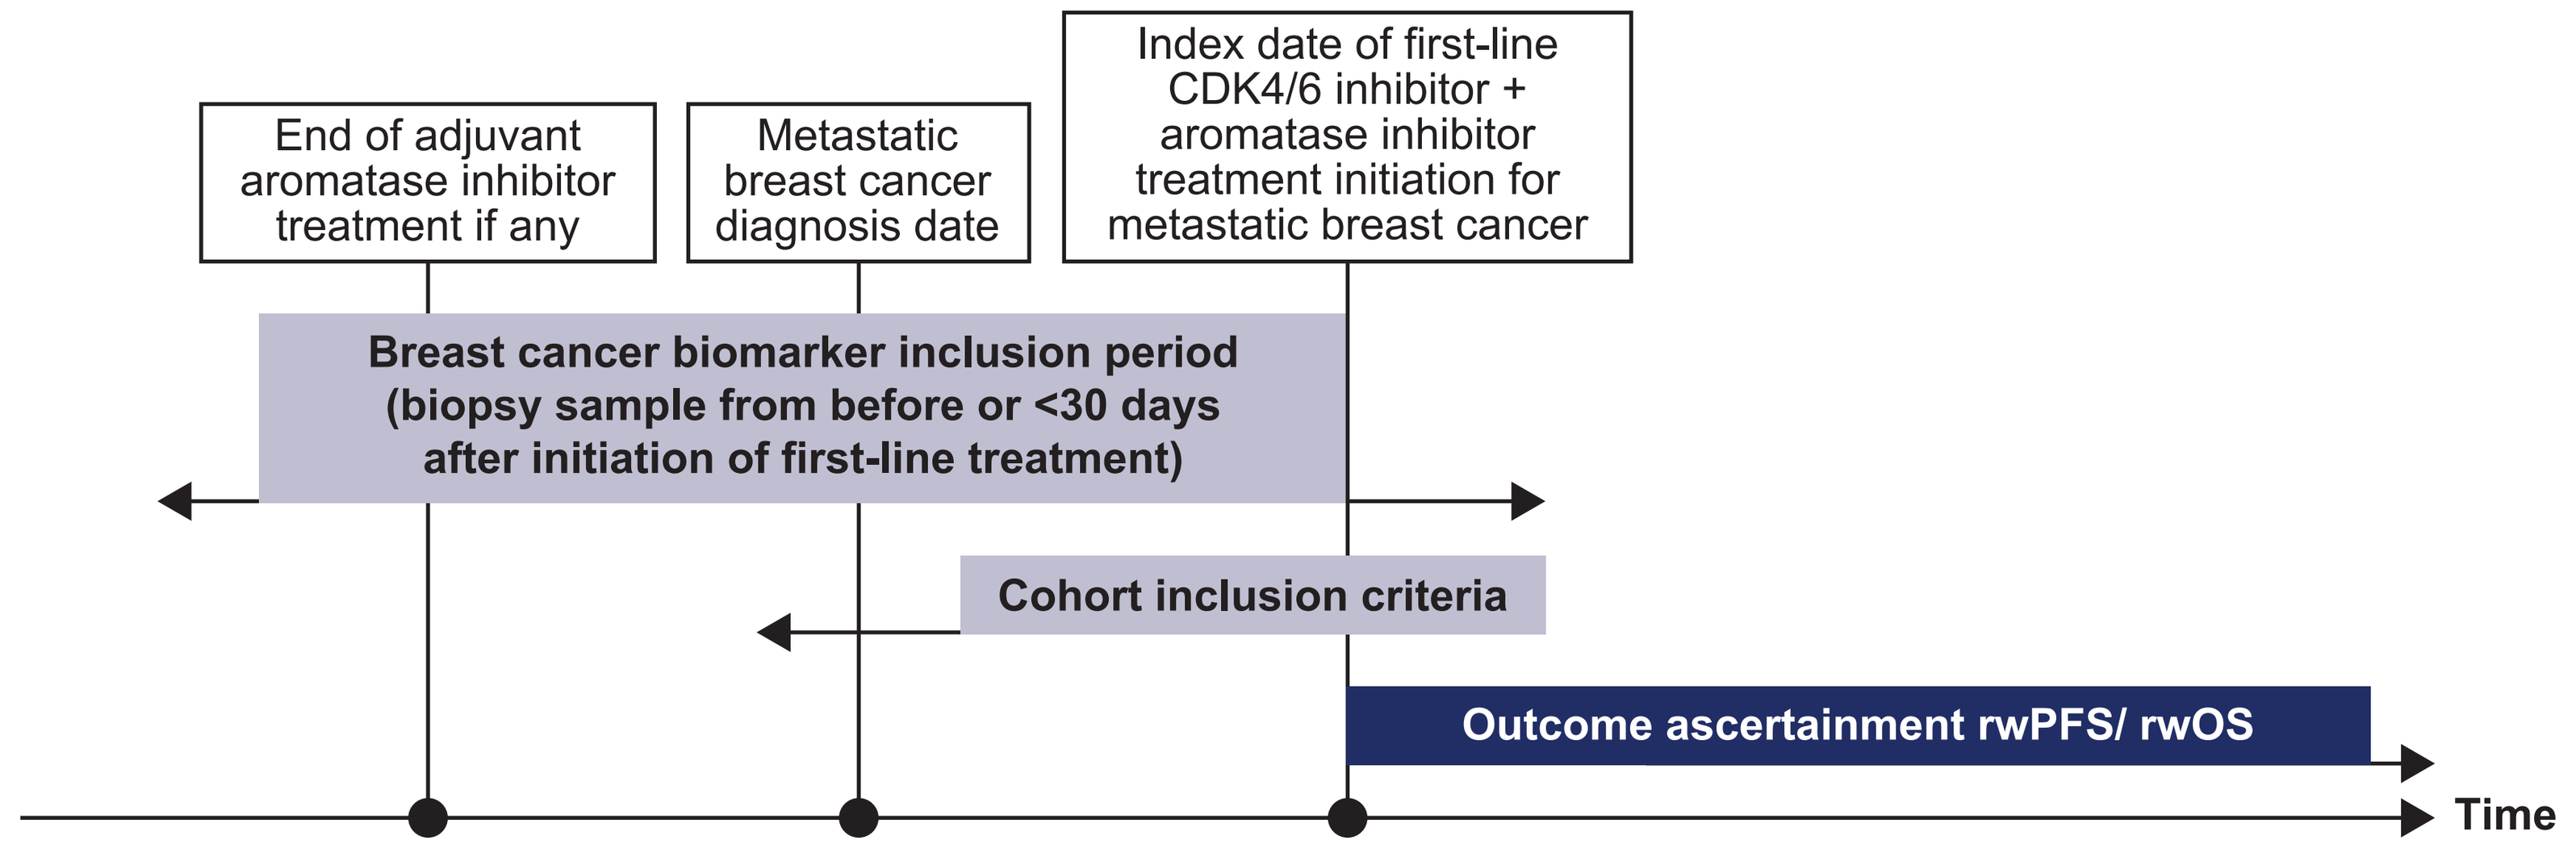
End of adjuvant aromatase inhibitor treatment if any
Metastatic breast cancer diagnosis date
Index date of first-line CDK4/6 inhibitor + aromatase inhibitor treatment initiation for metastatic breast cancer
Breast cancer biomarker inclusion period (biopsy sample from before or <30 days after initiation of first-line treatment)
Cohort inclusion criteria
Outcome ascertainment rwPFS/ rwOS
Time

- Curated clinical outcome data from the Tempus real-world multimodal database were leveraged. Genomic data included DNA sequencing (Tempus xT assay: xT, xT.v2, xT.v3, or xT.v4) and whole transcriptome sequencing (Tempus xR assay).

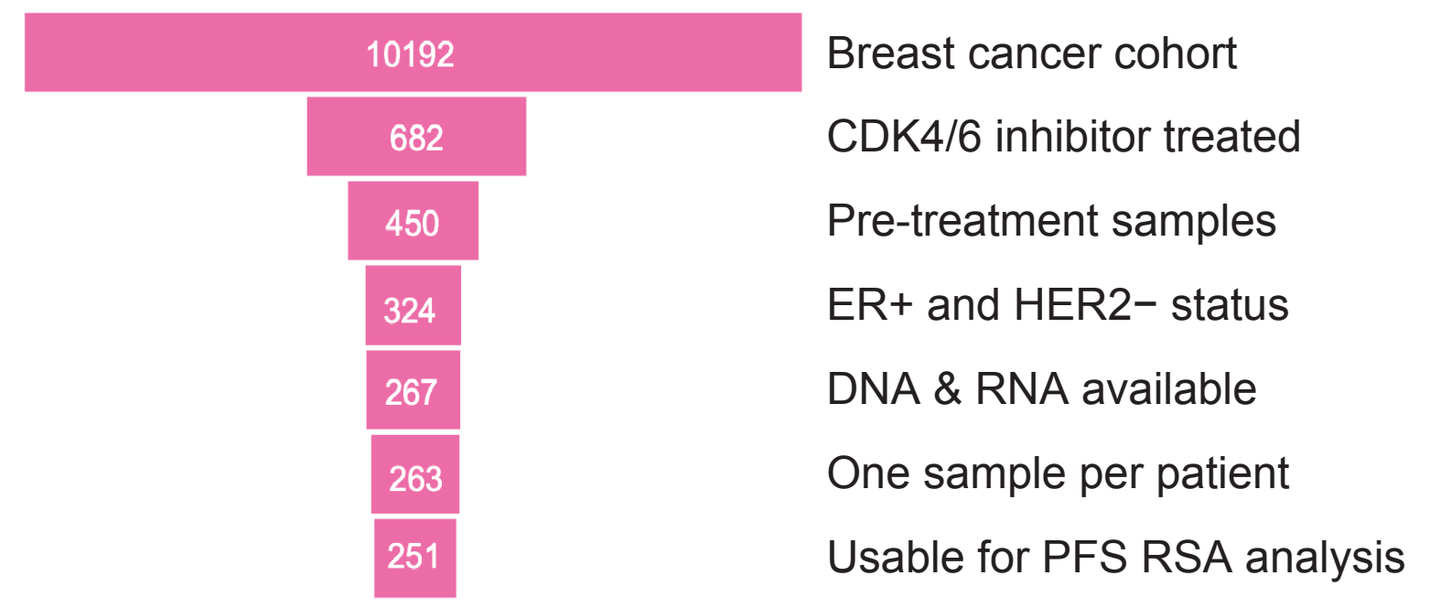
10192 Breast cancer cohort
682 CDK4/6 inhibitor treated
450 Pre-treatment samples
324 ER+ and HER2− status
267 DNA & RNA available
263 One sample per patient
251 Usable for PFS RSA analysis

Analyses

- Driver DNA alterations associated with breast cancer were extracted at the gene and pathway levels.[1]
- Genomic, transcriptomic, and clinicopathological features (age, tissue site, *de novo* disease, stage, early recurrence) and treatment characteristics (CDK4/6 inhibitor, endocrine therapy partner, adjuvant treatment) were used as explanatory variables in an integrative risk stratification model that used the OncoCast algorithm[2] to predict real-world progression-free survival (rwPFS) in patients treated with first-line CDK4/6 inhibitors.
- Patients were stratified into low- or high-risk groups based on their progression risk score.
- Cell type proportions and cell states were decomposed from bulk RNA sequencing data using EcoTyper.[3]
- Risk-set adjustment (RSA) was used to mitigate the effect of patients' delayed entry into the cohort due to the timing of their Tempus sequencing tests.

Results

Cohort overview

Demographics and clinical features

Characteristic	Category	N (%)
Age	≤30	3 (1.2)
	31–50	73 (29.1)
	51–70	140 (55.8)
	>70	35 (13.9)
Metastatic sites	1 site	95 (37.8)
	2–3 sites	116 (46.3)
	≥4 sites	40 (15.9)
Stage closest to biopsy	1	3 (1.2)
	2	9 (3.6)
	3	4 (1.6)
	4	166 (66.1)
	Unknown	69 (27.5)
Type	*De novo*	96 (38.2)
	Recurrent	155 (61.8)
Tempus assay	xT	2 (0.8)
	xT.v2	33 (13.2)
	xT.v3	47 (18.7)
	xT.v4	169 (67.3)

Treatment characteristics

Category	Subgroup	*De novo* disease (n=96)	Recurrent disease (n=155)
First-line treatment for metastatic breast cancer, n (%)*			
CDK4/6 inhibitor	Palbociclib	63 (65.6)	95 (61.3)
	Ribociclib	20 (20.8)	21 (13.6)
	Abemaciclib	16 (16.7)	44 (28.4)
Endocrine therapy partner	Aromatase inhibitor	80 (83.3)	75 (48.4)
	Fulvestrant	14 (14.6)	77 (49.7)
	Tamoxifen	3 (3.1)	2 (1.3)
Adjuvant treatment, n (%)			
Endocrine therapy	Any†	-	122 (78.7)
	Aromatase inhibitor	-	100 (64.5)
	Fulvestrant	-	4 (2.6)
	Tamoxifen	-	57 (36.8)
Treatment-free interval (TFI)	<12 months	-	74 (47.7)
	≥12 months	-	24 (15.5)
	Unknown	-	57 (36.8)

*Some patients received >1 CDK4/6 inhibitor or endocrine therapy as first-line treatment for metastatic breast cancer (*de novo* patients: >1 CDK4/6 inhibitor, n=3, and >1 endocrine therapy, n=3; recurrent patients: >1 CDK4/6 inhibitor, n=5, and >1 endocrine therapy, n=3).
†AI, fulvestrant or tamoxifen.

Overview of somatic alterations in known breast cancer-related genes

The frequency of somatic alterations in driver genes is consistent with previously reported analyses of hormone receptor-positive breast cancer.[4]

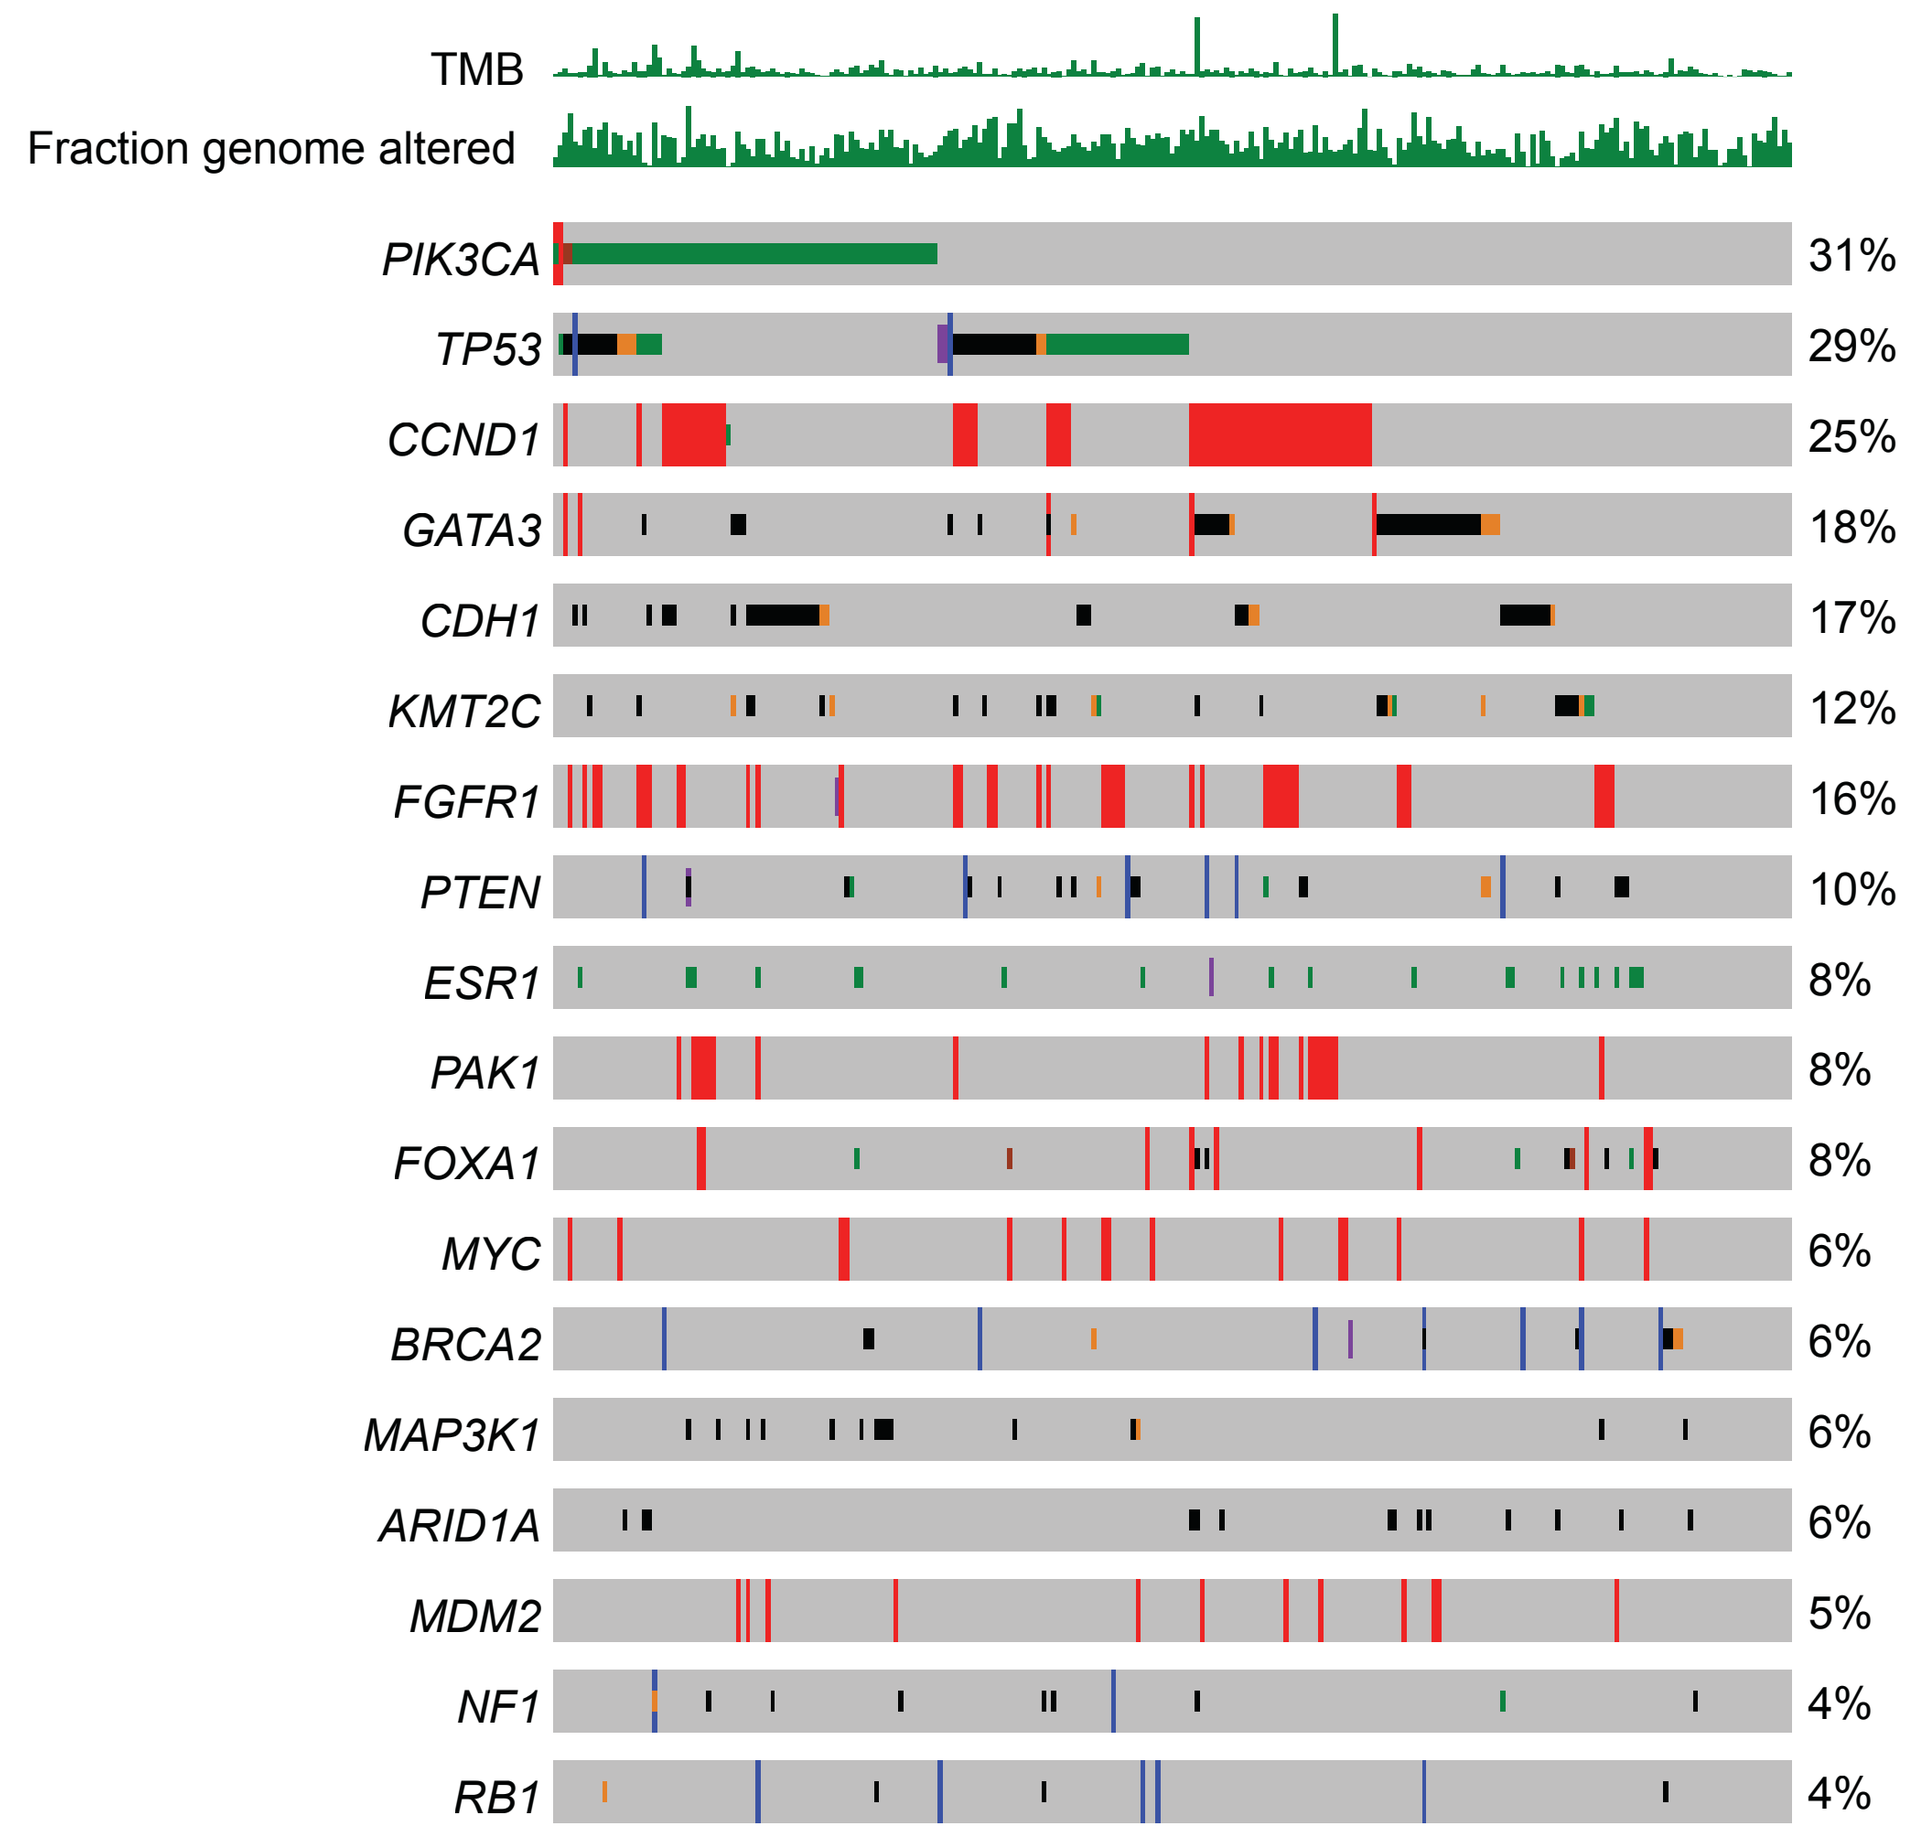
TMB
Fraction genome altered
PIK3CA 31%
TP53 29%
CCND1 25%
GATA3 18%
CDH1 17%
KMT2C 12%
FGFR1 16%
PTEN 10%
ESR1 8%
PAK1 8%
FOXA1 8%
MYC 6%
BRCA2 6%
MAP3K1 6%
ARID1A 6%
MDM2 5%
NF1 4%
RB1 4%

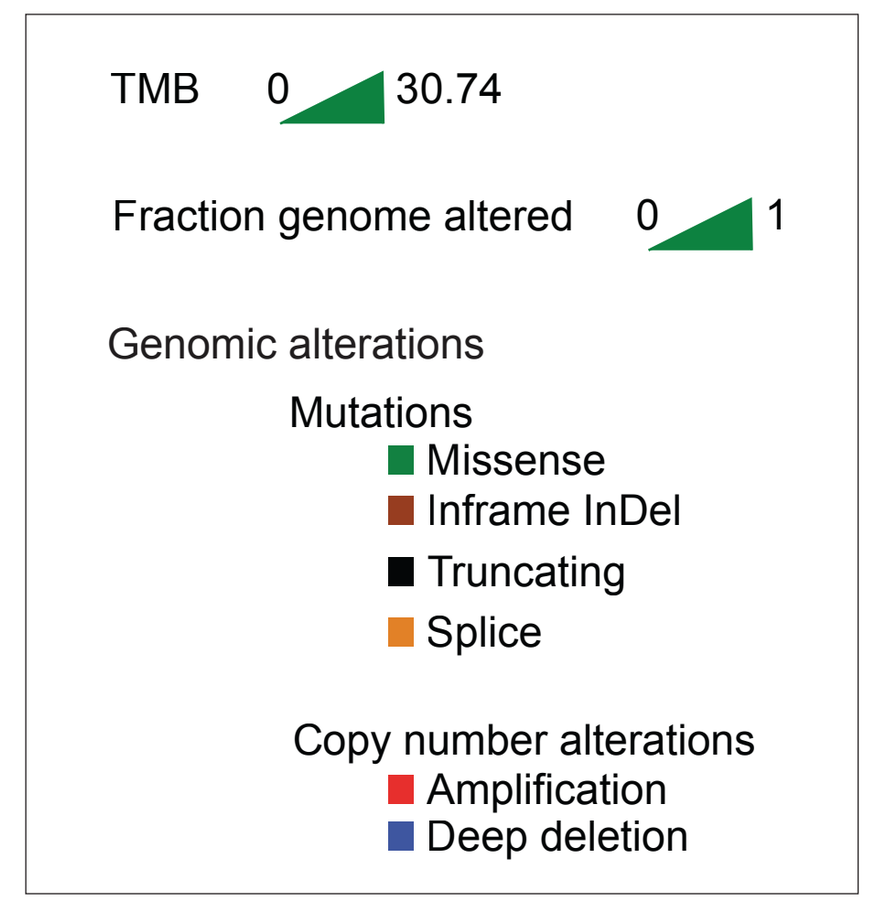
TMB 0 30.74
Fraction genome altered 0 1
Genomic alterations
Mutations
Missense
Inframe InDel
Truncating
Splice
Copy number alterations
Amplification
Deep deletion

- Copy number changes in driver genes correlate with RNA-Seq measurements.

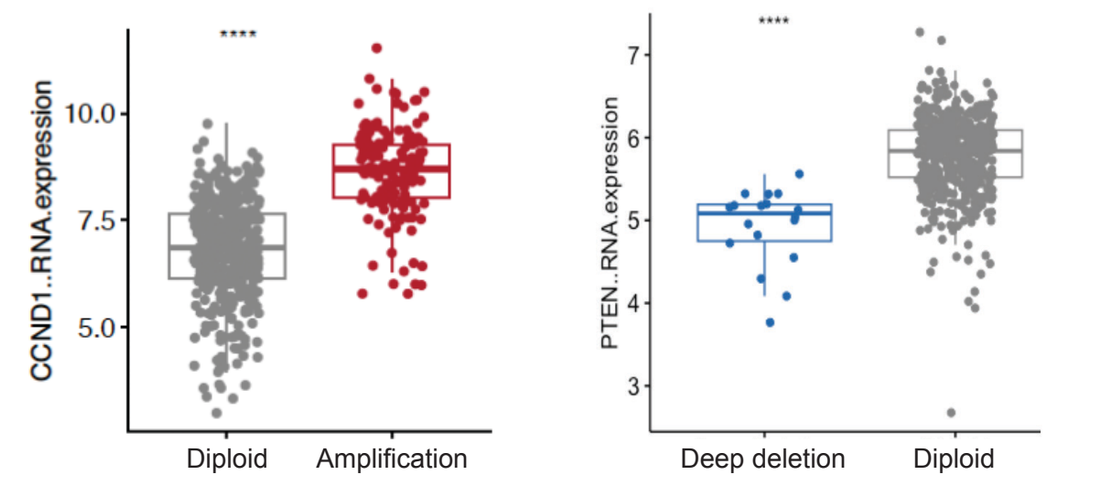
CCND1..RNA.expression
Diploid
Amplification
PTEN..RNA.expression
Deep deletion
Diploid

Patient stratification into low- and high-risk groups

- Patients were stratified into two risk groups (high-risk [median rwPFS 11.2 months, 95% confidence interval [CI]: 7.2–17.6]; low-risk [median rwPFS 18 months, 95% CI: 14.9–not reached]; log-rank p<0.001).

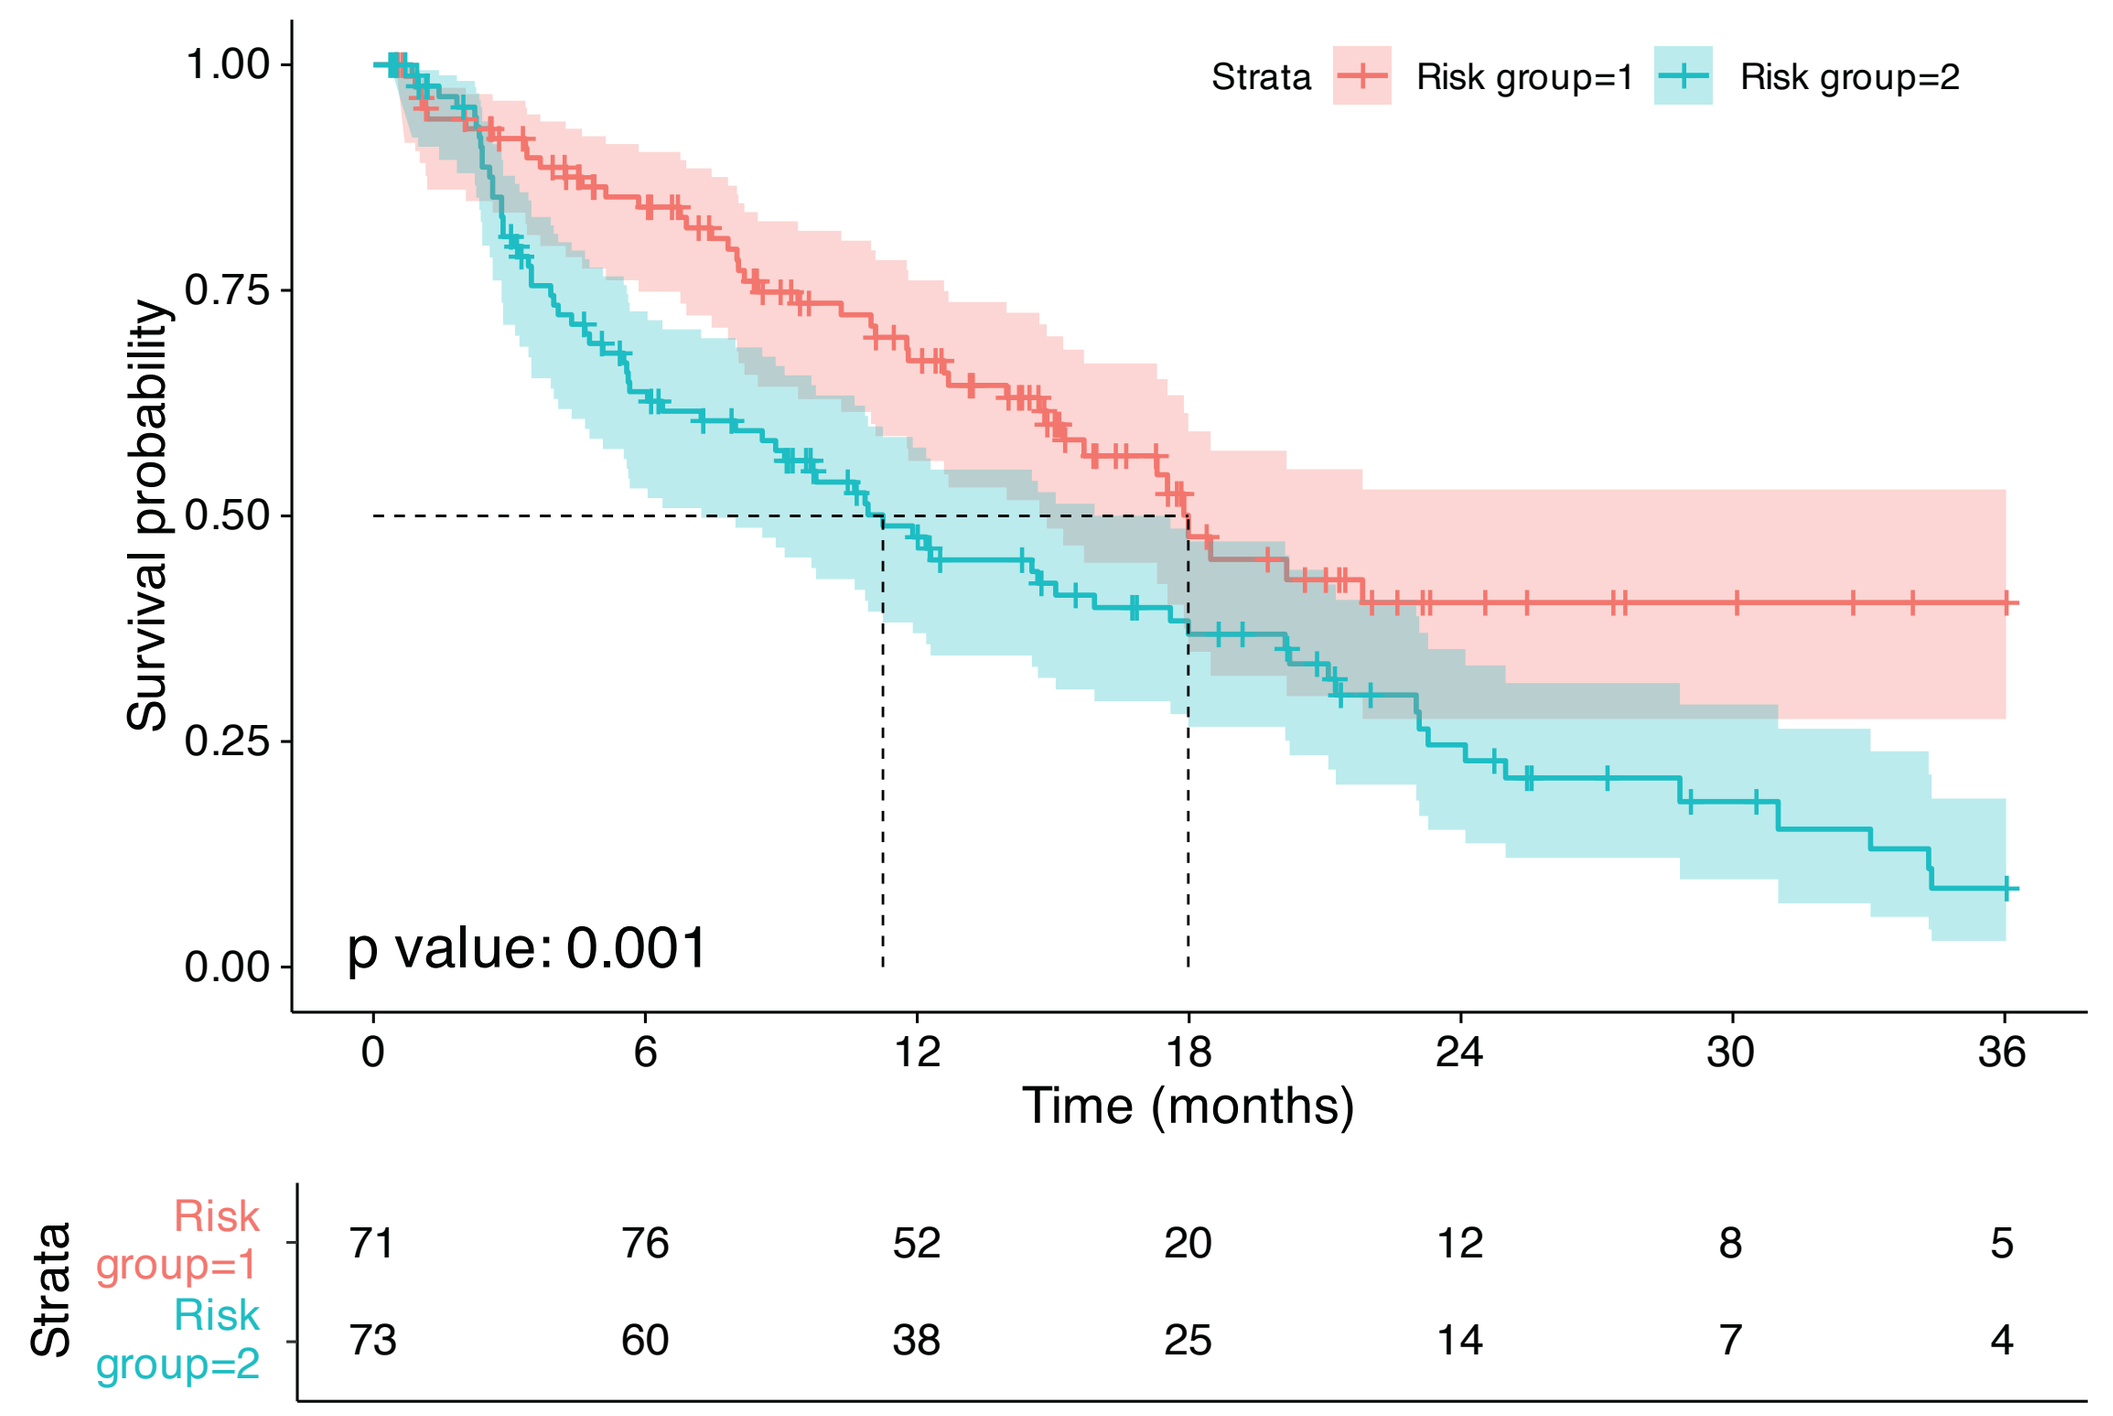
Strata
Risk group=1
Risk group=2
Survival probability
p value: 0.001
Time (months)
Risk group=1 71 76 52 20 12 8 5
Risk group=2 73 60 38 25 14 7 4

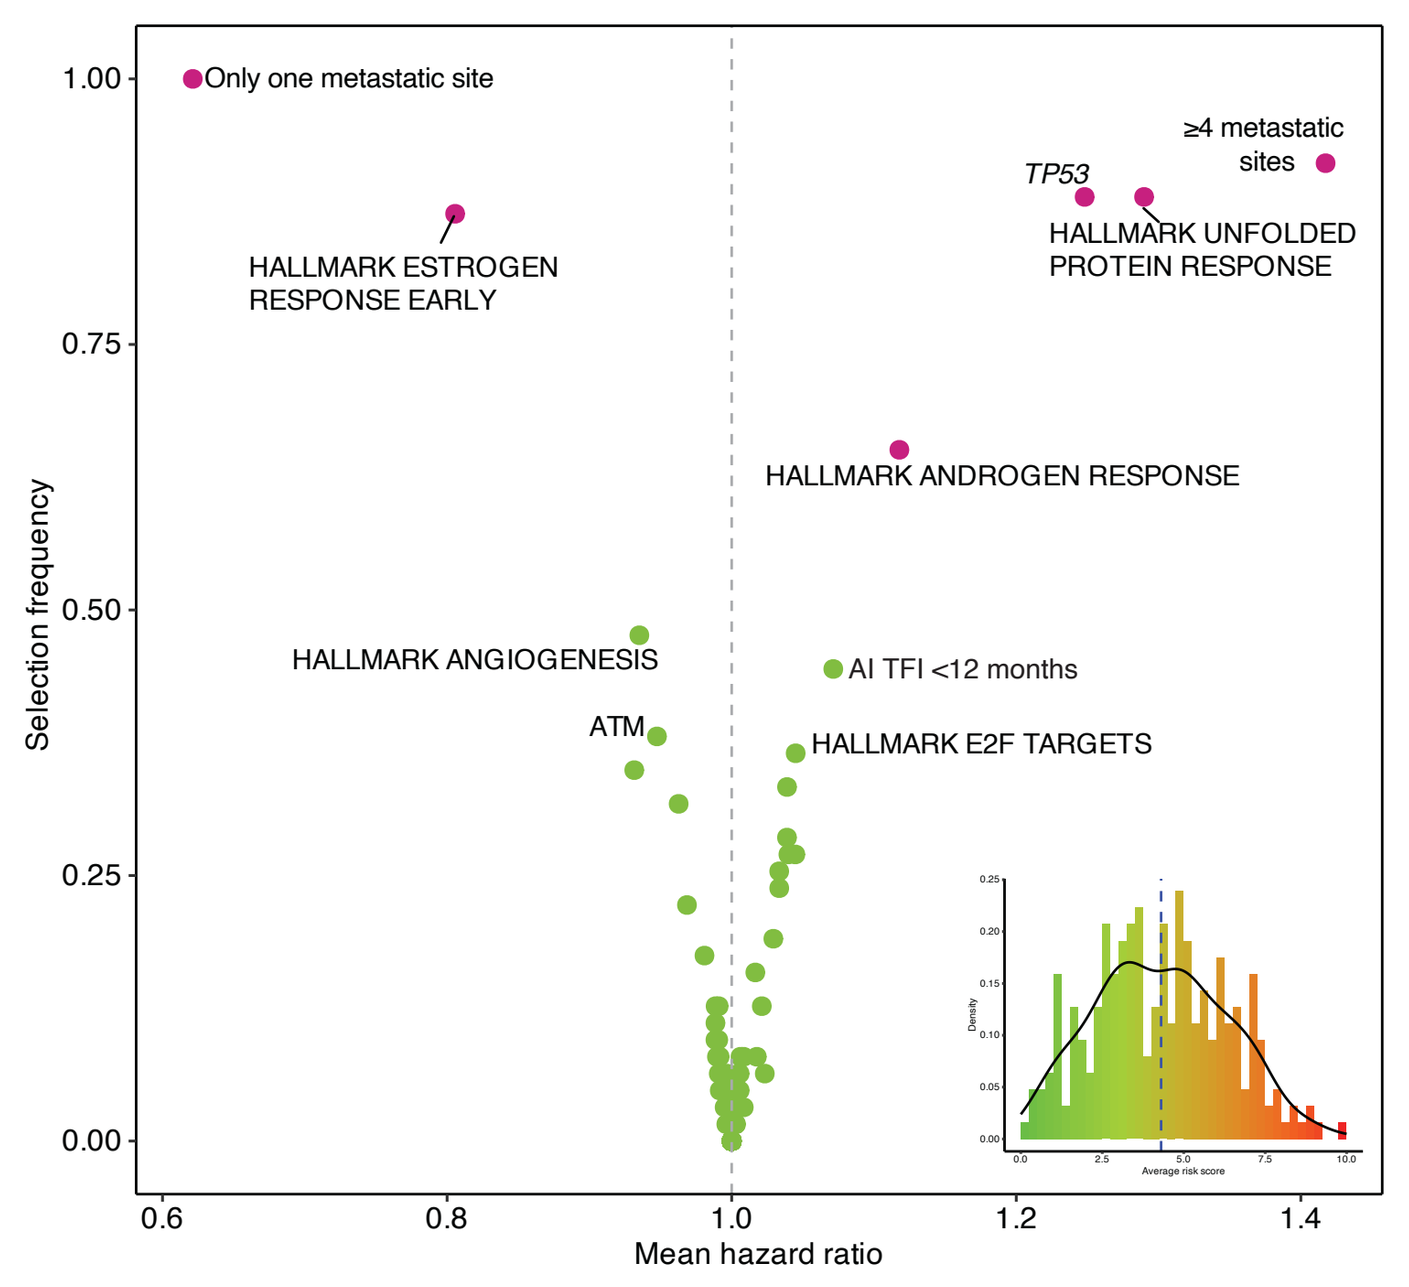
Only one metastatic site
HALLMARK ESTROGEN RESPONSE EARLY
TP53
≥4 metastatic sites
HALLMARK UNFOLDED PROTEIN RESPONSE
HALLMARK ANDROGEN RESPONSE
HALLMARK ANGIOGENESIS
AI TFI <12 months
ATM
HALLMARK E2F TARGETS
Selection frequency
Mean hazard ratio

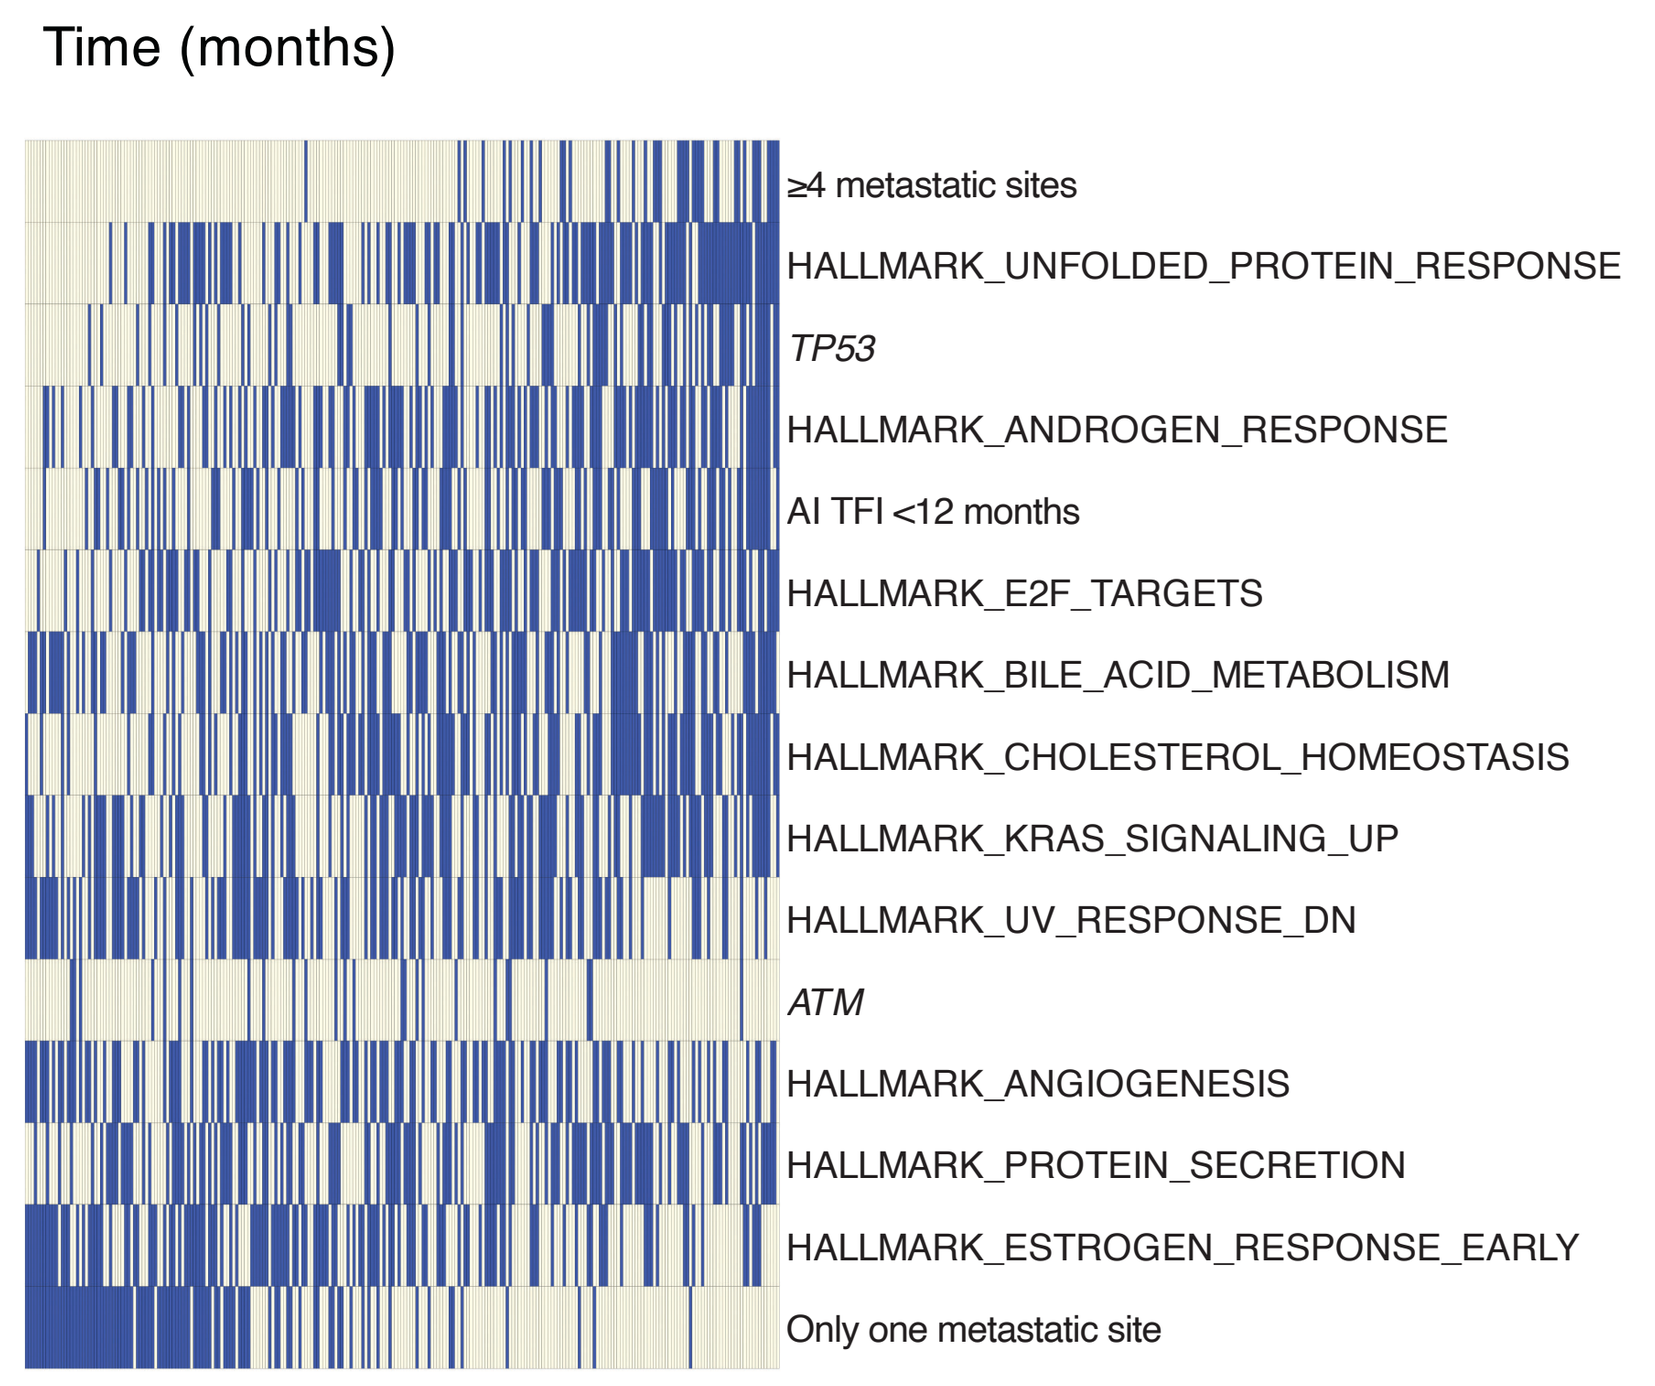
Time (months)
≥4 metastatic sites
HALLMARK_UNFOLDED_PROTEIN_RESPONSE
TP53
HALLMARK_ANDROGEN_RESPONSE
AI TFI <12 months
HALLMARK_E2F_TARGETS
HALLMARK_BILE_ACID_METABOLISM
HALLMARK_CHOLESTEROL_HOMEOSTASIS
HALLMARK_KRAS_SIGNALING_UP
HALLMARK_UV_RESPONSE_DN
ATM
HALLMARK_ANGIOGENESIS
HALLMARK_PROTEIN_SECRETION
HALLMARK_ESTROGEN_RESPONSE_EARLY
Only one metastatic site

- Features associated with the high-risk group included higher rates of TP53 alterations, >4 metastatic sites, and high expression of genes involved in unfolded protein response, cell cycle, metabolic, proliferative, and androgen response pathways.
- Features associated with the low-risk group included high expression of genes involved in early estrogen response pathway.

Comparison of transcriptional profiles between low- and high-risk groups

- We investigated differences in transcriptomic patterns as a function of risk status.
- Gene set enrichment analysis identified pathways with significant differences in expression across risk groups.

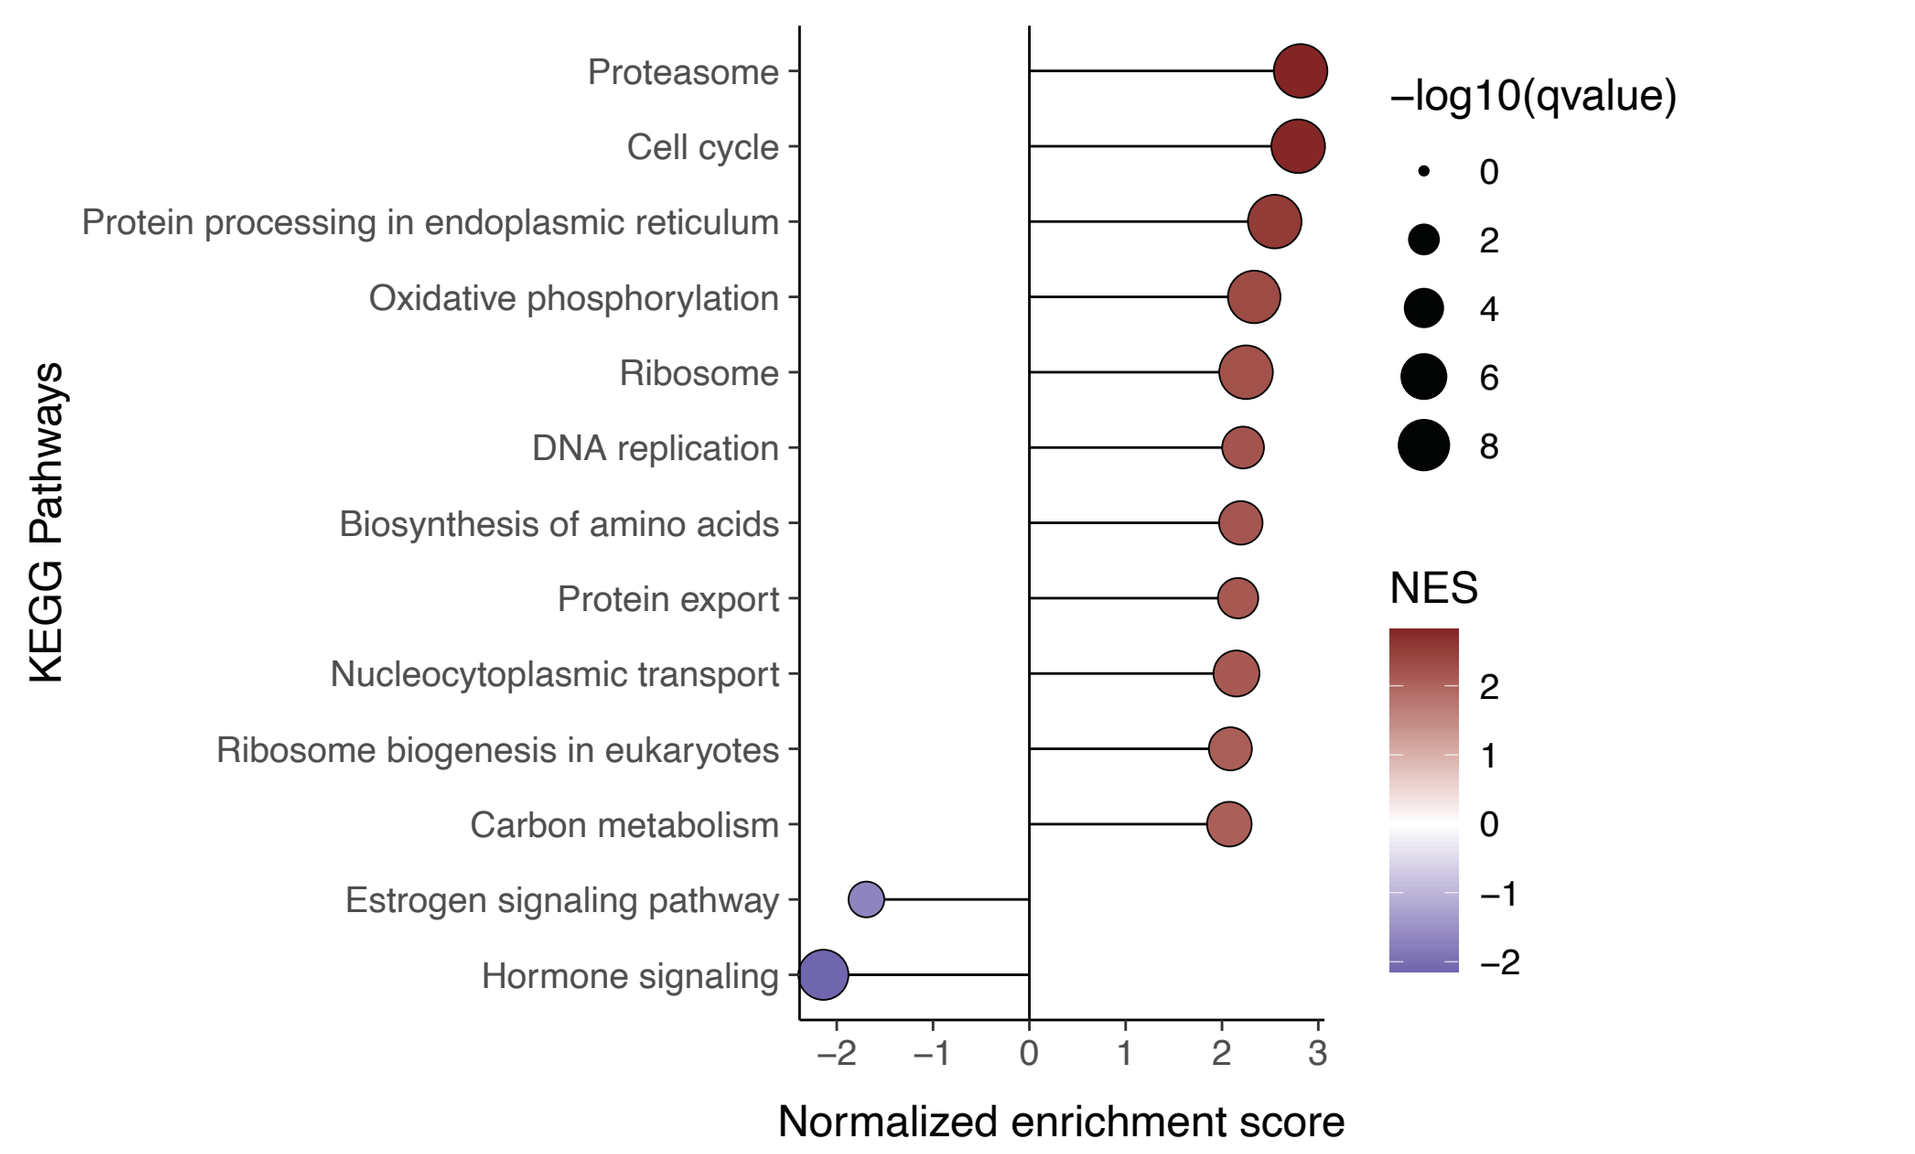
KEGG Pathways
Proteasome
Cell cycle
Protein processing in endoplasmic reticulum
Oxidative phosphorylation
Ribosome
DNA replication
Biosynthesis of amino acids
Protein export
Nucleocytoplasmic transport
Ribosome biogenesis in eukaryotes
Carbon metabolism
Estrogen signaling pathway
Hormone signaling
Normalized enrichment score
−log10(qvalue)
NES

- Differences in expression of individual genes are shown in the boxplots and heatmap.

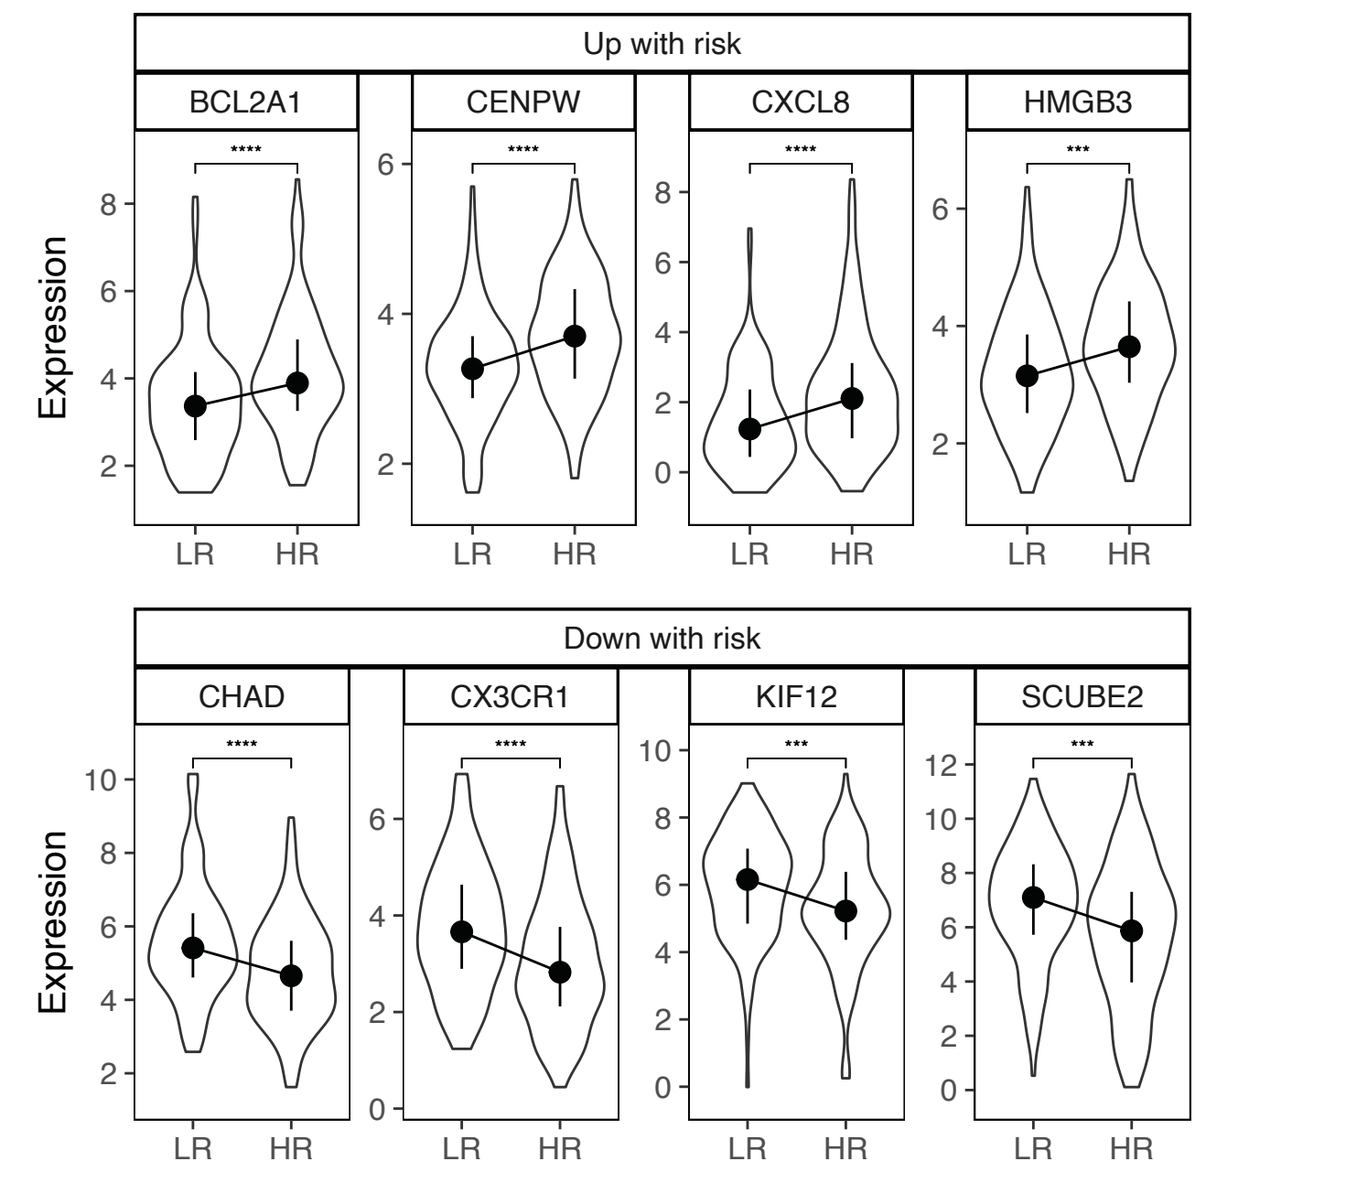
Up with risk
BCL2A1
CENPW
CXCL8
HMGB3
Down with risk
CHAD
CX3CR1
KIF12
SCUBE2
Expression
LR
HR

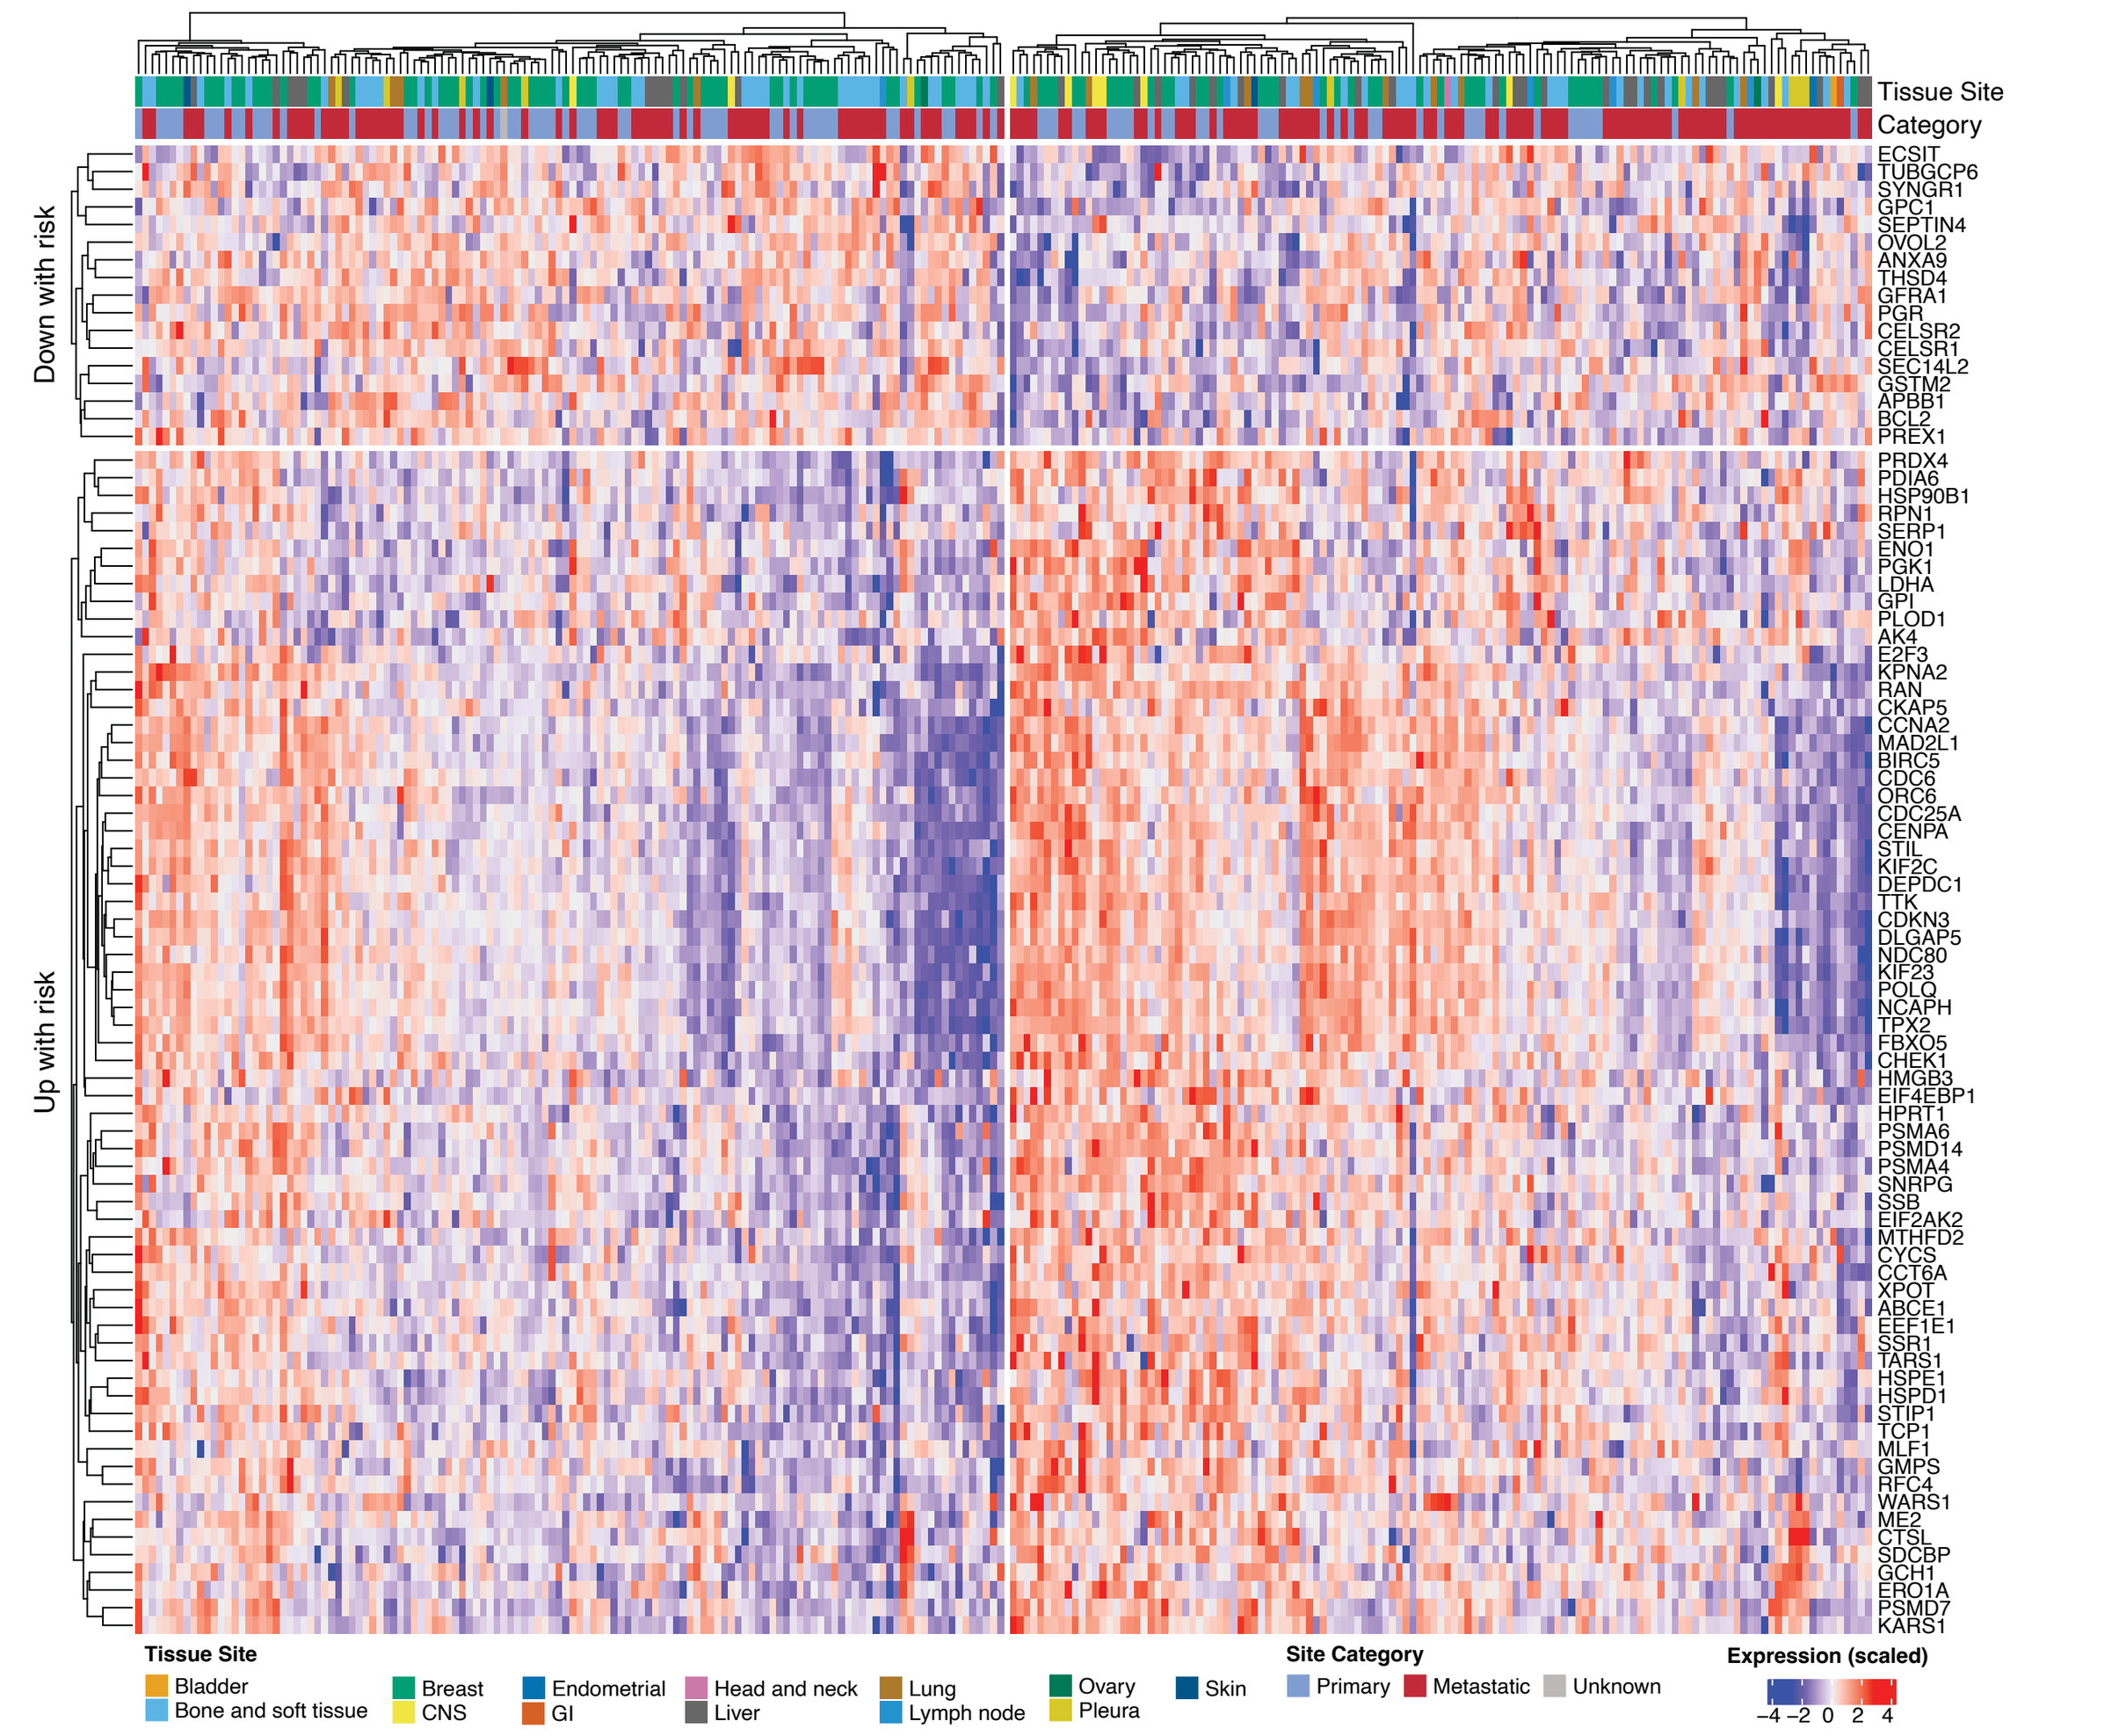
LR
HR
Tissue Site
Category
Down with risk
Up with risk
Tissue Site
Bladder
Bone and soft tissue
Breast
CNS
Endometrial
GI
Head and neck
Liver
Lung
Lymph node
Ovary
Pleura
Skin
Site Category
Primary
Metastatic
Unknown
Expression (scaled)

Conclusion

Our study shows how multimodal real-world data collected during routine care can provide valuable insights into the biology of response to CDK4/6 inhibitors in patients with metastatic breast cancer.

References

1. Sanchez-Vega F, et al. *Cell* 2018;173:321–337.e10. doi:10.1016/j.cell.2018.03.035.
2. Shen R, et al. *JCO Precis Oncol* 2019;3:PO.1800307. doi:10.1200/PO.18.00307.
3. Luca BA, et al. *Cell* 2021;184:5482–5496.e28. doi:10.1016/j.cell.2021.
4. Razavi P, et al. *Cancer Cell* 2018; 34:427–438.e6. doi: 10.1016/j.ccell.2018.08.008.

Please scan this quick response (QR) code with your smartphone camera or app to obtain a copy of this poster. Alternatively, please use the link here https://bit.ly/47whfAI
Copies of this poster obtained through QR Code are for personal use only and may not be reproduced without permission from SABCS® and the author