San Antonio Breast Cancer Symposium®, December 9–12, 2025
Poster number: PS2-06-18

Real-world second-line treatment use and clinical outcomes in patients with HR-positive/HER2-negative metastatic breast cancer (mBC) and an *ESR1* mutation

Spencer James,[1] Sushupta Vijapur,[2] Sameet Sreenivasan,[3] Gráinne Long,[4] Lídia Robert,[5] Cynthia Huang-Bartlett,[6] Massimo Cristofanilli[7]*

[1]Clinical Data Science, Oncology R&D, AstraZeneca, Barcelona, Spain; [2]Tempus AI, Inc., New York, NY, USA; [3]Clinical Data Science, Oncology R&D, AstraZeneca, Gaithersburg, MD, USA; [4]Clinical Data Science, Oncology R&D, AstraZeneca, Cambridge, UK; [5]Late Development, Oncology R&D, AstraZeneca, Barcelona, Spain; [6]Late Development, Oncology R&D, AstraZeneca, Gaithersburg, MD, USA; [7]Weill-Cornell Medicine / New York-Presbyterian Hospital, New York, NY, USA.

*Presenting author.

Objective

- To characterize real-world second-line treatment use and clinical outcomes (rwPFS and rwOS) in patients with mBC with a positive *ESR1*m test after first-line treatment with AI+CDK4/6i using data from electronic health databases

Conclusions

- In patients with *ESR1*m HR-positive mBC, the most common real-world second-line treatments were ET+CDK4/6i, elacestrant monotherapy, and ET+PI3Ki/AKTi/mTORi
- *ESR1*m defines a difficult-to-treat subgroup, with rwPFS outcomes highlighting the need for more effective strategies that address resistance to AI+CDK4/6i and improve patient outcomes
 - Across treatments, rwPFS estimates were consistent among patients who received ≥6 months or ≥12 months of first-line AI+CDK4/6i
- Heterogeneity in second-line treatment use and the poor median rwOS support the need for novel treatment options in this setting
- Results were broadly consistent across two independent US electronic health record databases (Flatiron Health and Tempus)

Poster
Supplementary materials
Narrated overview

Please scan this quick response (QR) code with your smartphone camera or app to obtain a copy of these materials. Alternatively, please use the link below.

https://bit.ly/43GpSaO

Copies of this poster obtained through this QR code are for personal use only and may not be reproduced without permission from SABCS® and the authors of this poster.

This presentation is the intellectual property of the author/presenter.
Corresponding author email address: spencer.james@astrazeneca.com

Introduction

- ET (most commonly an AI) plus CDK4/6i is the current standard-of-care first-line treatment for HR-positive/HER2-negative mBC[1,2]
 - Clinical outcomes with subsequent therapies tend to worsen following progression on first-line AI+CDK4/6i[3,4]
- *ESR1*m lead to constitutive (estrogen-independent) activation of estrogen receptor alpha and are rare (<5%) at initial diagnosis of mBC
- At disease progression following first-line treatment with an AI+CDK4/6i, *ESR1*m are detectable in the tumors of ~40% of patients[5–7] and are recognized as the most common mechanism of acquired resistance to AI treatment
- Current guidelines recommend *ESR1*m testing at the time of first-line treatment progression for informing optimal and targeted treatment decisions[1,2]

Methods

- This retrospective cohort study used data from two US de-identified electronic health record-derived data sets: primary analyses were conducted using data from the Flatiron Health database (**Figure 1**). A confirmatory analysis was performed using data derived from the Tempus database and is presented in the supplementary appendix
 - The US Flatiron Health Electronic Data Mart database comprises de-identified, structured and unstructured, longitudinal patient-level demographic, clinical, and genomic data curated via a technology-enabled abstraction method
 - The Tempus real-world multimodal database encompasses patient demographics, treatment histories, and outcomes, as well as molecular data
- Inclusion criteria included a confirmed diagnosis of HR-positive/HER2-negative mBC, first-line treatment with AI+CDK4/6i (initiated before December 31, 2024), a positive *ESR1*m test before initiation of second-line therapy, and a second-line therapy regimen adherent to NCCN Breast Cancer Guidelines v4.2025 (**Figure 2**)
- Patients were followed until June 30, 2025
- Descriptive statistics were used to summarize demographics, baseline clinical characteristics, and treatment patterns
- The primary objectives were to estimate rwPFS and rwOS following treatment with second-line therapies
- rwPFS was defined as time from second-line start to disease progression, start of third-line therapy, death, or censoring at last known activity; rwOS was defined as time from second-line start to death or censoring
- The index date comprised the start of second-line therapy
- Multivariable Cox proportional hazards regression was used to adjust for prespecified confounders, including age at second-line start, ECOG PS, metastatic site, and first-line treatment duration
- Hazard ratios of rwPFS and rwOS for each second-line therapy regimen were calculated relative to fulvestrant, which was chosen as the reference due to it being the historical standard of care in this setting
- 12-month survival rates were included, given the potential for short follow-up times to affect estimation of median real-world outcomes
- Sensitivity analyses were conducted by duration of first-line AI+CDK4/6i

Figure 1. Study design

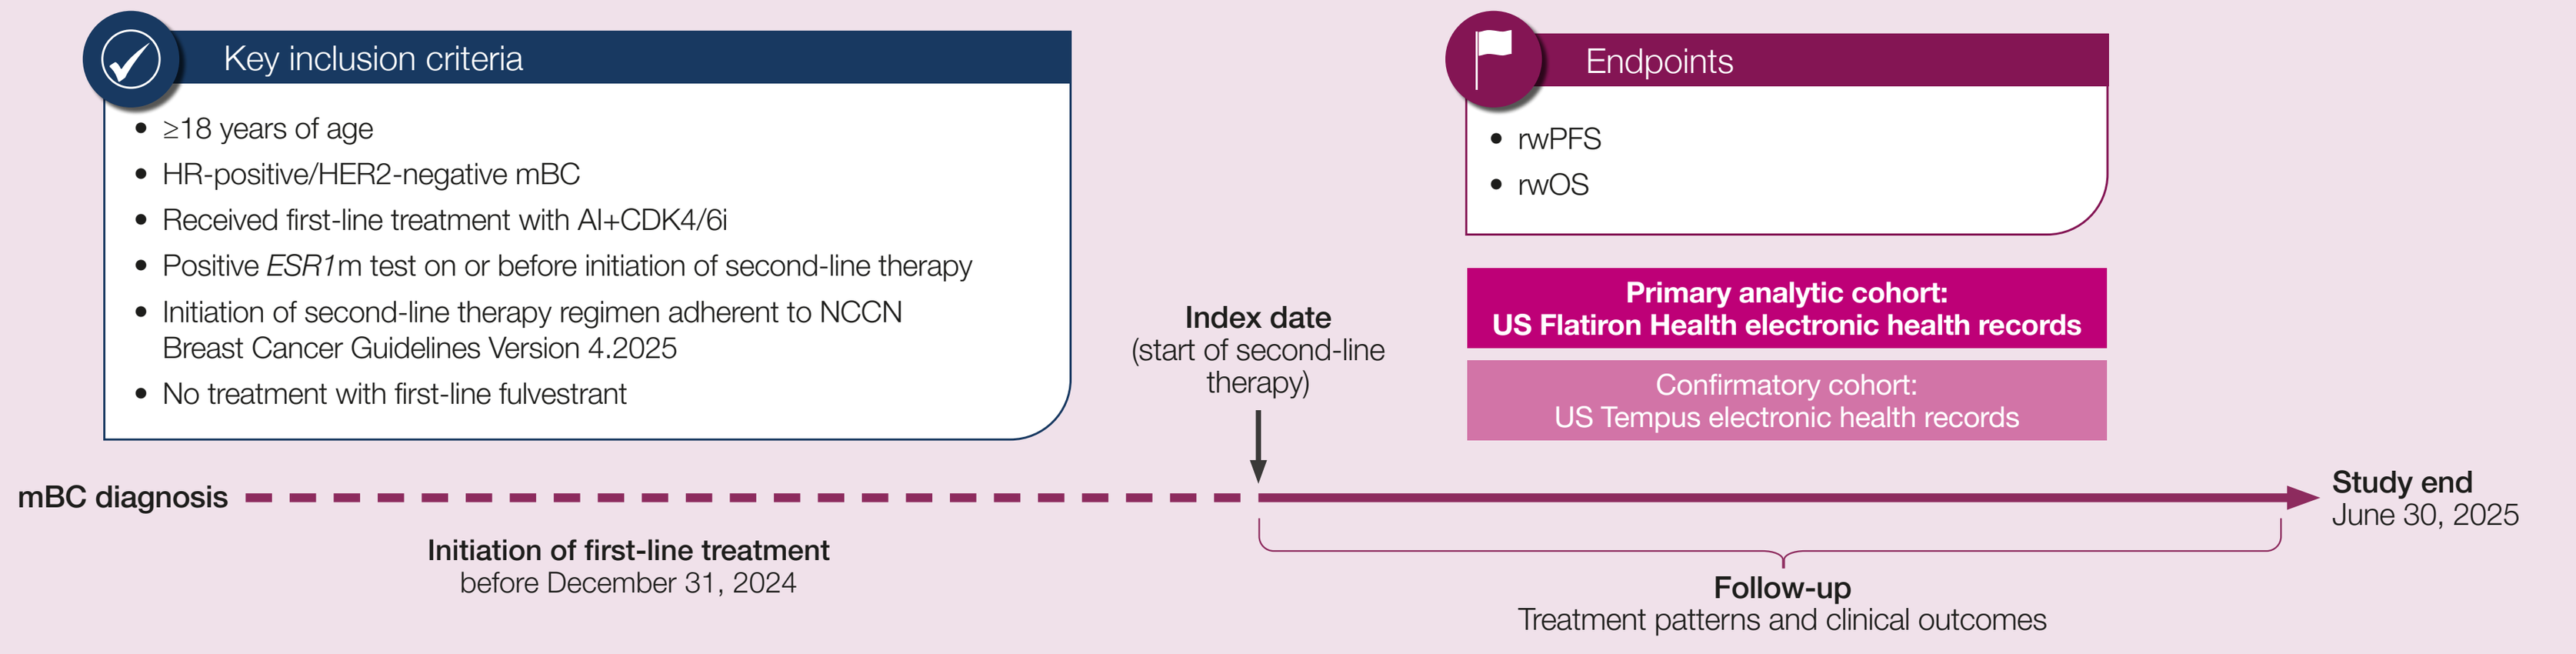
Key inclusion criteria
≥18 years of age
HR-positive/HER2-negative mBC
Received first-line treatment with AI+CDK4/6i
Positive ESR1m test on or before initiation of second-line therapy
Initiation of second-line therapy regimen adherent to NCCN Breast Cancer Guidelines Version 4.2025
No treatment with first-line fulvestrant
Endpoints
rwPFS
rwOS
Primary analytic cohort: US Flatiron Health electronic health records
Confirmatory cohort: US Tempus electronic health records
Index date (start of second-line therapy)
mBC diagnosis
Initiation of first-line treatment before December 31, 2024
Study end June 30, 2025
Follow-up
Treatment patterns and clinical outcomes

rwPFS was defined as time from second-line start to disease progression, start of third-line therapy, death, or censoring; rwOS was defined as time from second-line start to death or censoring.

Results

Baseline characteristics

- A total of 226 eligible patients were identified in the Flatiron Health database (**Figure 2**)
- The median age at diagnosis was 63 years (range: 57.0–71.0), most patients were White (74.1%) with ECOG PS ≤1 (71.2%). Liver metastases were common (51.8%), and 25.2% of patients had bone-only metastasis (**Table 1**)

Second-line treatment patterns

- Second-line treatments included ET+CDK4/6i, elacestrant monotherapy, ET+PI3Ki/AKTi/mTORi, chemotherapy/ADC, fulvestrant monotherapy, or other (**Table 2**)

Figure 2. Flow chart diagrams of patients included in the Flatiron Health analysis

- Patients with mBC (n=42,636)
- Patients ≥18 years of age (n=42,458)
- Patients with a metastatic diagnosis on or after February 3, 2015 (n=31,664)
- Receiving first-line treatment (n=27,535)
- Confirmed HR-positive/HER2-negative tumors (n=18,945)
- First-line started >6 months prior to data release (n=18,481)
- First-line treatment with CDK4/6i (n=9,331)
- First-line treatment with AI+CDK4/6i (n=6,471)
- Initiation of second-line therapy included in current NCCN guideline (n=3,168)
- Patients without prior fulvestrant use and not enrolled in a clinical trial (n=2,834)
- *ESR1*m results anytime on or before second-line initiation (n=995)
- Patients with a positive *ESR1*m result (n=226)

Effectiveness: rwPFS and rwOS

- In a multivariate adjusted model, rwPFS and rwOS estimates in patients with *ESR1*m that were treated with second-line therapies are shown in **Figure 3**
- median rwPFS remained <6 months across all guideline-recommended second-line treatments for patients with *ESR1*m
- Apart from second-line ET+CDK4/6i, the 95% CI for rwPFS and rwOS hazard ratios for the other treatments crossed 1, suggesting that real-world outcomes were not largely improved compared with fulvestrant
- A sensitivity analysis of rwPFS and rwOS in patients by duration of first-line AI+CDK4/6i is shown in **Table 3**
- rwPFS and rwOS were consistent between patients who received ≥6 months or ≥12 months of first-line AI+CDK4/6i
- Outcomes differed in patients who received other treatments by duration of first-line AI+CDK4/6i, but due to the small sample sizes (≥6 months, n=6; ≥12 months, n=3), results should be interpreted with caution

Confirmatory analysis in the Tempus database

- A total of 135 eligible patients were identified in the Tempus database (**Figure S1**). Baseline demographics and disease characteristics were similar to those of patients in the Flatiron database (**Table S1**)
- Second-line treatment patterns were broadly similar between the Flatiron Health database and the Tempus database (**Table S2**), although use of ET+CDK4/6i was more common in the Flatiron Health database (32.3% vs 13.3%)
- In patients with *ESR1*m treated in the Tempus database, median rwPFS was 4.1 (95% CI 3.4–5.4) months (**Table S3**)
- Median rwPFS varied by treatment, and estimates were comparable to those made from data in the Flatiron Health database
- In patients with *ESR1*m treated in the Tempus database, median rwOS was 25.2 (95% CI 20.1–36.6) months (**Table S3**)
- Trends in median rwOS should be interpreted with caution due to the short follow-up in both datasets and the differences in the duration of follow-up (**Table 1 and S1**)

Table 1. Baseline characteristics

Characteristic		Flatiron cohort (n=226)
Median (IQR) age at diagnosis, years		63.0 (57.0–71.0)
Race, n (%)	White	152 (74.1)
	Black or African American	32 (15.6)
	Asian	5 (2.4)
	Other	16 (7.8)
ECOG PS at start of second-line, n (%)	0	85 (37.6)
	1	76 (33.6)
	2	22 (9.7)
	3	6 (2.7)
	4	–
	Missing	37 (16.4)
Metastases, n (%)	Bone	197 (87.2)
	Liver	117 (51.8)
	Lung	74 (32.7)
	Pleural	45 (19.9)
	Brain	26 (11.5)
	Bone-only	57 (25.2)
Median (IQR) follow-up, months		12.6 (6.1–20.4)

Table 2. Treatment characteristics

Characteristic		Flatiron cohort (n=226)
Prior adjuvant AI		65 (28.8)
AI+CDK4/6i, n (%)	<6 months	32 (14.2)
	≥6 months	194 (85.8)
	≥12 months	163 (72.1)
	≥18 months	133 (58.8)
Treatment-free interval, n (%)	<12 months	36 (15.9)
	≥12 months	65 (28.8)
	No prior	125 (55.3)
Time from *ESR1*m detection to second-line therapy, n (%)	0–28 days	96 (42.5)
	29+ days	130 (57.5)
Second-line therapy, n (%)	ET+CDK4/6i	73 (32.3)
	Elacestrant monotherapy	51 (22.6)
	ET+PI3Ki/AKTi/mTORi	43 (19.0)
	Chemotherapy/ADC	27 (11.9)
	Fulvestrant	25 (11.1)
	Other*	7 (3.1)

*Other treatments included olaparib (n=5), abemaciclib (n=1), and talazoparib (n=1).

Figure 3. Real-world PFS and OS in patients with *ESR1*m treated with second-line therapies in the Flatiron Health database, by treatment

		Median (95% CI), months	12-month rate (95% CI), %
rwPFS	All patients (n=226)	4.8 (4.0–5.6)	22.9 (17.5–29.9)
	Fulvestrant (n=25) (**reference**)	3.0 (2.8–6.3)	24.1 (13.6–42.7)
	ET+CDK4/6i (n=73)	5.2 (3.6–6.1)	27.9 (18.9–41.3)
	Elacestrant monotherapy (n=51)	5.3 (4.4–7.5)	21.4 (4.2–100.0)
	ET+PI3Ki/AKTi/mTORi (n=43)	5.7 (4.0–10.2)	29.1 (17.3–49.2)
	Chemotherapy/ADC (n=27)	4.8 (2.8–6.7)	13.4 (4.7–37.9)
	Other (n=7)*	2.5 (0.9–NE)	NA (NE–NE)
rwOS	All patients (n=226)	24.9 (18.8–30.9)	72.3 (66.1-79.1)
	Fulvestrant (n=25) (**reference**)	17.4 (11.8–30.9)	63.7 (43.8–92.6)
	ET+CDK4/6i (n=73)	39.1 (19.8–NE)	73.2 (63.0–85.1)
	Elacestrant monotherapy (n=51)	17.8 (16.2–NE)	72.9 (60.4–88.1)
	ET+PI3Ki/AKTi/mTORi (n=43)	26.7 (15.12–NE)	80.6 (68.6–94.7)
	Chemotherapy/ADC (n=27)	17.6 (13.8–NE)	71.2 (55.2–91.8)
	Other (n=7)*	7.1 (0.9–NE)	42.9 (18.2–100.0)

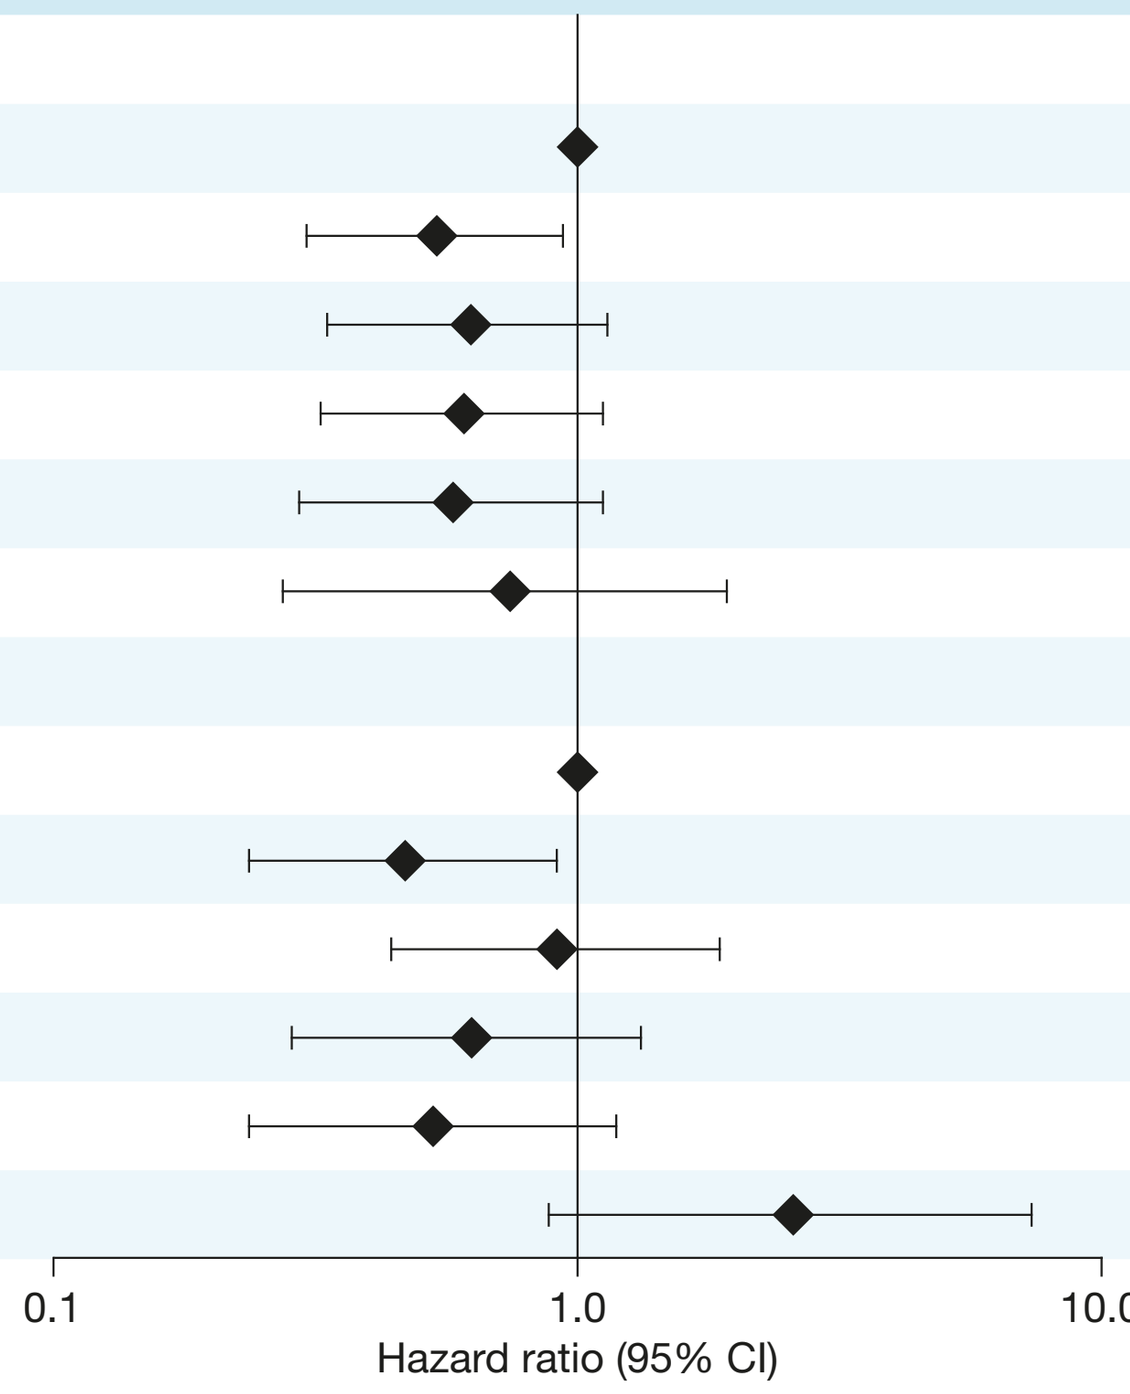
0.1
1.0
10.0
Hazard ratio (95% CI)

*Other treatments included olaparib (n=5), abemaciclib (n=1), and talazoparib (n=1).

Table 3. Real-world PFS in patients with *ESR1*m treated with second-line therapies in the Flatiron Health database by treatment and duration of first-line therapy

Duration of first-line AI+CDK4/6i		n=	rwPFS median (95% CI), months	12-month PFS rate (95% CI), %	rwOS median (95% CI), months	12-month OS rate (95% CI), %
≥6 months	All patients	194	4.6 (3.7–5.6)	20.9 (15.4–28.4)	23.5 (17.8–30.9)	72.2 (65.6–79.6)
	Fulvestrant	21	2.8 (1.6–NE)	NA (NE–NE)	19.1 (11.8–NE)	63.9 (42.5–96.3)
	Chemotherapy/ADC	24	3.5 (2.8–7.6)	14.5 (5.2–40.9)	17.6 (13.8–NE)	68.4 (51.5–91.0)
	ET+CDK4/6i	52	4.6 (2.7–5.9)	21.0 (12.2–36.4)	39.1 (19.8–NE)	72.3 (60.4–86.4)
	ET+PI3Ki/AKTi/mTORi	42	5.4 (4.0–10.2)	27.0 (15.4–47.4)	26.7 (15.1–NE)	80.0 (67.7–94.6)
	Elacestrant monotherapy	49	5.7 (4.5–7.5)	25.2 (14.3–44.3)	17.8 (16.2–NE)	73.8 (61.0–89.3)
	Other*	6	4.3 (2.07–NE)	25.0 (5.0–100)	19.2 (3.5–NE)	50.0 (22.5–100.0)
≥12 months	All patients	163	5.3 (4.4–5.9)	23.6 (17.2–32.2)	26.7 (23.5–47.1)	78.5 (71.8–85.9)
	Fulvestrant	17	3.0 (1.1–NE)	NA (NE–NE)	19.3 (12.7–NE)	74.6 (52.8–100.0)
	Chemotherapy/ADC	17	5.6 (3.0–NE)	21.2 (7.8–57.1)	NA (NE–NE)	80.7 (63.2–100.0)
	ET+CDK4/6i	42	4.6 (2.7–6.1)	21.3 (11.6–39.2)	39.1 (24.9–NE)	78.8 (66.7–93.1)
	ET+PI3Ki/AKTi/mTORi	37	5.8 (4.2–12.2)	30.0 (17.3–52.2)	26.7 (15.1–NE)	83.7 (71.5–98.0)
	Elacestrant monotherapy	47	5.7 (4.5–7.5)	25.7 (14.6–45.3)	17.8 (16.2–NE)	73.1 (60.1–89.0)
	Other*	3	31.3 (2.5–NE)	66.7 (30.0–100.0)	31.30 (NE–NE)	100 (100.0–100.0)

Hazard ratios were adjusted for the *a priori* confounders age, ECOG PS at second-line initiation, duration of first-line AI+CDK4/6i, and site of metastasis.
*Other treatments included olaparib, abemaciclib, and talazoparib.

Abbreviations

ADC, antibody–drug conjugate; AI, aromatase inhibitor; AKTi, protein kinase B inhibitor; CI, confidence interval; CDK4/6i, cyclin-dependent kinase 4/6 inhibitor; ECOG PS, Eastern Cooperative Oncology Group performance status; *ESR1*m, mutations in the *ESR1* gene; ET, endocrine therapy; HER2, human epidermal growth factor receptor 2; HR, hormone receptor; IQR, interquartile range; mBC, metastatic breast cancer; mTORi, mechanistic target of rapamycin inhibitor; NA, not available; NCCN, National Comprehensive Cancer Network; NE, non-evaluable; PI3Ki, phosphoinositide 3-kinase inhibitor; (rw)OS, (real-world) overall survival; (rw)PFS, (real-world) progression-free survival; SD, standard deviation.

References

1. NCCN Clinical Practice Guidelines in Oncology (NCCN Guidelines®) for Breast Cancer Version 5.2025. © National Comprehensive Cancer Network, Inc. 2025. All rights reserved.
2. NCCN Clinical Practice Guidelines in Oncology (NCCN Guidelines®) for Breast Cancer Version 4.2024. © National Comprehensive Cancer Network, Inc. 2024. All rights reserved.
3. Herrscher H, et al. *Breast Cancer Res Treat* 2020;179(2):371–376.
4. Kimura T, et al. *Breast Cancer Res Treat* 2025;211(1):233–244.
5. Bidard F-C, et al. *J Clin Oncol* 2022;40(28):3246–56.
6. Bhave MA, et al. *Breast Cancer Res Treat* 2024;207(3):599–609.
7. Chaudhary N, et al. *Nat Commun* 2016;7:11579.

Acknowledgments and disclosures

- Massimo Cristofanilli reports grants to Weill Cornell Medicine from AstraZeneca, personal fees as a consultant for AstraZeneca, BriaCell, Datar Genomics, Menarini Silicon Biosystems, Merck, Olaris, and Repare Therapeutics, and personal fees as a speaker and consultant for Pfizer
- The study is sponsored by AstraZeneca
- Medical writing assistance was provided by Suzanne Patel, PhD, and Leigh-Ann Booth, PhD, from BOLDSCIENCE Inc., funded by AstraZeneca